Demographic, clinical characteristics and treatment outcomes of C3 glomerulopathy in China: A nationwide retrospective cohort study

2023

TH-PO641

Authors: **Nie Sheng[1], Zhou Shiyu[1], Gao Qi[1], Zhang Shaosen[2], Li Ping[2], Gao Fei[2]**

[1]Division of Nephrology, Nanfang Hospital, Southern Medical University, China. [2]Beijing Novartis Pharma. Co. Ltd., Shanghai, China.

①Background:

- C3 glomerulopathy (C3G) is a rare glomerular disease, and there is limited data on the epidemiology and treatment outcomes of C3G in China.

②Methods:

- We conducted the multicentre observational study using China Renal Data System database (including 24 regional central hospitals across 18 cities in China from 2013 to 2021), which represented a nationwide real-world kidney disease database. We identified potential C3G patients using medical terms in Chinese as no ICD code for C3G so far. We defined the index date as the recorded date of first confirmed diagnosis of C3G in the database. This study provided a descriptive analysis of the clinical characteristics (from 90 days pre-index to 30 days post-index date) and treatment regimen (from the index date to 6 months post-index date). We used t-test for continuous variables and chi-square test for categorical variables for comparisons.

③Results:

- In total of 470,000 patients with chronic kidney disease (CKD) were enrolled in the study, of which 102 patients were identified with C3G based on renal biopsy records (Fig 1 and Table 1).

Fig 1. Patients Characteristics

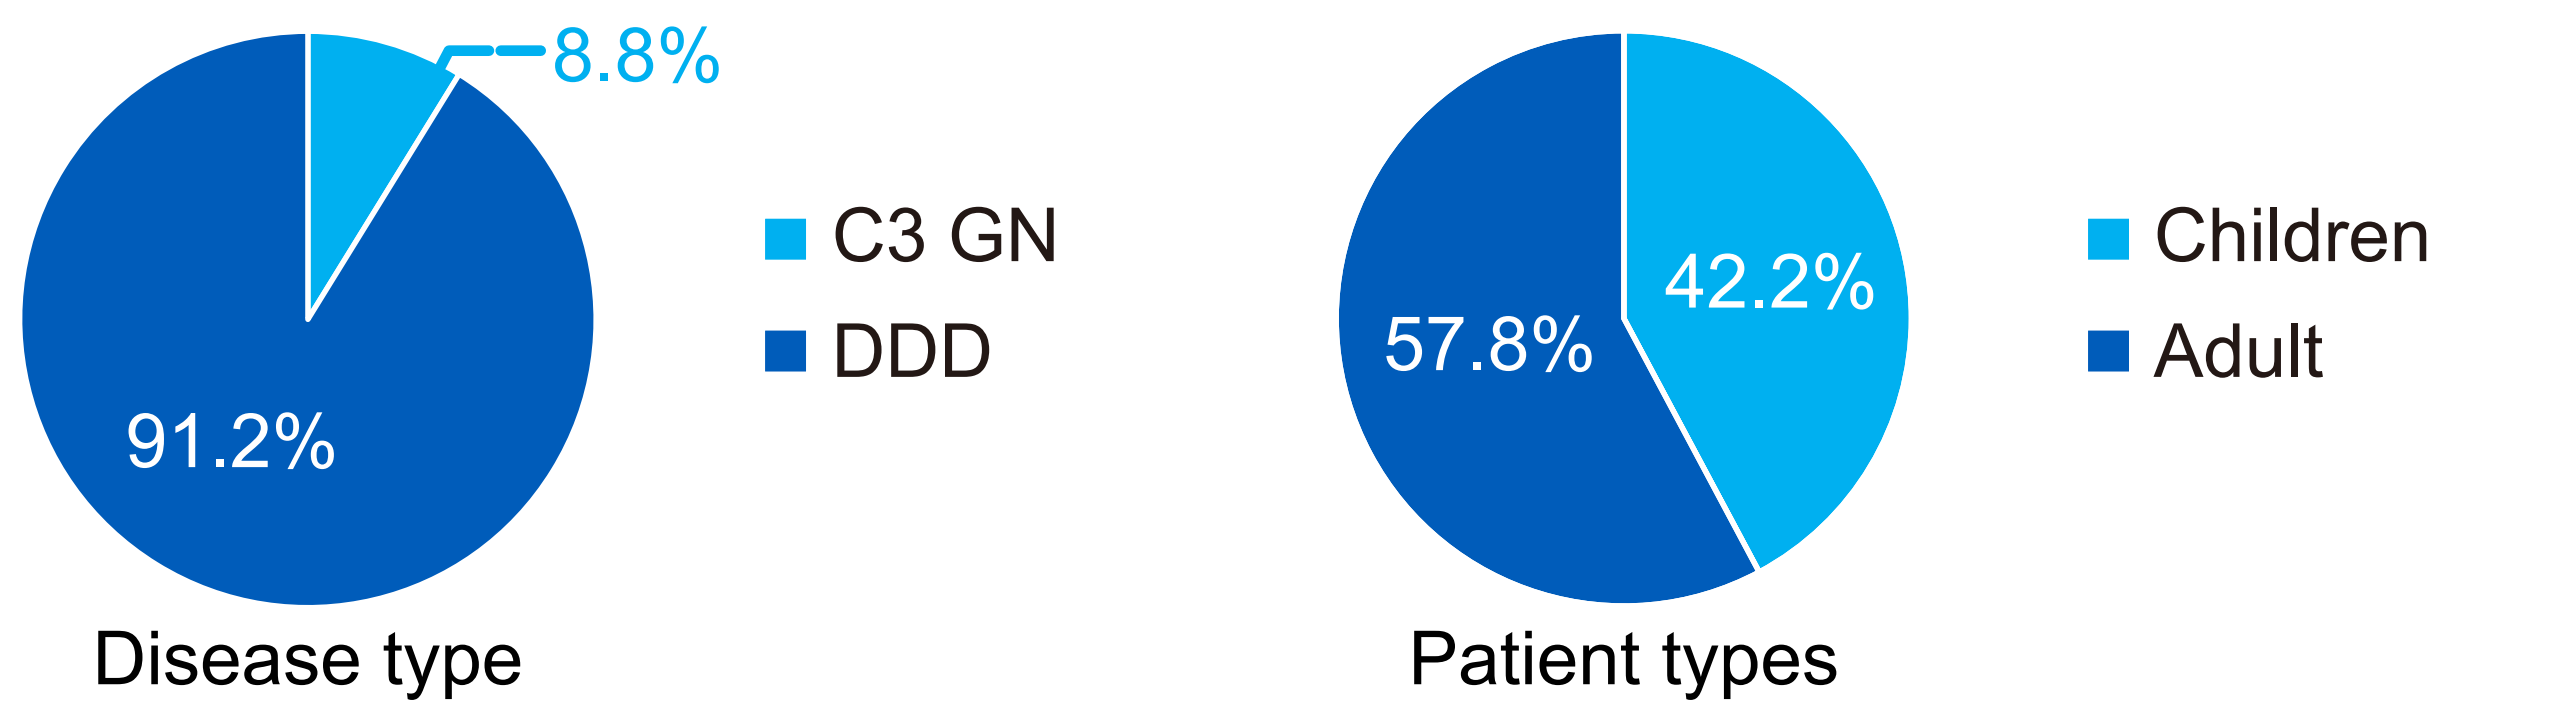
8.8%
91.2%
C3 GN
DDD
Disease type
42.2%
57.8%
Children
Adult
Patient types

Table 1. Baseline characteristics of patients with C3G stratified by age

Demography		
Characteristics	**Children^(N=43)**	**Adult(N=59)**
Age	8.9(6.1-11.2)#	42.1(29.4-53.8)#
Male,n(%)	24(55.8)*	29(49.2)*

^:Children < 16 years old *:percentage; #:Quartiles (Q1andQ3)

DDD dense deposit disease; C3 GN C3 glomerulonephritis;

- At the time of diagnosis, 46.2% C3G patients had an eGFR<60 ml/min/1.73m², 47.1% presented serum complement C3 below normal, 48.8% presented low level of serum C4, 22.5% had abnormal blood/urine protein electrophoresis, and 3% had elevated serum globulin.
- Adult patients had significantly higher proportions of hypertension, severe proteinuria, and hematuria than children (Fig 2) . Adult patients had significantly higher creatinine values, 24-hour urine protein excretion, and lower glomerular filtration capacity than children (Fig 3) .

Fig 2. Clinical presentations

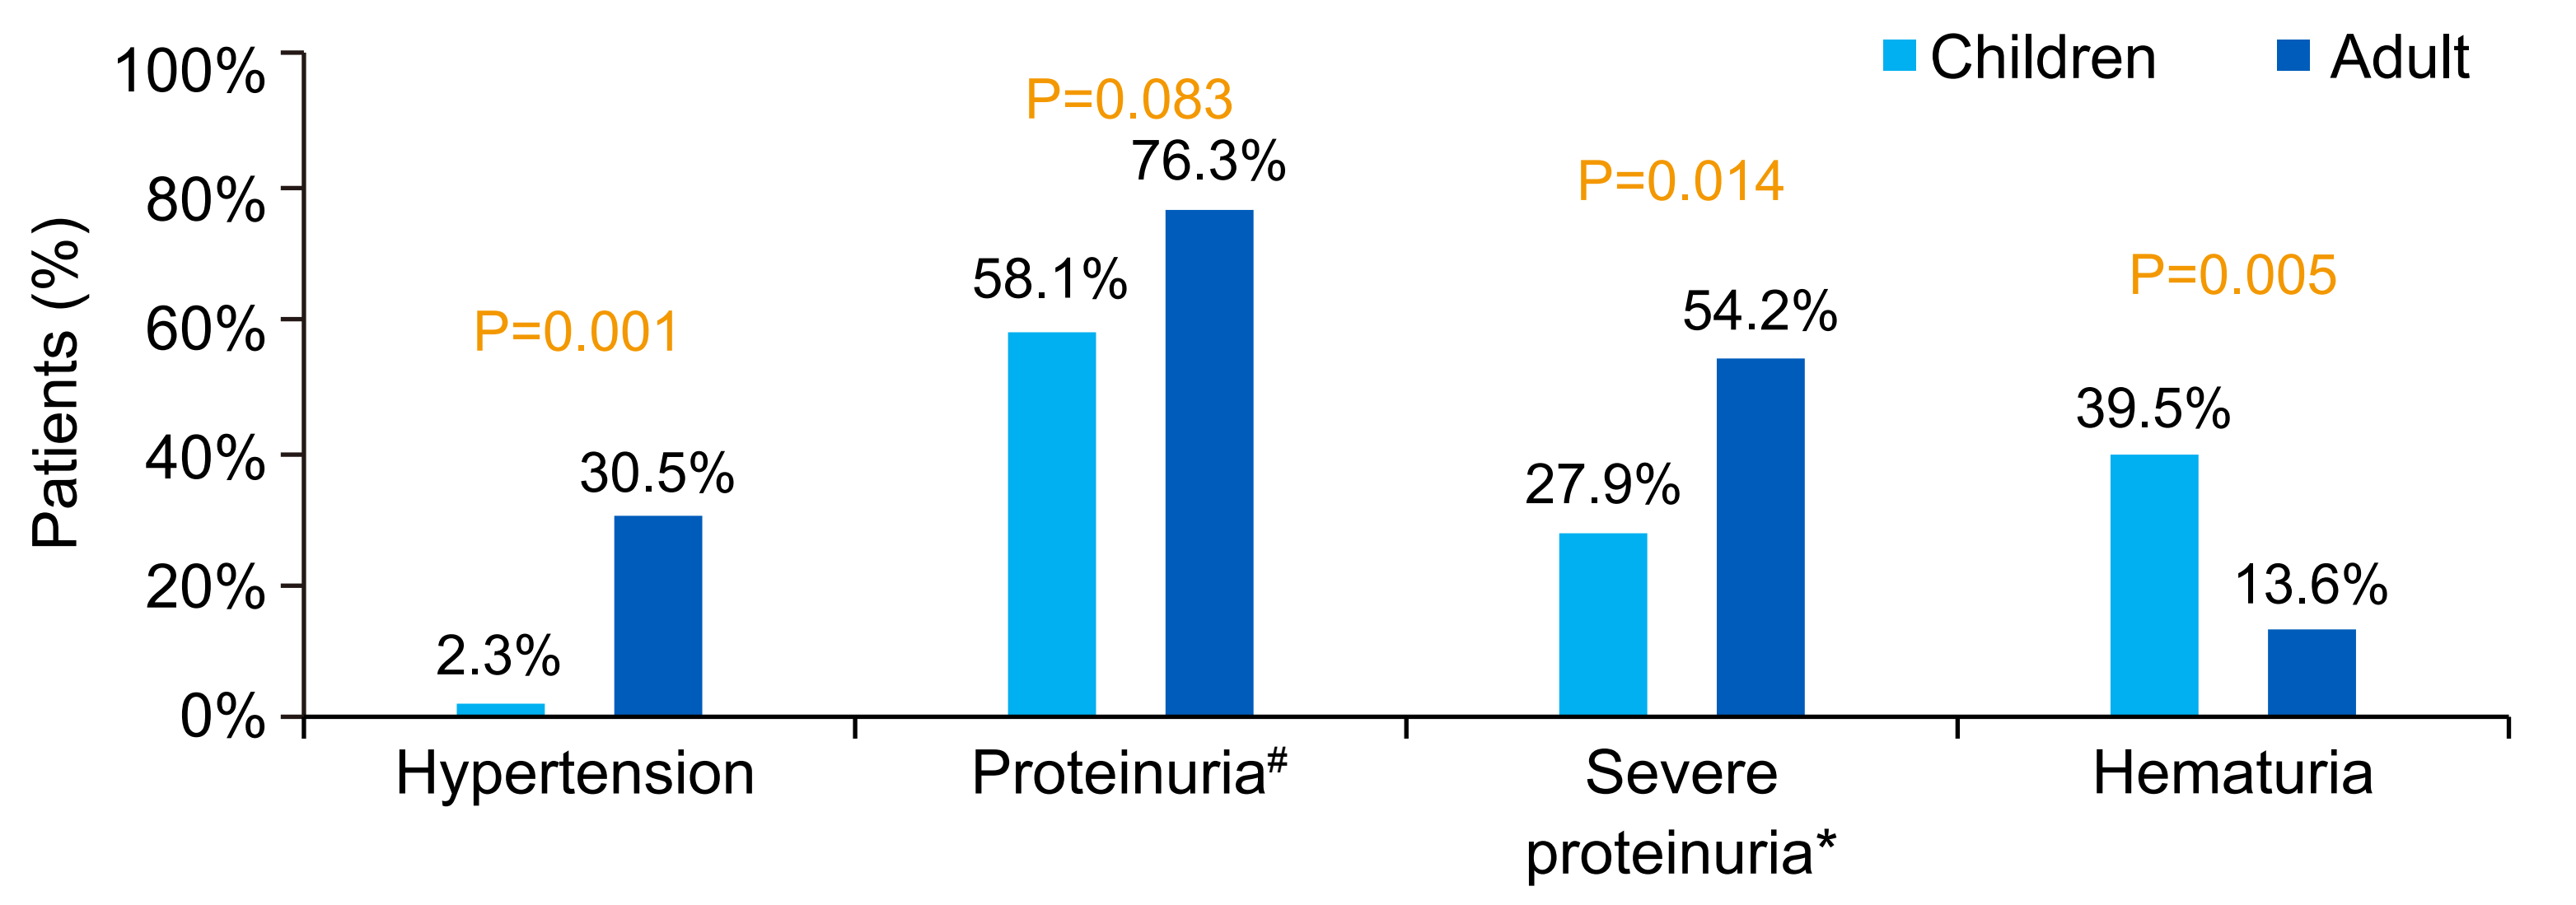
Children
Adult
Patients (%)
100%
80%
60%
40%
20%
0%
P=0.001
2.3%
30.5%
Hypertension
P=0.083
58.1%
76.3%
Proteinuria#
P=0.014
27.9%
54.2%
Severe proteinuria*
P=0.005
39.5%
13.6%
Hematuria

#Proteinuria: urinary protein qualitative≥1+ or UACR >30mg/g or 24-hour urinary protein qualitative >0.15g.
* Severe proteinuria defined as urinary protein qualitative ≥3+ or UACR >300mg/g or 24-hour urinary protein qualitative >3.5g

Fig 3. Laboratory tests

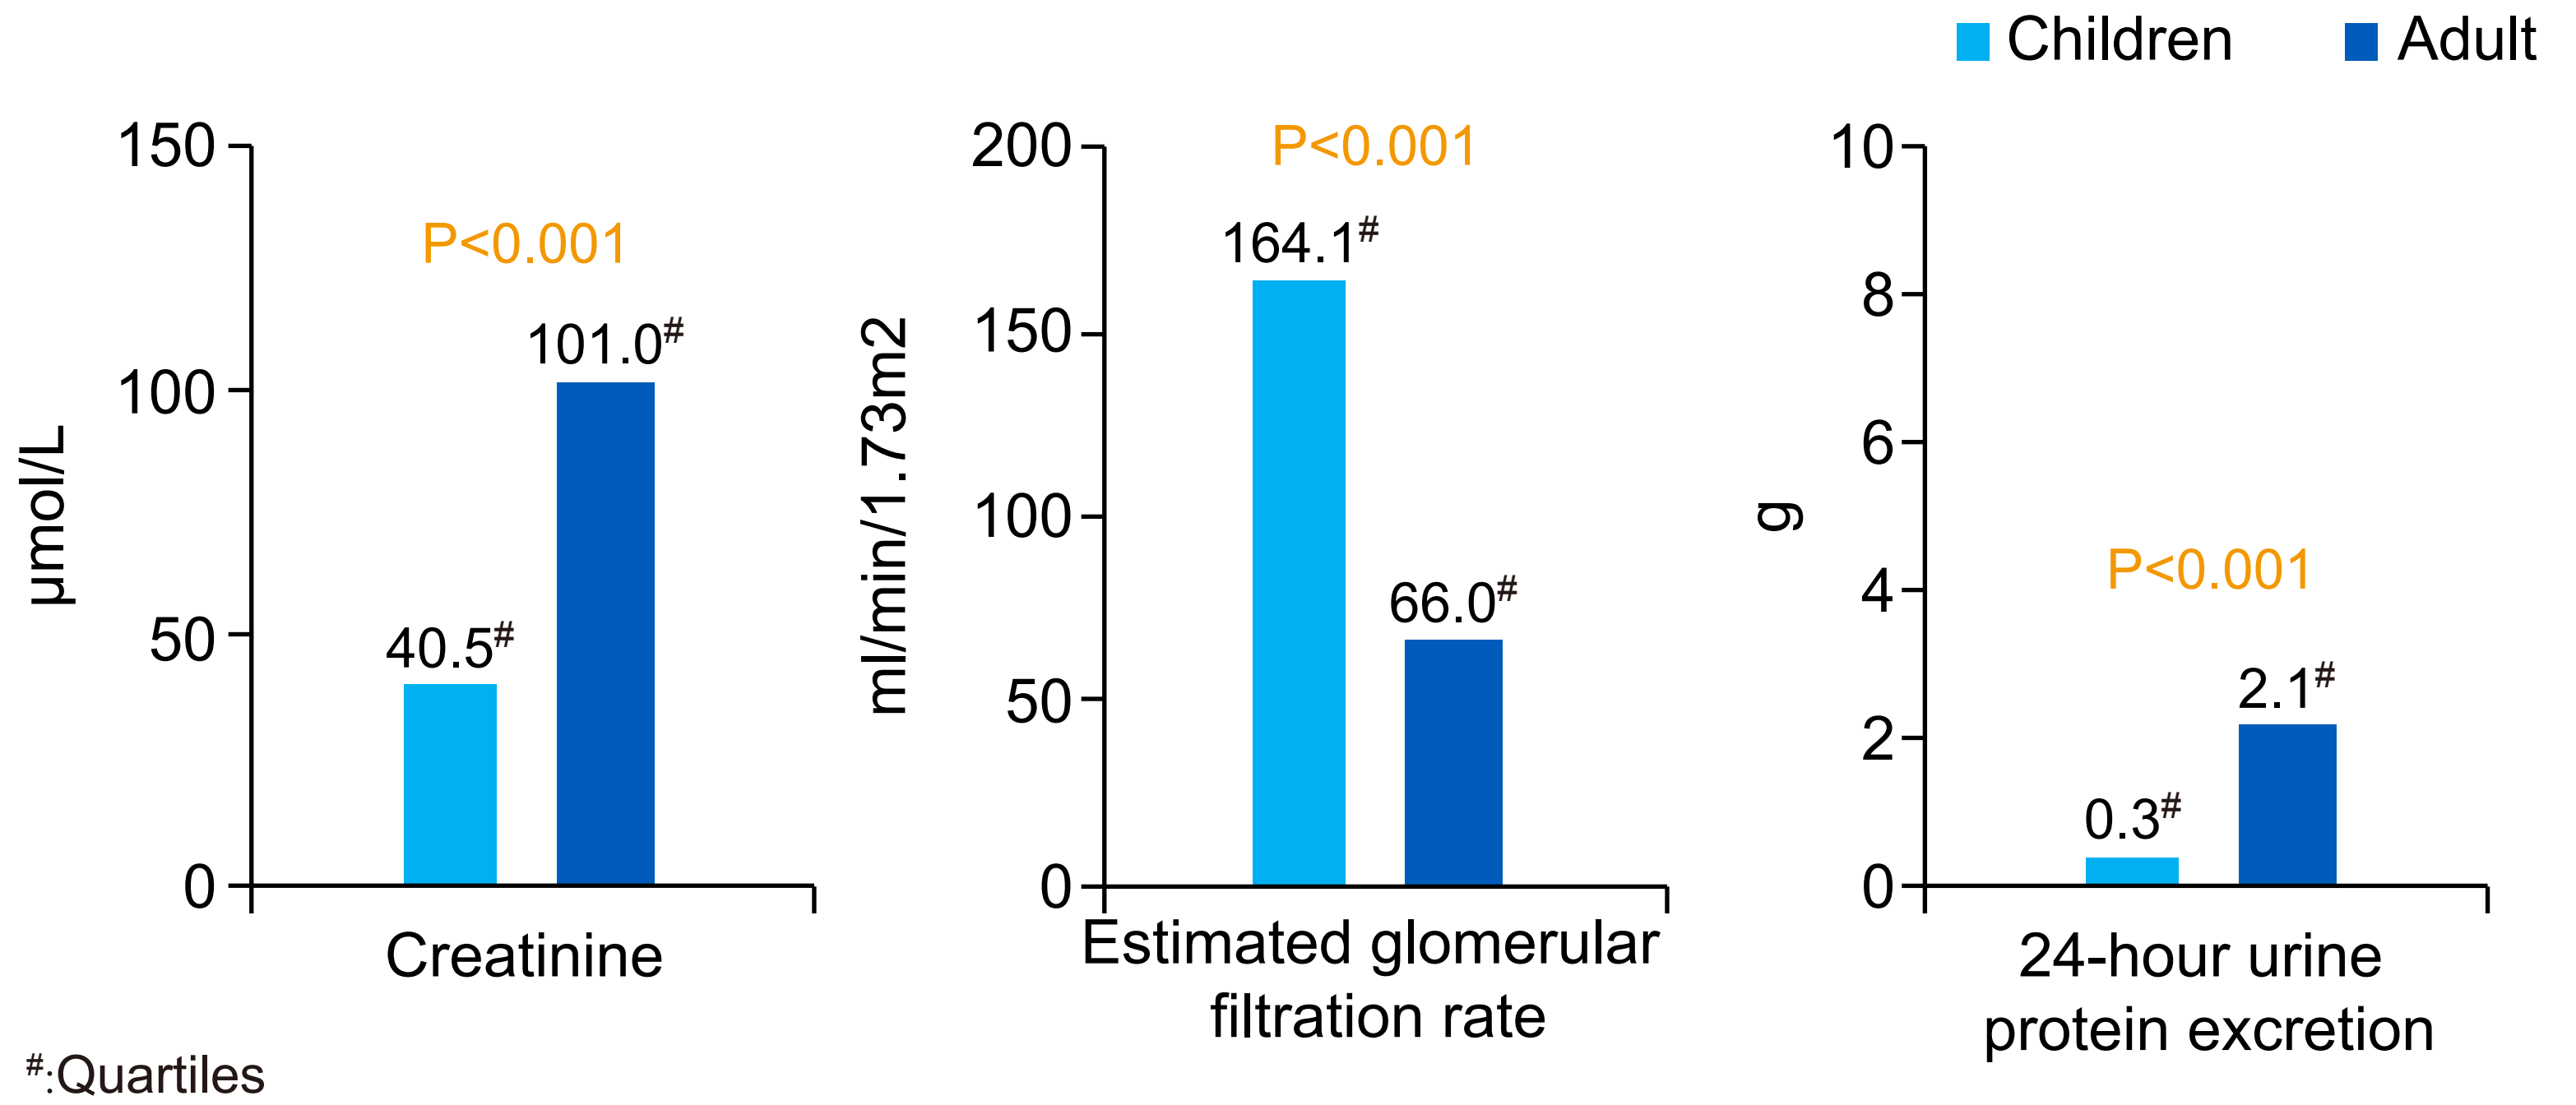
Children
Adult
μmol/L
150
100
50
0
P<0.001
40.5#
101.0#
Creatinine
ml/min/1.73m2
200
150
100
50
0
P<0.001
164.1#
66.0#
Estimated glomerular filtration rate
g
10
8
6
4
2
0
P<0.001
0.3#
2.1#
24-hour urine protein excretion

#:Quartiles

- As for treatment, the proportion of RASI use was 53.9%, while 47.1% of patients received glucocorticoids and half of them (23.5% of total) non-glucocorticoid immunosuppressants(including Cyclosporine, MMF, tacrolimus, etc.)(Fig 4).

Fig 4. Treatment

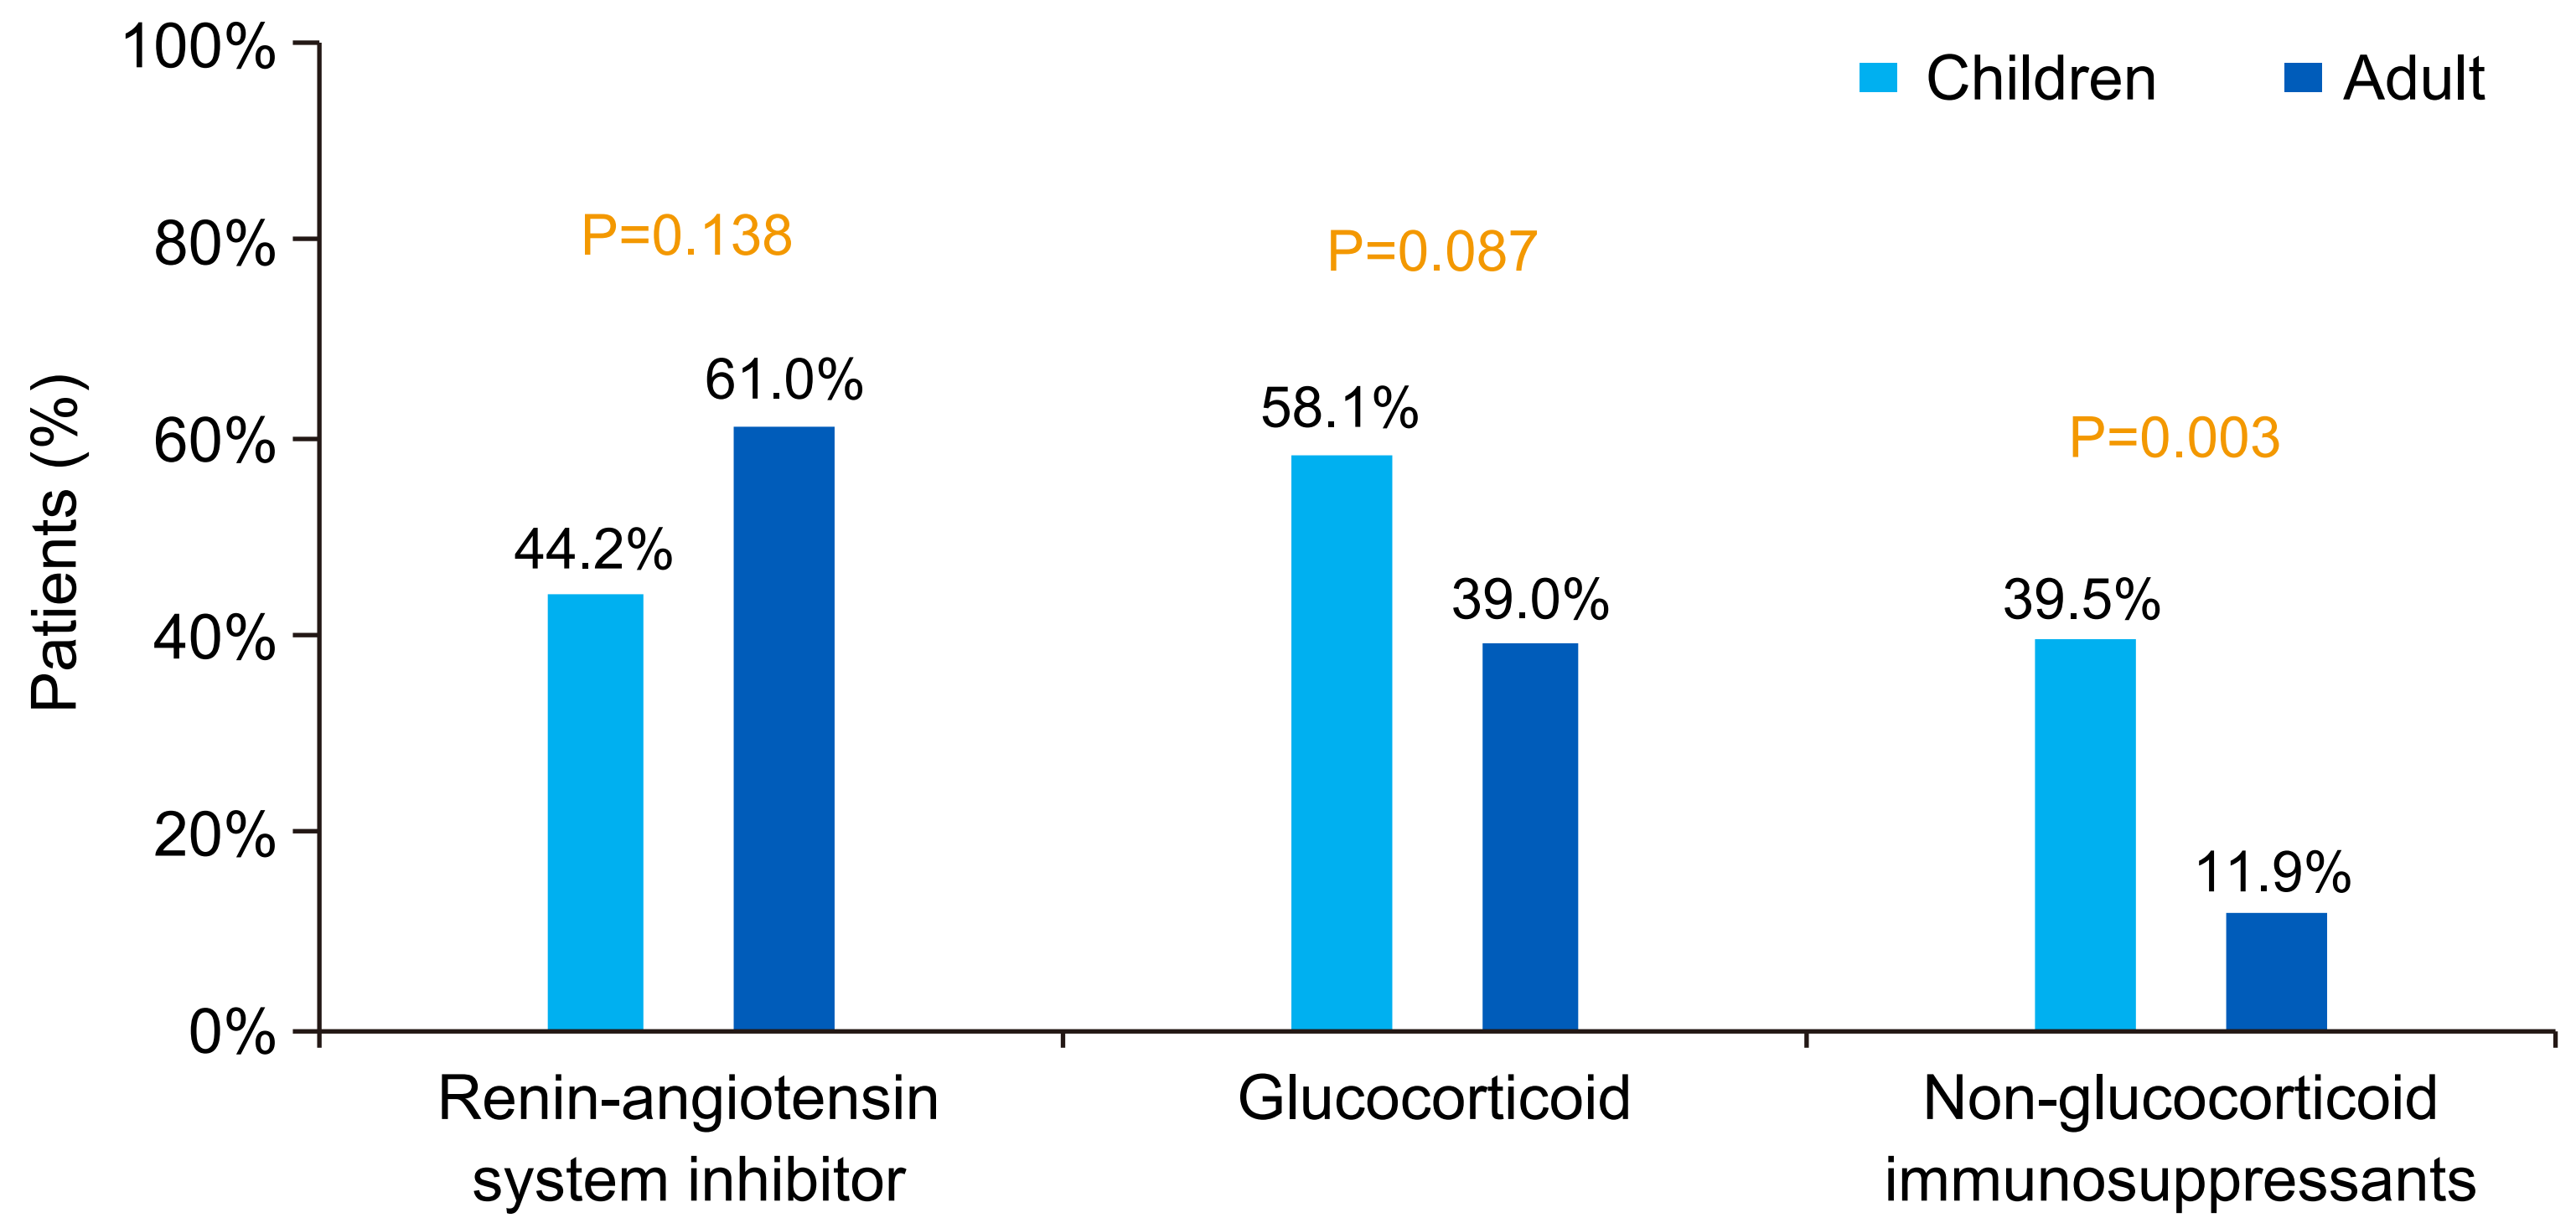
Children
Adult
Patients (%)
100%
80%
60%
40%
20%
0%
P=0.138
44.2%
61.0%
Renin-angiotensin system inhibitor
P=0.087
58.1%
39.0%
Glucocorticoid
P=0.003
39.5%
11.9%
Non-glucocorticoid immunosuppressants

- Sixteen patients were followed for more than one year. Among them, 12.5% had progression, as measured by a sustained > 40% decrease in eGFR from baseline, and 18.7% developed kidney failure within one year.

④Conclusion:

- C3G has heterogeneous clinical characteristics, and adult patients presented more serious symptoms at diagnosis. Unmet needs are identified for C3G patients standardized treatment in China, and targeted therapy is urgently needed.

⑤Disclosures:

- The authors have no disclosures or conflicts of interest to report.