Presenting author: Irene L. Noronha | irenenor@usp.br

Poster Board: SUN-051 Abstract No: WCN24-AB-1868

ASSIST: A randomized, double-blind, placebo-controlled crossover trial of atrasentan in patients with IgA nephropathy (IgAN) on SGLT2i

Irene L. Noronha,[1] Jonathan Barratt,[2] Jose Luis Gorriz,[3] Sradha Kotwal,[4] Soo Kun Lim,[5] Valeria Beckett,[6] Marianne Camargo,[6] Amy K. Mottl,[7] Hiddo J. L. Heerspink[8]

[1]Hospital Beneficência Portuguesa de São Paulo, São Paulo, Brazil; [2]University of Leicester, Leicester, United Kingdom; [3]Hospital Clínico Universitario, University of València, València, Spain; [4]The George Institute for Global Health, Newtown, Australia; [5]University of Malaya, Kuala Lumpur, Malaysia; [6]Chinook Therapeutics, A Novartis Company, Seattle, USA; [7]University of North Carolina at Chapel Hill, Chapel Hill, USA; [8]University Medical Center Groningen, Groningen, Netherlands

Scan to obtain:
- Poster

https://bit.ly/IKCWCN

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

SUMMARY

- Atrasentan, a potent and selective ETA antagonist, has potential to reduce proteinuria and preserve kidney function in IgAN
- The ASSIST crossover study will evaluate the safety and efficacy of atrasentan in combination with SGLT2i in patients with IgAN with persistent proteinuria despite maximized RASi

This study is sponsored by Chinook Therapeutics, a Novartis Company
Poster presented at the World Congress of Nephrology 2024, Buenos Aires, Argentina, April 13–16, 2024

INTRODUCTION

Glomerular disease and proteinuria

- IgA nephropathy (IgAN) is the leading cause of primary glomerulonephritis, with approximately 30–45% of patients with IgAN progressing to kidney failure over a period of 20–25 years[1,2]
- Proteinuria is the strongest predictor of disease progression in IgAN[1,3,4]
- Endothelin A (ETA) receptor activation may contribute to mesangial cell activation, proteinuria, kidney inflammation and fibrosis in IgAN[5] (**Figure 1**)

Figure 1. Mechanisms of ETA receptor mediated kidney injury in IgAN[5]

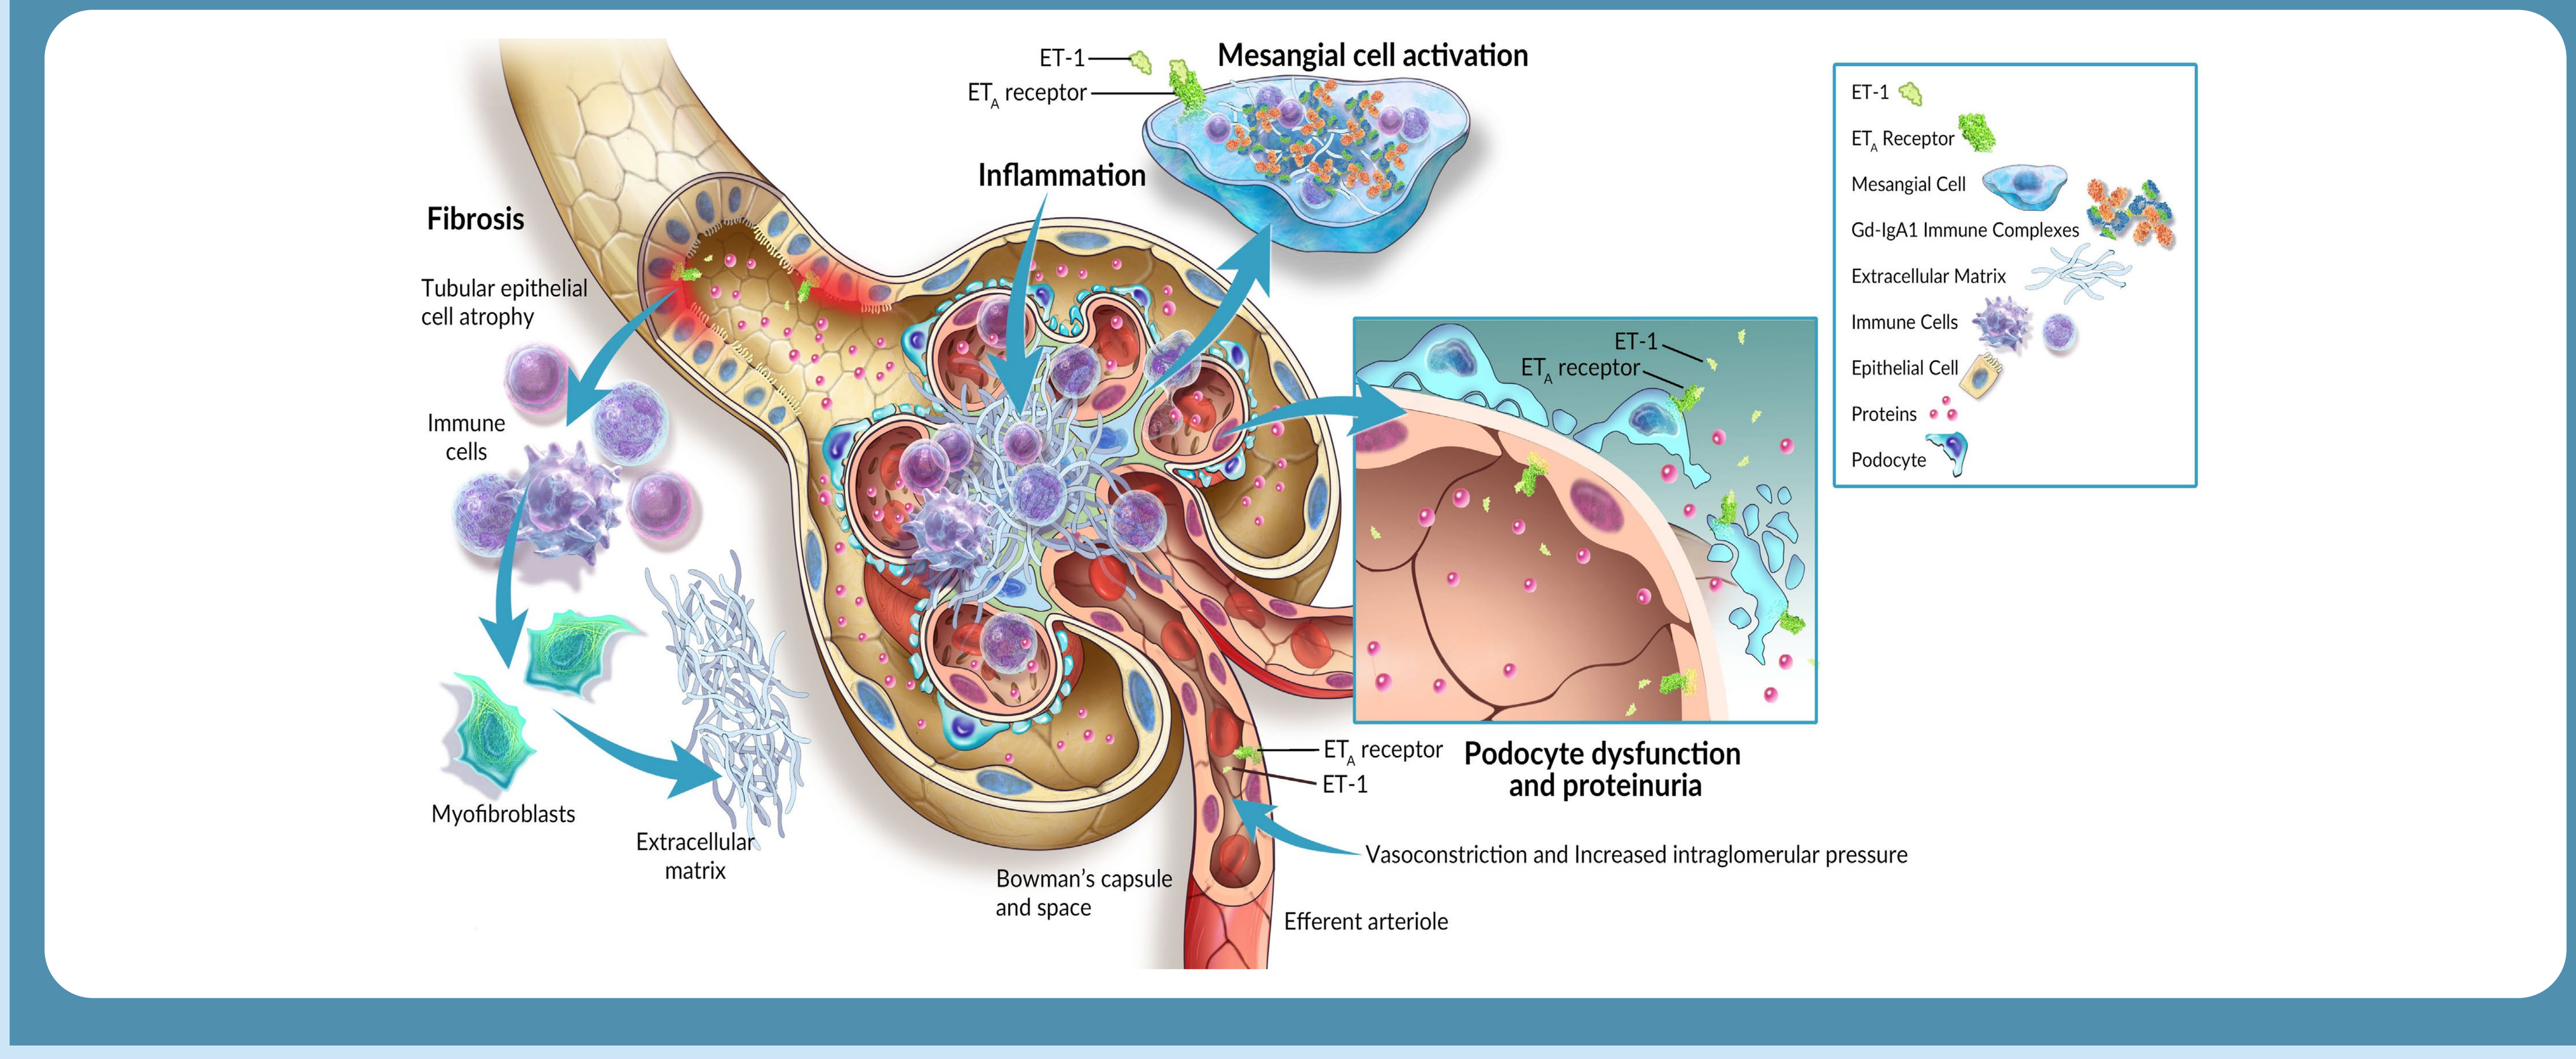
ET-1
ET_A receptor
Mesangial cell activation
Inflammation
Fibrosis
Tubular epithelial cell atrophy
Immune cells
Myofibroblasts
Extracellular matrix
Bowman's capsule and space
Efferent arteriole
ET_A receptor
ET-1
Podocyte dysfunction and proteinuria
Vasoconstriction and Increased intraglomerular pressure
ET-1
ET_A Receptor
Mesangial Cell
Gd-IgA1 Immune Complexes
Extracellular Matrix
Immune Cells
Epithelial Cell
Proteins
Podocyte

Atrasentan and SGLT2i

- Atrasentan is a potent and selective ETA antagonist with the potential to reduce proteinuria and preserve kidney function in IgAN[6]
 - Interim results of a Phase 2, open-label study in patients with IgAN (AFFINITY, NCT04573920) demonstrated that atrasentan was well tolerated and resulted in clinically meaningful and sustained proteinuria reductions in patients receiving a maximally tolerated and optimized dose of a renin-angiotensin system inhibitor (RASi)[6]
- Sodium glucose cotransporter-2 inhibitors (SGLT2i) are approved for use in adults with chronic kidney disease (CKD) at risk of disease progression[7,8]
 - In a post-hoc analysis of the global Phase 3 SONAR study in patients with type 2 diabetes and CKD, 6-week treatment with atrasentan and SGLT2i in a small number of patients (n=14) further decreased albuminuria and decreased body weight, a surrogate for fluid retention, compared with atrasentan alone[9]

ASSIST STUDY

Study objective

- ASSIST™ (NCT05834738) is a randomized, double-blind, placebo-controlled, crossover study to evaluate the safety and efficacy of atrasentan vs. placebo in adults with IgAN on stable SGLT2i and RASi with persistent proteinuria (**Figure 2**)
- Approximately 52 patients will be enrolled

Figure 2. Study design

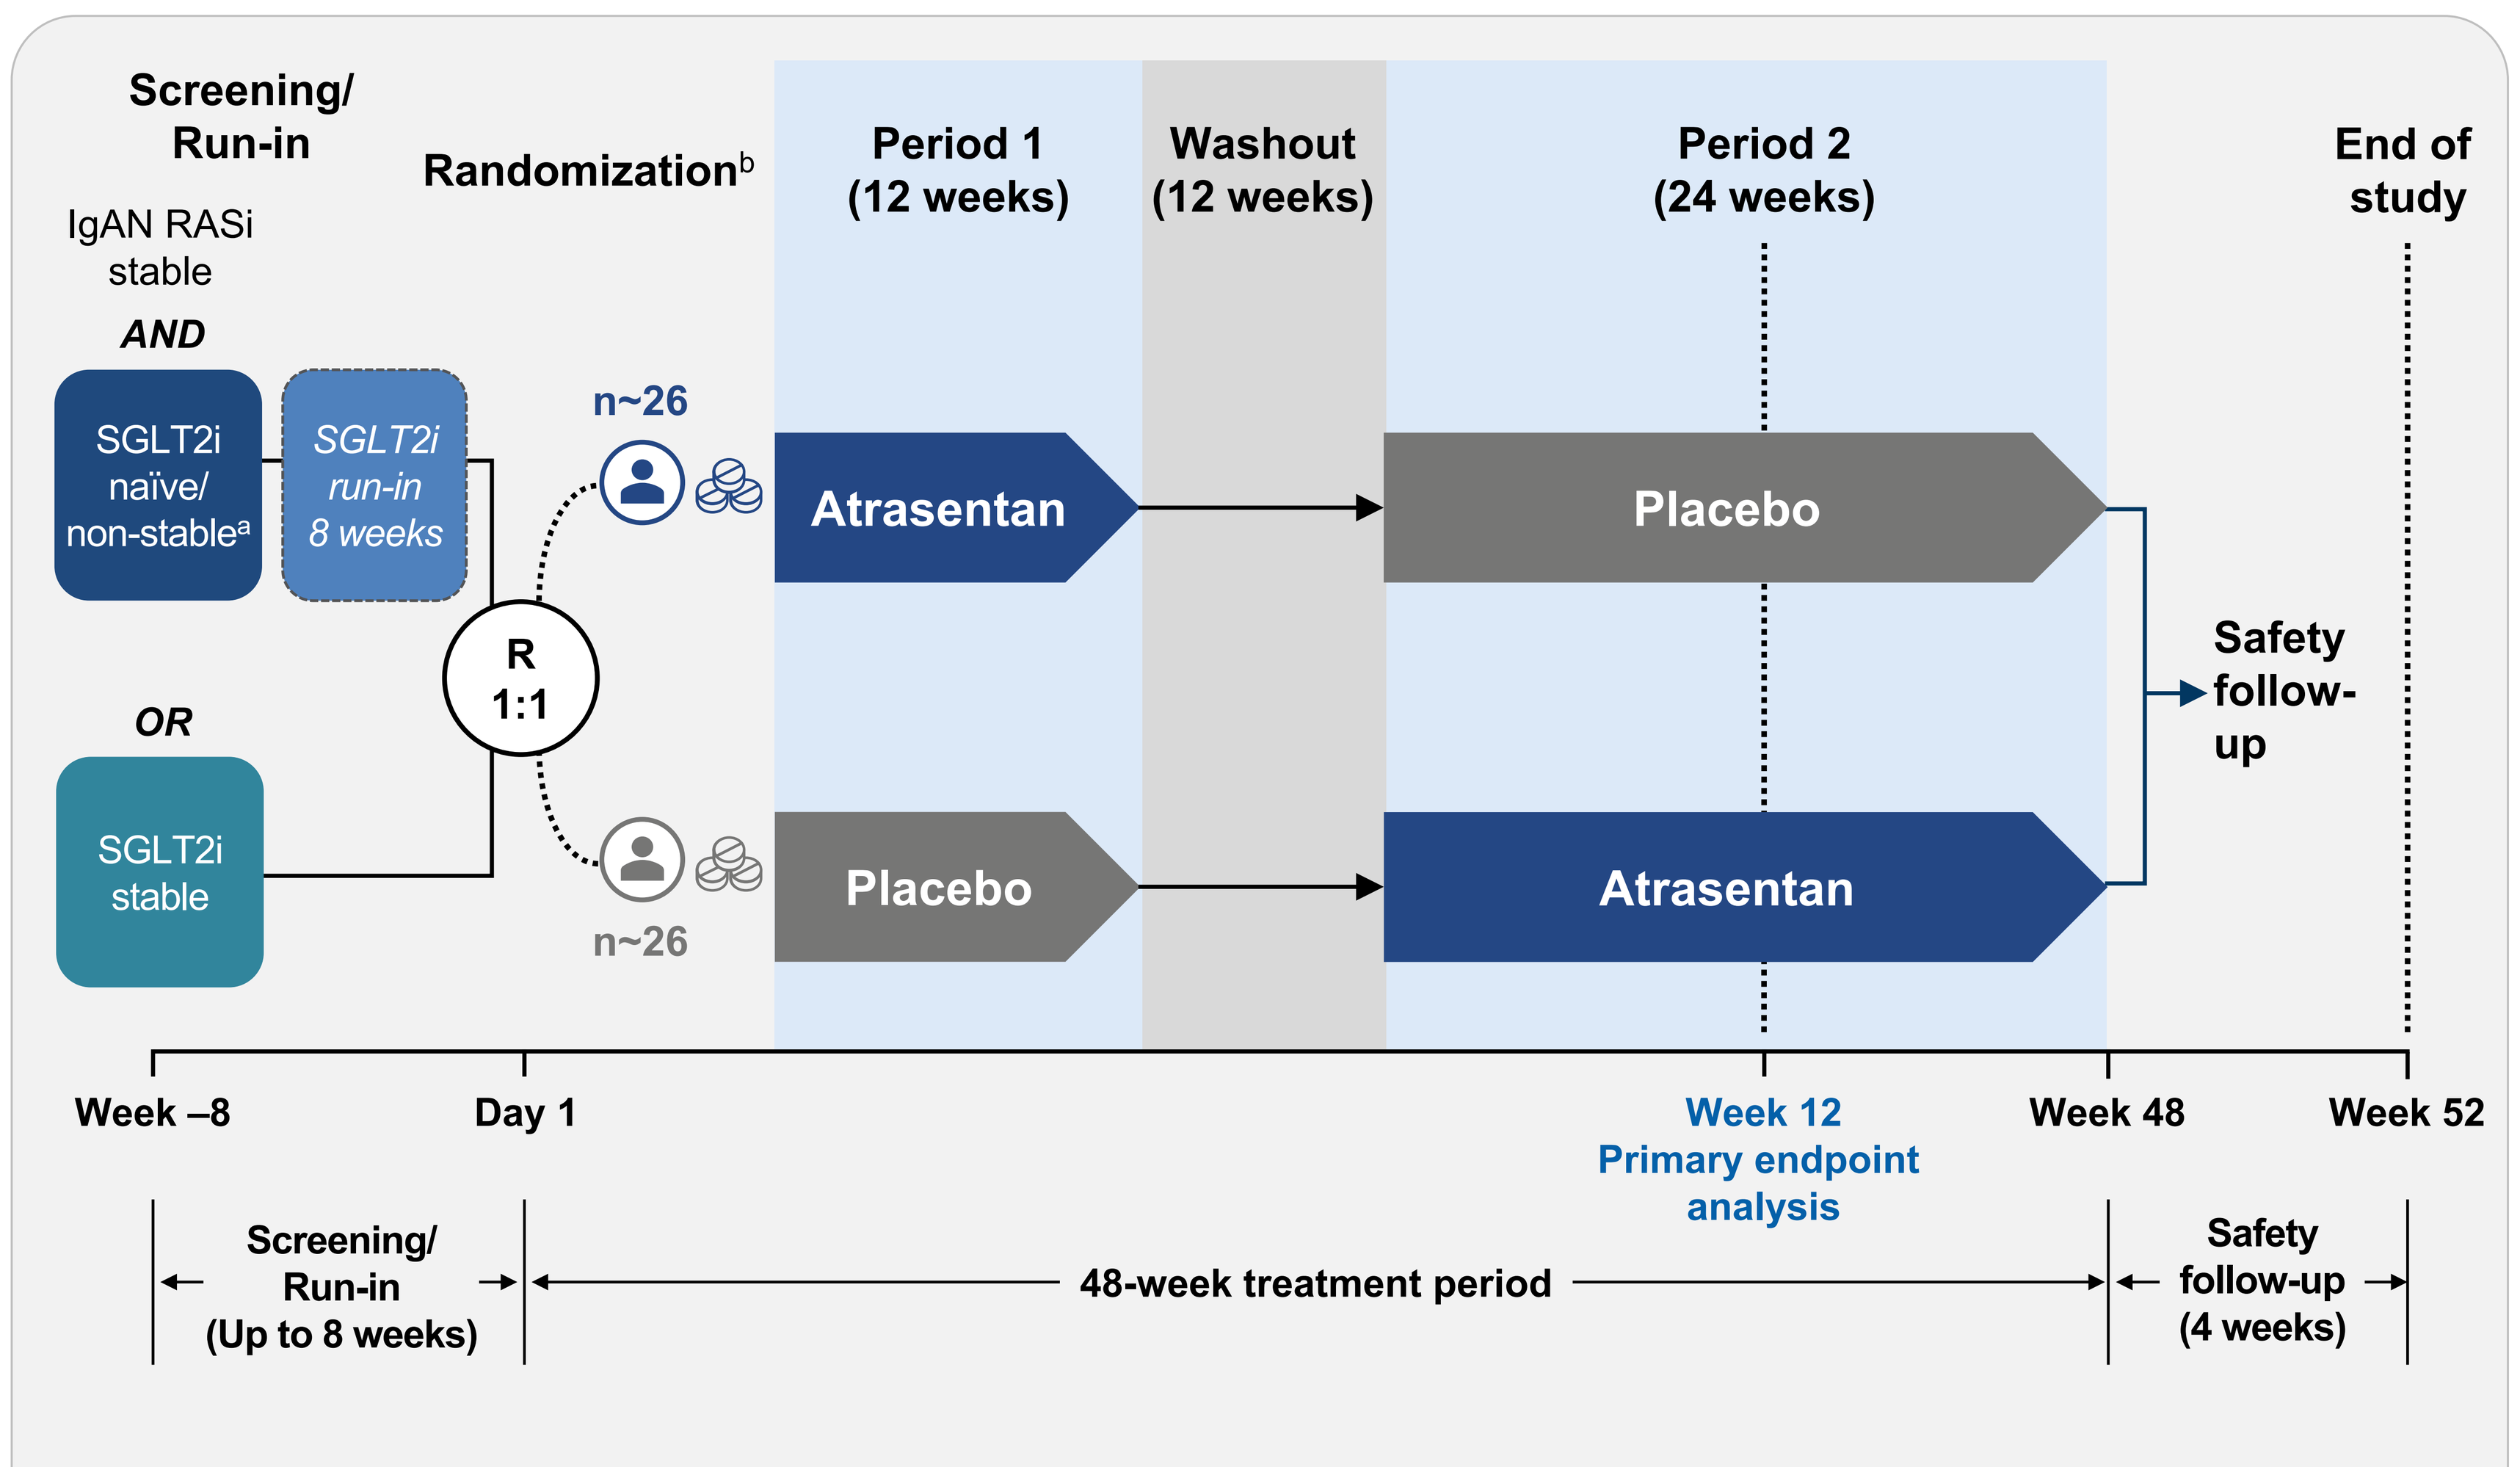
Screening/ Run-in
Randomization[b]
Period 1 (12 weeks)
Washout (12 weeks)
Period 2 (24 weeks)
End of study
IgAN RASi stable
AND
SGLT2i naïve/ non-stable[a]
SGLT2i run-in 8 weeks
OR
SGLT2i stable
R 1:1
n~26
n~26
Atrasentan
Placebo
Placebo
Atrasentan
Safety follow-up
Week –8
Day 1
Week 12 Primary endpoint analysis
Week 48
Week 52
Screening/ Run-in (Up to 8 weeks)
48-week treatment period
Safety follow-up (4 weeks)

[a]Patients who have not been on a stable dose of SGLT2i prior to study entry are required to complete the 8-week run-in period; [b]Stratified by SGLT2i status (stable vs run-in).

ASSIST™

The ASSIST study is currently enrolling
For more information, scan QR or visit
https://clinicaltrials.gov/ct2/show/NCT05834738

Study endpoints

Primary: Change in proteinuria (UPCR from a 24-hr urine collection) from baseline to Week 12

Key secondary: In Treatment Period 2, the change in proteinuria (UPCR from a 24-hr urine collection) from baseline to Week 24

Safety: Type, incidence, severity, and relatedness of adverse events (AEs) and serious AEs

Exploratory: In Treatment Period 2, change in eGFR from baseline to Week 24

Key inclusion criteria

Table 1. Key inclusion criteria

Patient group	
All patients	✓ Adults with biopsy-proven IgAN, not due to secondary causes
	✓ Receiving maximum tolerated and stable RASi ≥12 weeks prior to screening
	✓ eGFR ≥30 mL/min/1.73 m^2 (CKD-EPI equation) at screening
SGLT2i stable	✓ Receiving SGLT2i at stable dose ≥8 weeks prior to screening
	✓ 24-hour total urine protein >0.5 g/d at screening
SGLT2i naïve or non-stable	✓ 24-hour total urine protein >0.85 g/d at screening
	✓ Complete 8-week run-in period on a stable and well tolerated dose of an SGLT2i
	✓ After run-in period: 24-hour total urine protein >0.5 g/d confirmed at end of run-in; eGFR of ≥30 mL/min/1.73 m^2 (CKD-EPI equation) at end of run in

Disclosures

Atrasentan is an investigational drug that has not been approved by regulatory authorities. Efficacy and safety have not been established. There is no guarantee that it will become commercially available for the use(s) under investigation.

References

1. Reich HN, et al. *J Am Soc Nephrol*. 2007;18:3177–83;
2. Selvaskandan H, et al. *Kidney Int Rep*. 2024;9:203–213;
3. Thompson A, et al. *CJASN*. 2019;14(3): 469–481;
4. KDIGO. *Kidney Int*. 2021;100:S1–S276;
5. Kohan DE, et al. *Kidney Int Rep*. 2023;8:2198–2210;
6. Rastogi A, et al. ASN Kidney Week 2022; Poster TH-PO497;
7. Wheeler DC, et al. *Kidney Int*. 2021;100:215–24;
8. Jafar TH. *Lancet*. 2021:398(10297):283–84;
9. Heerspink HJL, et al. *Kidney Int*. 2021;99:346–49.