Impact of immunoglobulin A nephropathy on work productivity and activity impairment in the United States: Results from a real-world study

Briana Ndife,[1] Michel Kroes,[2] Carolina Aldworth,[1] Aneesh Thomas George,[3] Luis Prieto,[1] Jonathan DeCourcy,[4] Jade Garratt-Wheeldon,[4] Richard Lafayette[5]

[1]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [2]Novartis Pharma AG, Basel, Switzerland; [3]Novartis Healthcare Pvt. Ltd, Hyderabad, India; [4]Adelphi Real World, Bollington, UK; [5]Department of Medicine, Division of Nephrology, Stanford University School of Medicine, Stanford, CA, USA

Introduction

- IgAN is the most prevalent form of primary glomerulonephritis, affecting 7–21 adults per million each year in the United States[1–4]
- IgAN typically affects younger adults (20–30 years) more than older adults (>65 years)[2] and reduces a patient's life expectancy by >10 years[5]
- Up to 50% of patients with IgAN develop kidney failure within 20 years of diagnosis necessitating dialysis and/or kidney transplant,[6] placing considerable socioeconomic burden on patients, their caregivers, and healthcare systems
- In this analysis, we describe the economic burden in the form of work productivity and activity impairment due to IgAN in the United States

Methods

- This was a retrospective analysis of secondary data from the Adelphi Real World IgAN Disease Specific Programme, a cross-sectional survey of IgAN-treating nephrologists and their consecutively consulting patients with IgAN from the United States
- The survey was conducted in the US (as well as in the UK, Spain, France, Germany, Italy, China, and Japan) from June to October 2021
- Participating nephrologists completed patient record forms for their consecutively consulting patients diagnosed with IgAN, providing a representative "point in time" sample of consulting patients. The DSP methodology has been previously published in detail[7]
- The same consulting patients were then invited by their physicians to fill out a patient–self-completed form on a voluntary basis
- Patient-reported data were matched to the physician-reported clinical data to allow direct comparison of clinical characteristics and patient-reported outcomes
- The survey captured data on insurance coverage of patients and impact of the disease on patients' routine activities and their work using the Work Productivity and Activity Impairment – General Health questionnaire (WPAI-GH)

Results

- A total of 68 patients completed their forms and were included in this analysis
- The median patient-reported age when IgAN symptoms were observed was 36 years. Mean duration of diagnosis of IgAN at the time of survey was 4.1±5.7 years (**Table**)
- Nearly two-third (65.7%) of patients had proteinuria ≥1 g/day at the time of survey, which is a known risk factor for IgAN progression (**Table**)

Conclusions

- The results of this analysis indicate that IgAN typically affects the prime, working-age population in the United States, resulting in noticeable impairment in work productivity and activity
- Patients with high proteinuria (≥1 g/day) report greater severity of disease and show greater impairment in work productivity and activity, highlighting the need for therapies that reduce proteinuria, a known risk-factor for disease progression in IgAN

Key results

Patient-reported insurance plan/ coverage options

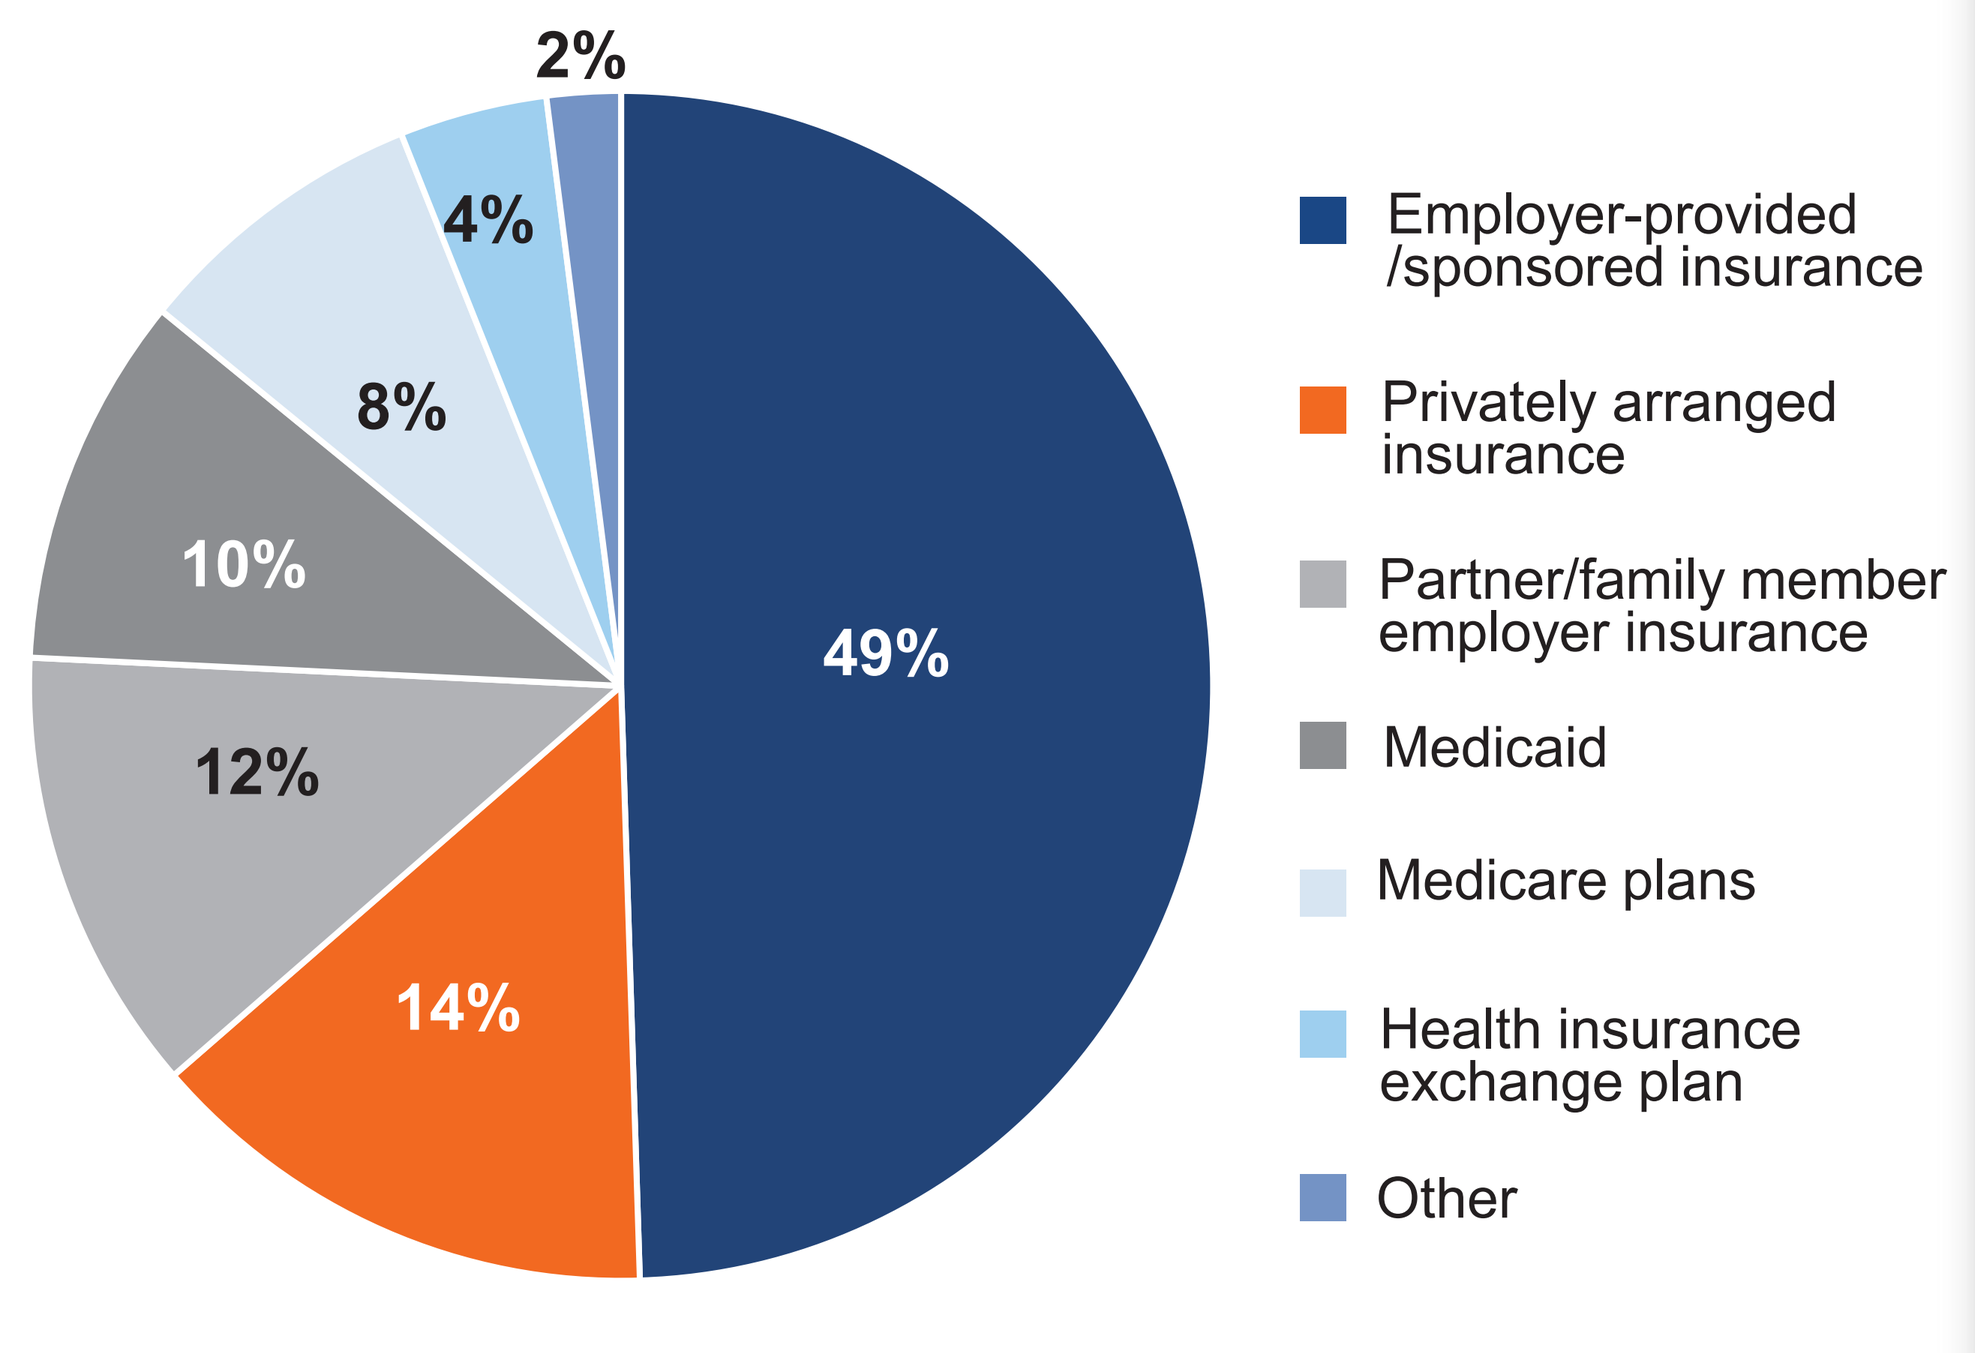
2%
4%
8%
10%
12%
14%
49%
Employer-provided /sponsored insurance
Privately arranged insurance
Partner/family member employer insurance
Medicaid
Medicare plans
Health insurance exchange plan
Other

Number of patients with available data = 49

- Nearly all patients (96%; n=55/57) had health insurance that covered IgAN treatment. Among the surveyed patients, 49% had employer-provided/sponsored insurance (n=24/49), while 8% (n=4/49) were covered under various Medicare plans and 10% (n=5/49) had Medicaid (or equivalent) coverage

Current employment status of the patients surveyed

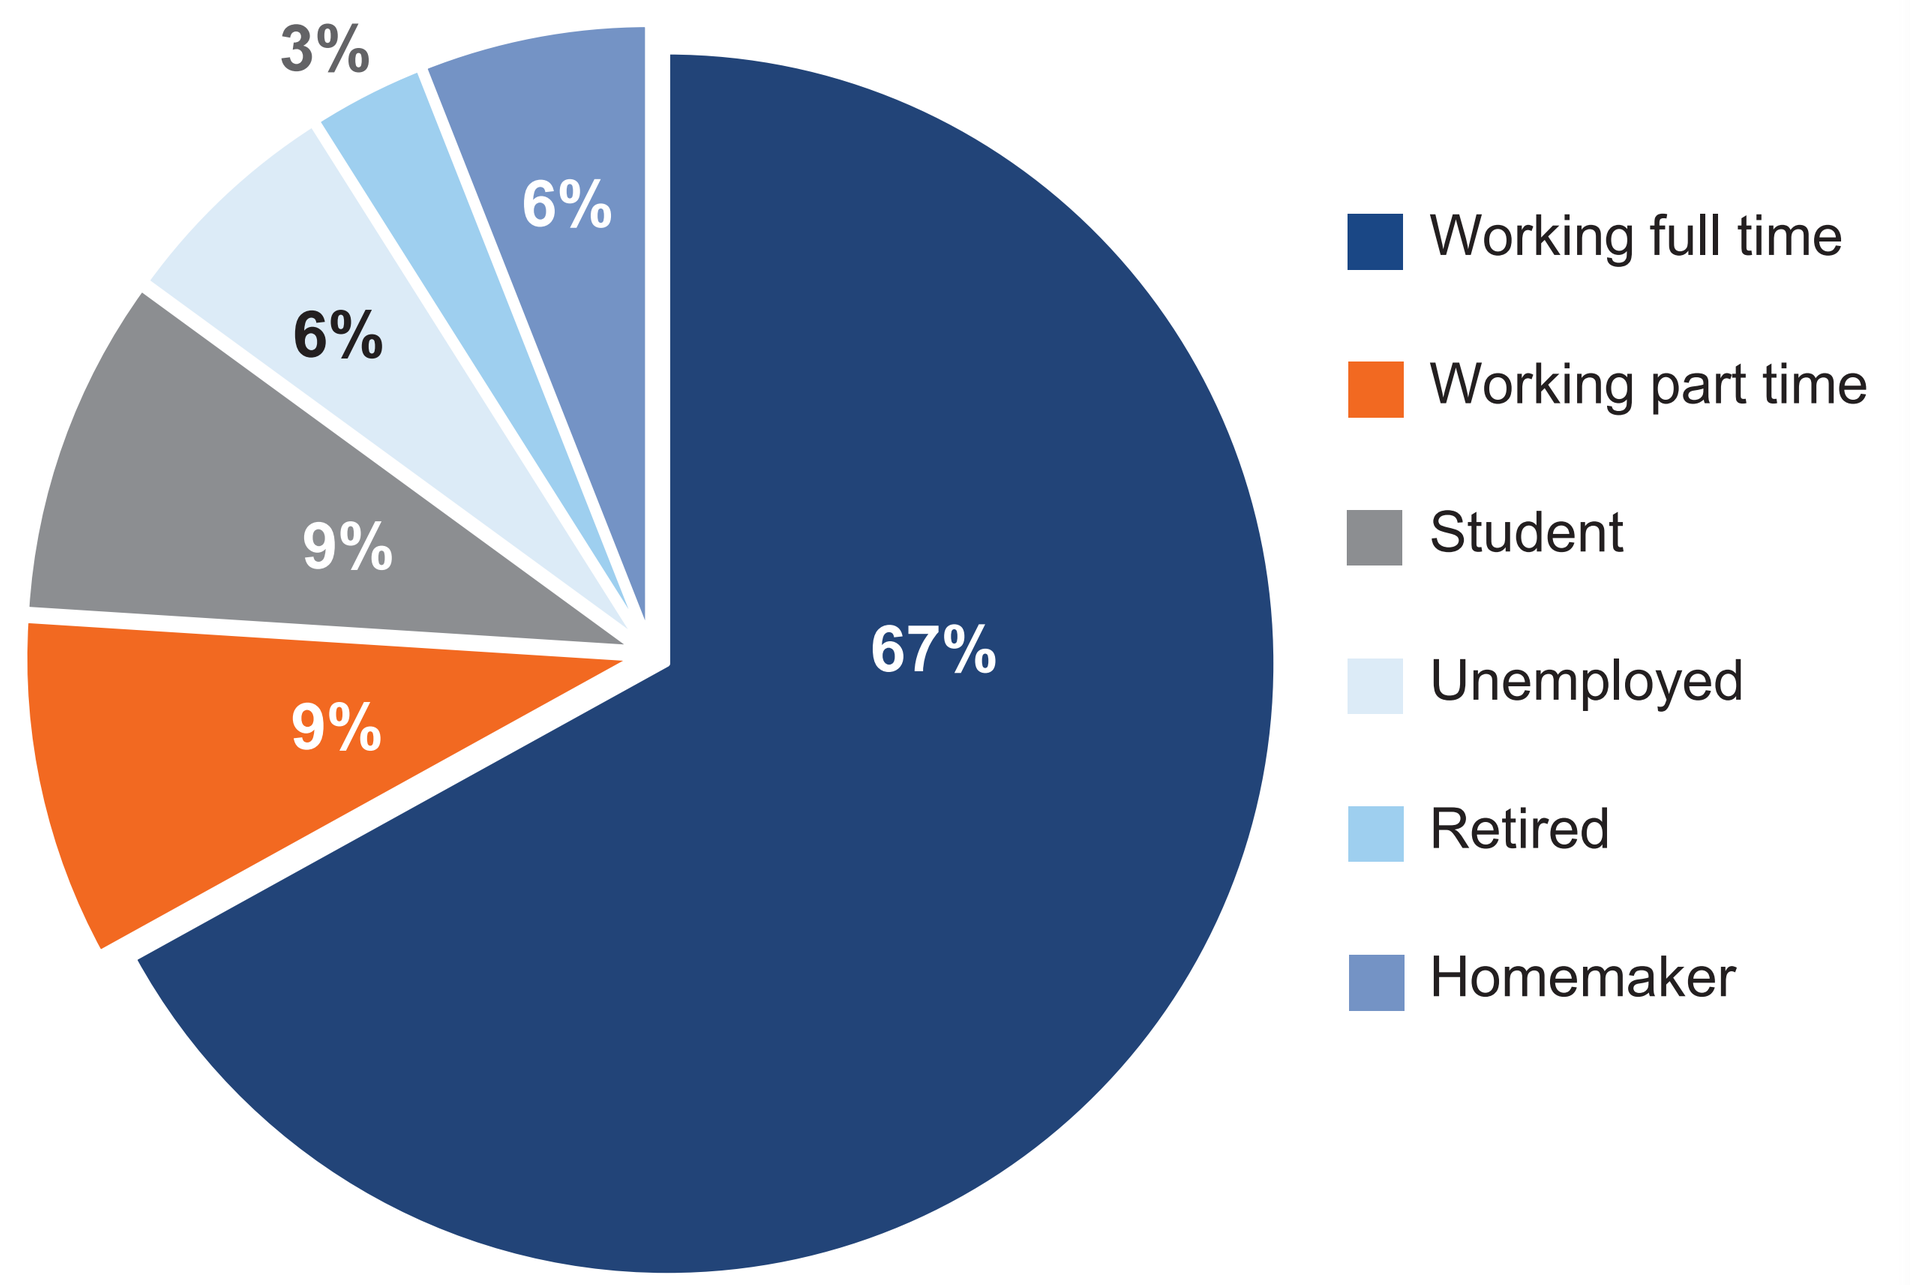
3%
6%
6%
9%
9%
67%
Working full time
Working part time
Student
Unemployed
Retired
Homemaker

Number of patients with available data = 66

- A majority of patients (>75%; n=50/66) were employed either full time or part time

Patient-reported WPAI due to IgAN by proteinuria levels

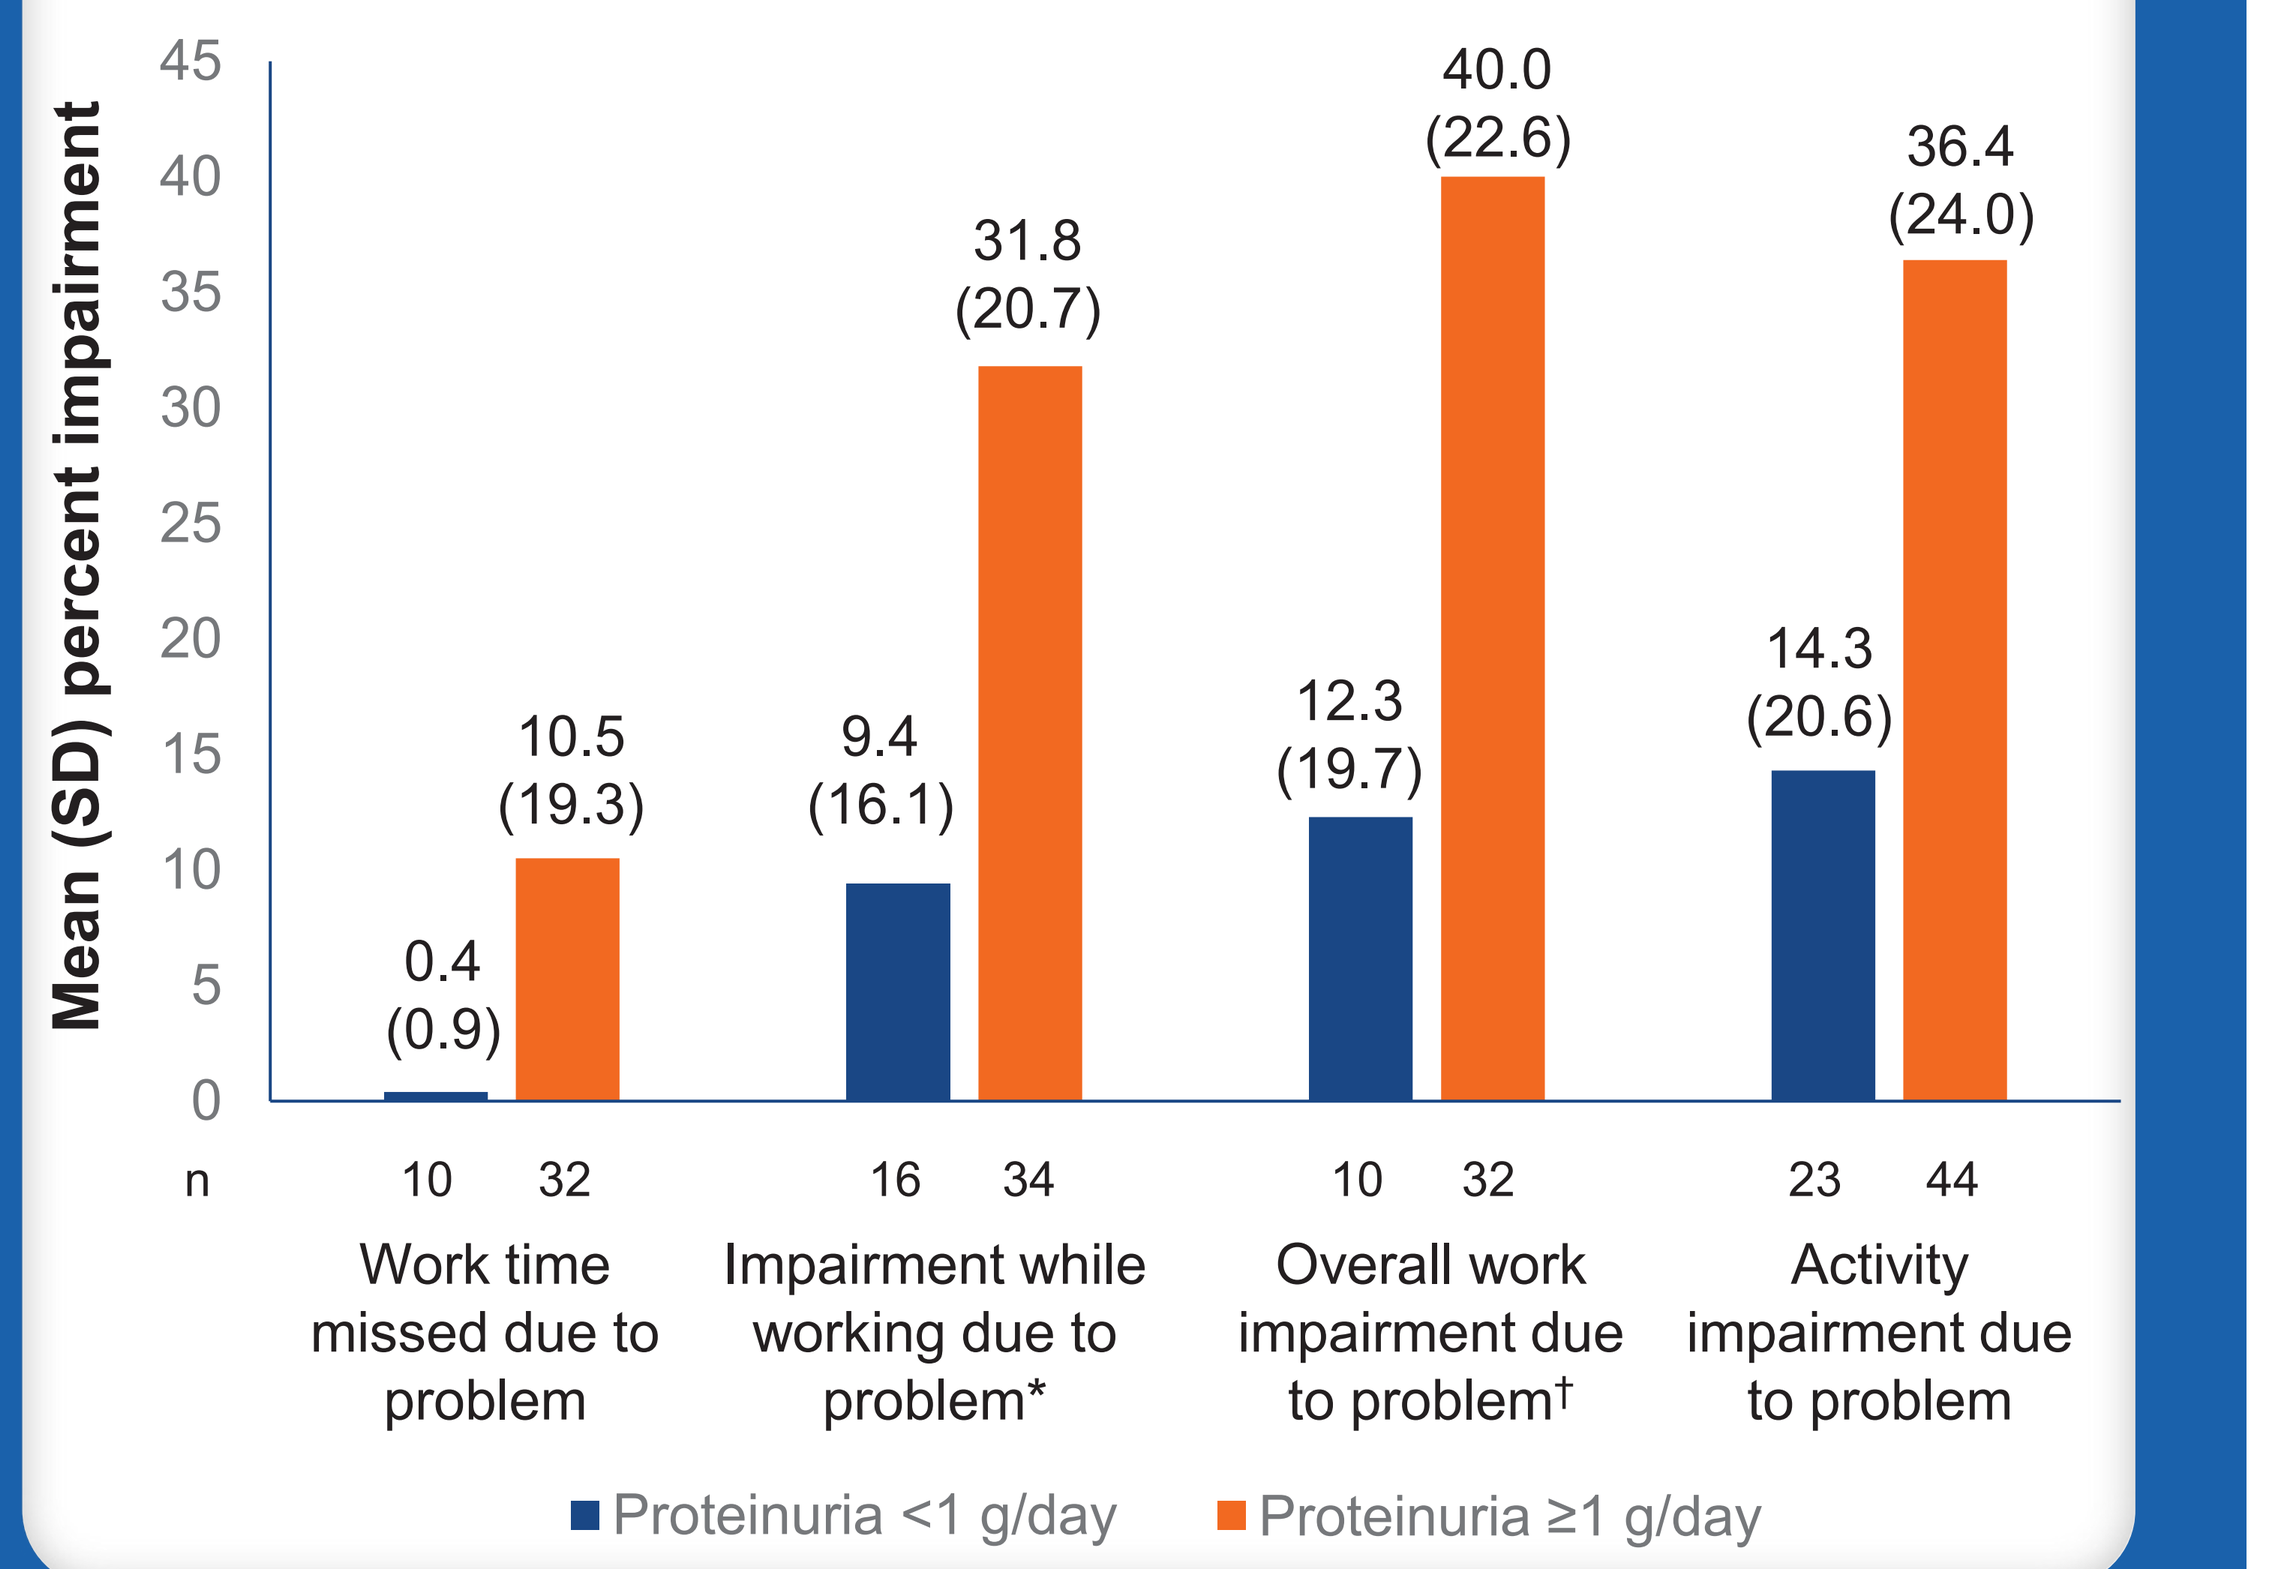
Mean (SD) percent impairment
45
40
35
30
25
20
15
10
5
0
0.4 (0.9)
10.5 (19.3)
9.4 (16.1)
31.8 (20.7)
12.3 (19.7)
40.0 (22.6)
14.3 (20.6)
36.4 (24.0)
n
10 32
16 34
10 32
23 44
Work time missed due to problem
Impairment while working due to problem*
Overall work impairment due to problem†
Activity impairment due to problem
Proteinuria <1 g/day
Proteinuria ≥1 g/day

- At the time of the survey, on average, patients reported 8.1% missed work time (n=42), 24.6% impairment while working (n=50), 33.4% overall work impairment (n=42), and 28.8% activity impairment (n=67) due to IgAN

*The effect that the disease has had on the patient's productivity while working in the last 7 days; †Percent overall work impairment considers this interference in combination with the working hours missed in the last 7 days.

Poster presented at: Academy of Managed Care Pharmacy (AMCP) 2023; March 21–24, 2023; San Antonio, USA.

Table: Patient-reported demographics and clinical characteristics

Characteristics*	Value
Age, years, mean±SD	43.5±12.9
Sex, (%)	
Male	50
Female	50
Race, (%) (n=67)	
White/Caucasian	54
Asian‡	24
Hispanic/Latino	13
African American	4
Other	4
BMI, kg/m^2 (mean±SD)	26.2±4.7
Age when IgAN symptoms were first noticed (median [min, max]) (n=66)	36.0 (9.0, 70.0)
Proteinuria (mean±SD) at the time of survey (n=67)	2.0±3.7
<1 g/day, n (%)	23 (34.3)
≥1 g/day, n (%)	44 (65.7)

*N=68, unless otherwise specified; ‡Asian (Indian subcontinent), Southeast Asian, and Asian (other).

Patient-reported severity of IgAN at the time of survey by proteinuria levels

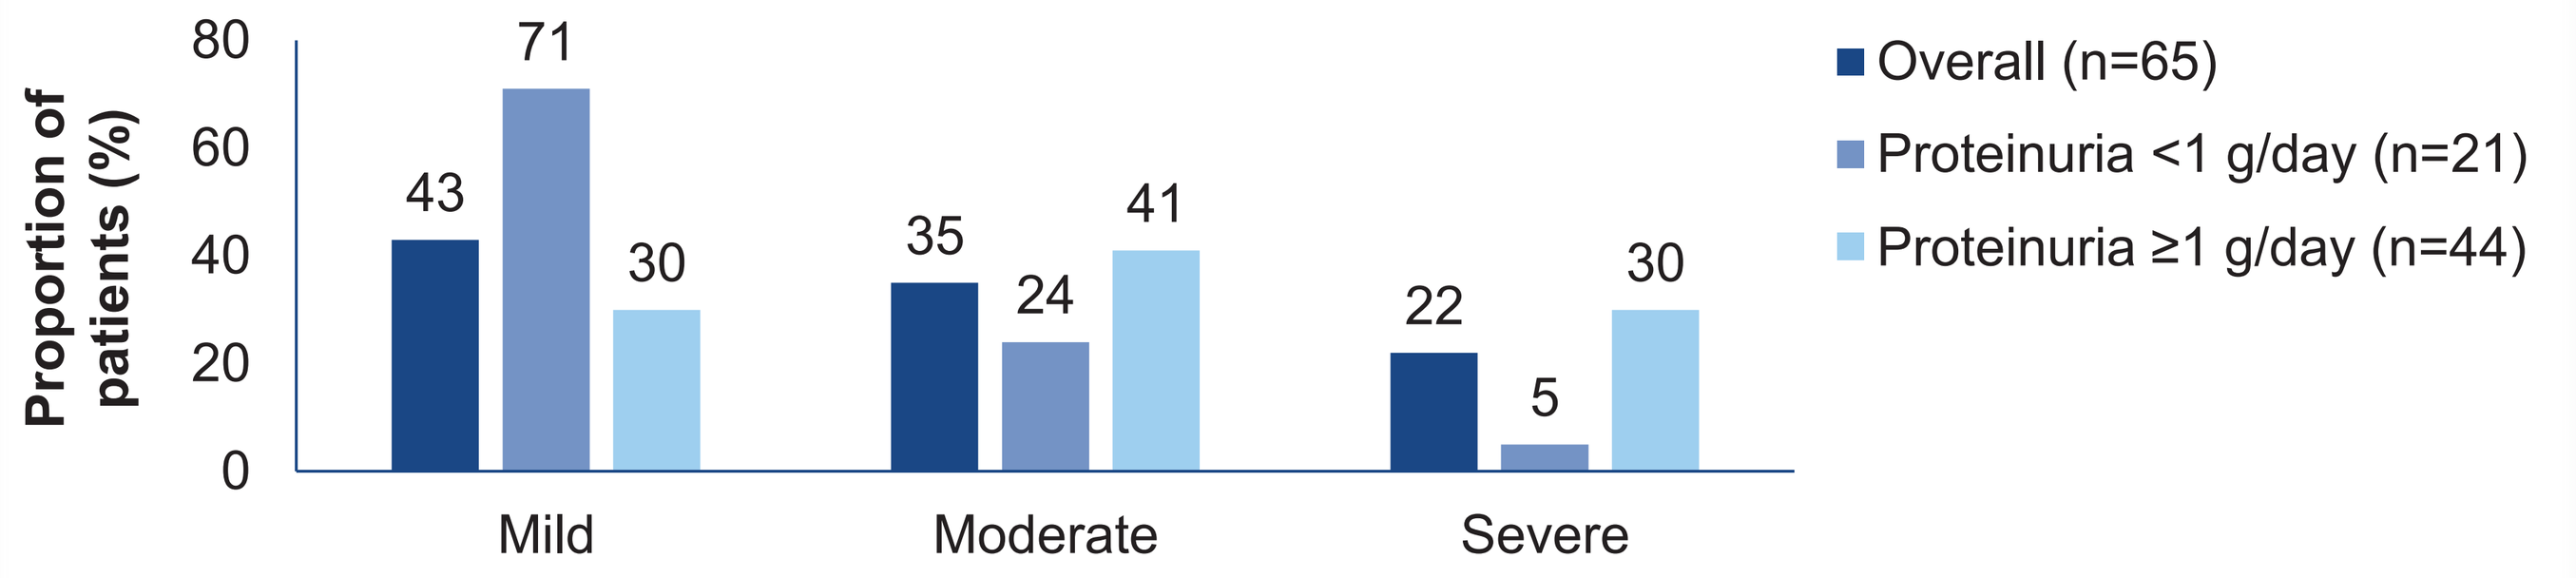
Proportion of patients (%)
80
60
40
20
0
43
71
30
35
24
41
22
5
30
Mild
Moderate
Severe
Overall (n=65)
Proteinuria <1 g/day (n=21)
Proteinuria ≥1 g/day (n=44)

- More than half of patients reported that their IgAN was moderate or severe at the time of survey; a higher proportion of patients with proteinuria ≥1 g reported that their IgAN was severe

Abbreviations

BMI, body mass index; IgAN, IgA nephropathy; SD, standard deviation; WPAI, work productivity and activity impairment

Funding

Data collection was undertaken by Adelphi Real World as part of an independent survey. Novartis is one of the subscribers to the IgAN Disease Specific Programme.

Disclosures

M. Kroes, L. Prieto, C. Aldworth, and B. Ndife are employees and shareholders of Novartis; A.T. George is an employee of Novartis; J. DeCourcy and J. Garratt-Wheeldon are employees of Adelphi Real World; R. Lafayette is an employee of Stanford University Medical Center; his employers have received research funding from Omeros, Vera, Chinook, Alexion, Otsuka, Calliditas and has provided consultancy for: Omeros, Vera, Calliditas, Chinook, Alexion, Otsuka, Novartis.

References

1. Wyatt RJ, et al. N Engl J Med. 2013;368:2402–14.
2. Lai KN, et al. Nat Rev Dis Primers. 2016;2:16001.
3. Sim JJ, et al. Am J Kidney Dis. 2016;68:533–544.
4. Swaminathan S, et al. Clin J Am Soc Nephrol. 2006;1:483–487.
5. Rizk DV, et al. Front Immunol. 2019;10:504.
6. Kwon CS, et al. J Health Econ Outcomes Res. 2021;8:36–45.
7. Anderson P, et al. Curr Med Res and Opin. 2008;24:3063–72.

Acknowledgments

Medical writing assistance was provided by Nagabhushana Ananthamurthy and design support was provided by Ravikishor Babu (both Novartis Healthcare Pvt Ltd, Hyderabad, India).

Scan QR Code for a copy of this poster