WCN23-0373

Alternative complement pathway pharmacodynamics of iptacopan

Robert Schmouder[1], Guido Junge[2], Julie Milojevic[2]*, Prasanna Kumar Nidamarthy[3], and Kenneth Kulmatycki[4]

[1]Novartis Pharmaceuticals Corporation, East Hanover, New Jersey, USA; [2]Novartis Pharma AG, Basel, Switzerland; [3]Novartis Healthcare Pvt. Ltd., Hyderabad, India; [4]Novartis Institutes of BioMedical Research, Cambridge, Massachusetts, USA

*Affiliation at the time of analysis

Conclusions

- Iptacopan (25–200 mg BID) demonstrated a rapid, selective, and sustained inhibition of the AP in a dose-dependent manner
- Participants who received iptacopan 200 mg BID showed the greatest (>80%) inhibition of the AP
- Consistent with its mechanism of action,[1] iptacopan administration was not associated with changes in CP activity
- Throughout the administration period, levels of plasma Bb (a fragment of factor B) were decreased in all iptacopan groups vs placebo, without apparent dose dependency
- Iptacopan was well tolerated in healthy participants with no serious AEs or AEs leading to study drug discontinuation
- The results of this study support the clinical development of iptacopan and provide confidence of durable AP inhibition in patients treated with iptacopan

Introduction

- Iptacopan (LNP023) is an oral, first-in-class, highly potent, selective inhibitor of factor B (FB), a key component of the alternative complement pathway (AP)[1]
- Phase 3 studies are currently ongoing to investigate the efficacy and safety of iptacopan in patients with paroxysmal nocturnal hemoglobinuria, C3 glomerulopathy, IgA nephropathy, atypical hemolytic uremic syndrome, and immune complex-mediated membranoproliferative glomerulonephritis[2–6]
- Here we report the PD of iptacopan, including inhibition of AP activity, in healthy participants as part of a Phase 1 study

Objectives

- To evaluate the PD and AP inhibitory activity of MAD of iptacopan using the Wieslab® ex vivo AP activity assay and circulating levels of Bb and sC5b-9
- To evaluate the safety and tolerability of MAD of iptacopan in healthy participants

Methods

Study design

- A randomized, first-in-human, Phase 1 study; eligible participants included healthy males and females (of non-childbearing potential) aged 18–55 years
 - The study consisted of three parts: a SAD study, a MAD study, and a food-effect evaluation study
- The MAD phase was a participant-blinded, placebo-controlled study; eligible participants were randomized in a 3:1 ratio to receive iptacopan (25 mg, 50 mg, 100 mg, or 200 mg) or matched placebo over a 14-day administration period (**Figure 1**)
 - On Days 1–13, participants received BID dosing; on Day 14, participants received only a single dose
 - A 5-day follow-up period included three follow-up evaluations and an EoS visit

Figure 1. Study design – MAD phase

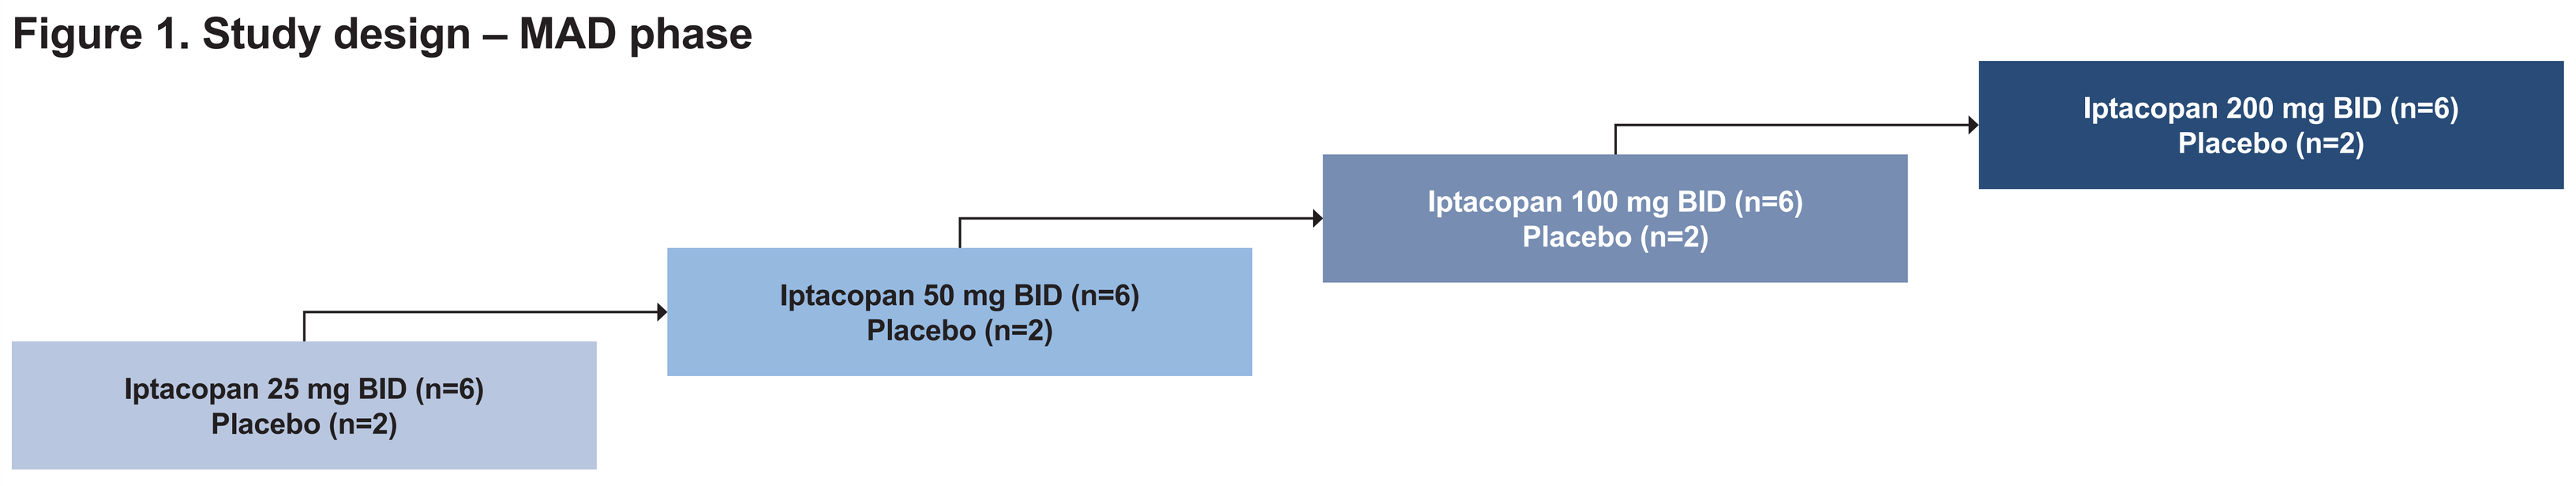

Iptacopan 200 mg BID (n=6)
Placebo (n=2)
Iptacopan 100 mg BID (n=6)
Placebo (n=2)
Iptacopan 50 mg BID (n=6)
Placebo (n=2)
Iptacopan 25 mg BID (n=6)
Placebo (n=2)

On Days 1–13, participants received BID dosing of iptacopan or matched placebo; on Day 14, participants received only a single dose.
BID, twice daily; MAD, multiple-ascending dose.

PD and safety assessments

- AP inhibition was measured using a Wieslab® assay,[7] which was validated for the quantitative assessment of functional AP activity after ex vivo pathway activation in participants' serum
 - AP activity was quantified at 13 timepoints: baseline; –1, 2, and 12 h (Day 1); 24 h (Day 2); pre-morning dose on Days 3, 6, 10, and 12; –1, 2, and 12 h on Day 14; and 24 h post-final dose (Day 15)
- Classical complement pathway (CP) activity was measured using a CH50 assay, validated for the quantitative assessment of CP activity after ex vivo pathway activation in participants' serum
 - CP activity was quantified at six time points: baseline; –1, 2, and 12 h (Day 1); 24 h (Day 2); and 24 h post-final dose (Day 15)
- Plasma concentrations of soluble complement biomarkers, Bb (a fragment of FB) and sC5b-9, were measured at 12 timepoints: baseline; −1, 2, and 12 h (Day 1); 24 h (Day 2); pre-morning dose on Days 3, 6, 10, and 12; −1 and 2 h on Day 14; and 24 h post-final dose (Day 15)
- Safety assessments included reporting of AEs, physical examinations, vital signs, ECG, and clinical laboratory evaluations

Statistical analysis

- Sample size was derived based on a ≥80% probability for an AE with an underlying occurrence rate of ≥24% at least once for the given doses of iptacopan
- The PD analysis set included all participants with available PD data; the safety analysis set included all participants that received any study drug
- Descriptive statistical analysis was performed using SAS version 9.4 (SAS Institute Inc., Cary, North Carolina, USA)

Results

- 32 participants enrolled in the study; all participants completed the study
- Demographic data were similar across the iptacopan dose groups (**Table 1**)

Table 1. Demographics

Characteristic	Iptacopan 25 mg BID (n=6)	Iptacopan 50 mg BID (n=6)	Iptacopan 100 mg BID (n=6)	Iptacopan 200 mg BID (n=6)	Pooled placebo (n=8)	Total (N=32)
Age, years	46.3 (9.2)	38.0 (12.7)	42.3 (12.4)	35.7 (12.4)	45.5 (9.6)	41.8 (11.3)
Sex, n (%)						
Female	1 (17)	0	0	1 (17)	0	2 (6)
Male	5 (83)	6 (100)	6 (100)	5 (83)	8 (100)	30 (94)
Race, n (%)						
White	6 (100)	6 (100)	6 (100)	6 (100)	8 (100)	32 (100)
Not Hispanic or Latino, n (%)	5 (83)	6 (100)	6 (100)	6 (100)	8 (100)	31 (97)
Weight, kg	76.5 (9.3)	78.1 (3.5)	86.1 (13.2)	74.9 (9.4)	85.9 (5.1)	80.6 (9.4)
Height, cm	180.3 (7.0)	179.2 (5.6)	181.0 (10.4)	170.0 (9.1)	181.6 (4.6)	178.6 (8.2)
BMI, kg/m²	23.5 (1.7)	24.4 (2.4)	26.3 (3.3)	25.9 (2.3)	26.0 (1.3)	25.3 (2.4)

Data are mean (SD) unless stated otherwise.
BID, twice daily; BMI, body mass index; SD, standard deviation.

Analysis of PD

AP activity

- Administration of iptacopan at all dose levels led to a rapid and near-complete inhibition (>87%) of AP activity 2 h after the first dose (**Figure 2A**)
- Over the administration period, iptacopan generally led to sustained inhibition (>60%) of AP activity; duration and magnitude of inhibition were dose-dependent
- The iptacopan 200 mg BID dose demonstrated greatest inhibition (>80%) of the AP
 - After treatment cessation, participants administered the 200 mg BID dose showed the longest persistence of AP inhibition
- Minimal changes in AP activity were observed in participants receiving placebo

CP activity

- No consistent magnitude of change in CP activity was observed following the administration of iptacopan 100 mg BID, 200 mg BID, or placebo (**Figure 2B**)

Figure 2. Mean percentage change* in complement activity†

A. AP activity (Wieslab® assay) **B. CP activity (CH50 assay)**

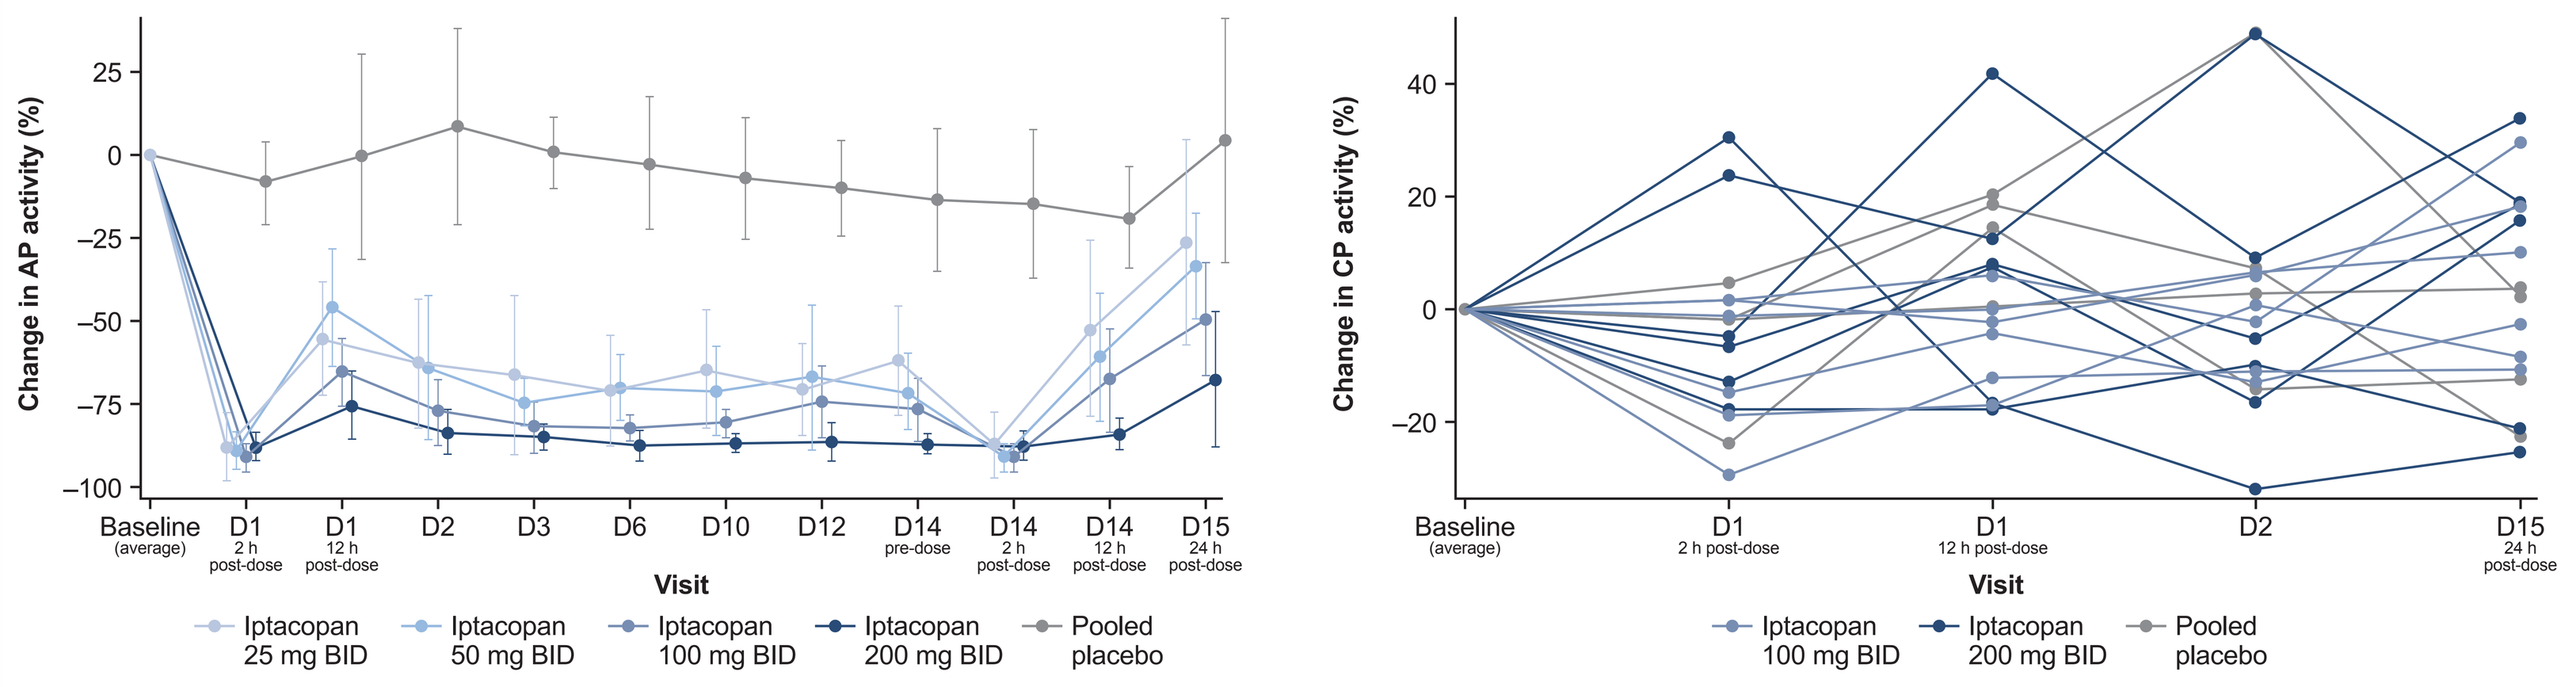

Change in AP activity (%)
Baseline (average)
D1 2 h post-dose
D1 12 h post-dose
D2
D3
D6
D10
D12
D14 pre-dose
D14 2 h post-dose
D14 12 h post-dose
D15 24 h post-dose
Visit
Iptacopan 25 mg BID
Iptacopan 50 mg BID
Iptacopan 100 mg BID
Iptacopan 200 mg BID
Pooled placebo
Change in CP activity (%)
Baseline (average)
D1 2 h post-dose
D1 12 h post-dose
D2
D15 24 h post-dose
Visit
Iptacopan 100 mg BID
Iptacopan 200 mg BID
Pooled placebo

Data are (A) mean (SD) time profiles and (B) overlaying individual time profiles of percentage change in complement pathway activity.
*Defined as the percentage change from the mean of baseline (Day –2 to –1) and Day 1 pre-dose values; †Values <LLOQ and >ULOQ were imputed as LLOQ/2 and ULOQ, respectively.
AP, alterative pathway; BID, twice daily; D, day; h, hour; LLOQ, lower limit of quantification; SD, standard deviation; ULOQ, upper limit of quantification.

Plasma biomarkers of complement activity

- Iptacopan administration led to a decrease from baseline in mean plasma Bb levels at 2 h post-first dose in all dose groups, which continued until 12 h post-first dose (**Figure 3A**)
- No consistent changes in mean plasma Bb levels from baseline were seen in participants receiving placebo (**Figure 3A**)
- Over the administration period, decreases in plasma sC5b-9 levels from baseline were observed at all iptacopan doses; however, sC5b-9 levels also decreased from baseline in the placebo group (**Figure 3B**)

Figure 3. Mean plasma Bb and plasma sC5b-9 concentrations

A. Plasma Bb **B. Plasma sC5b-9**

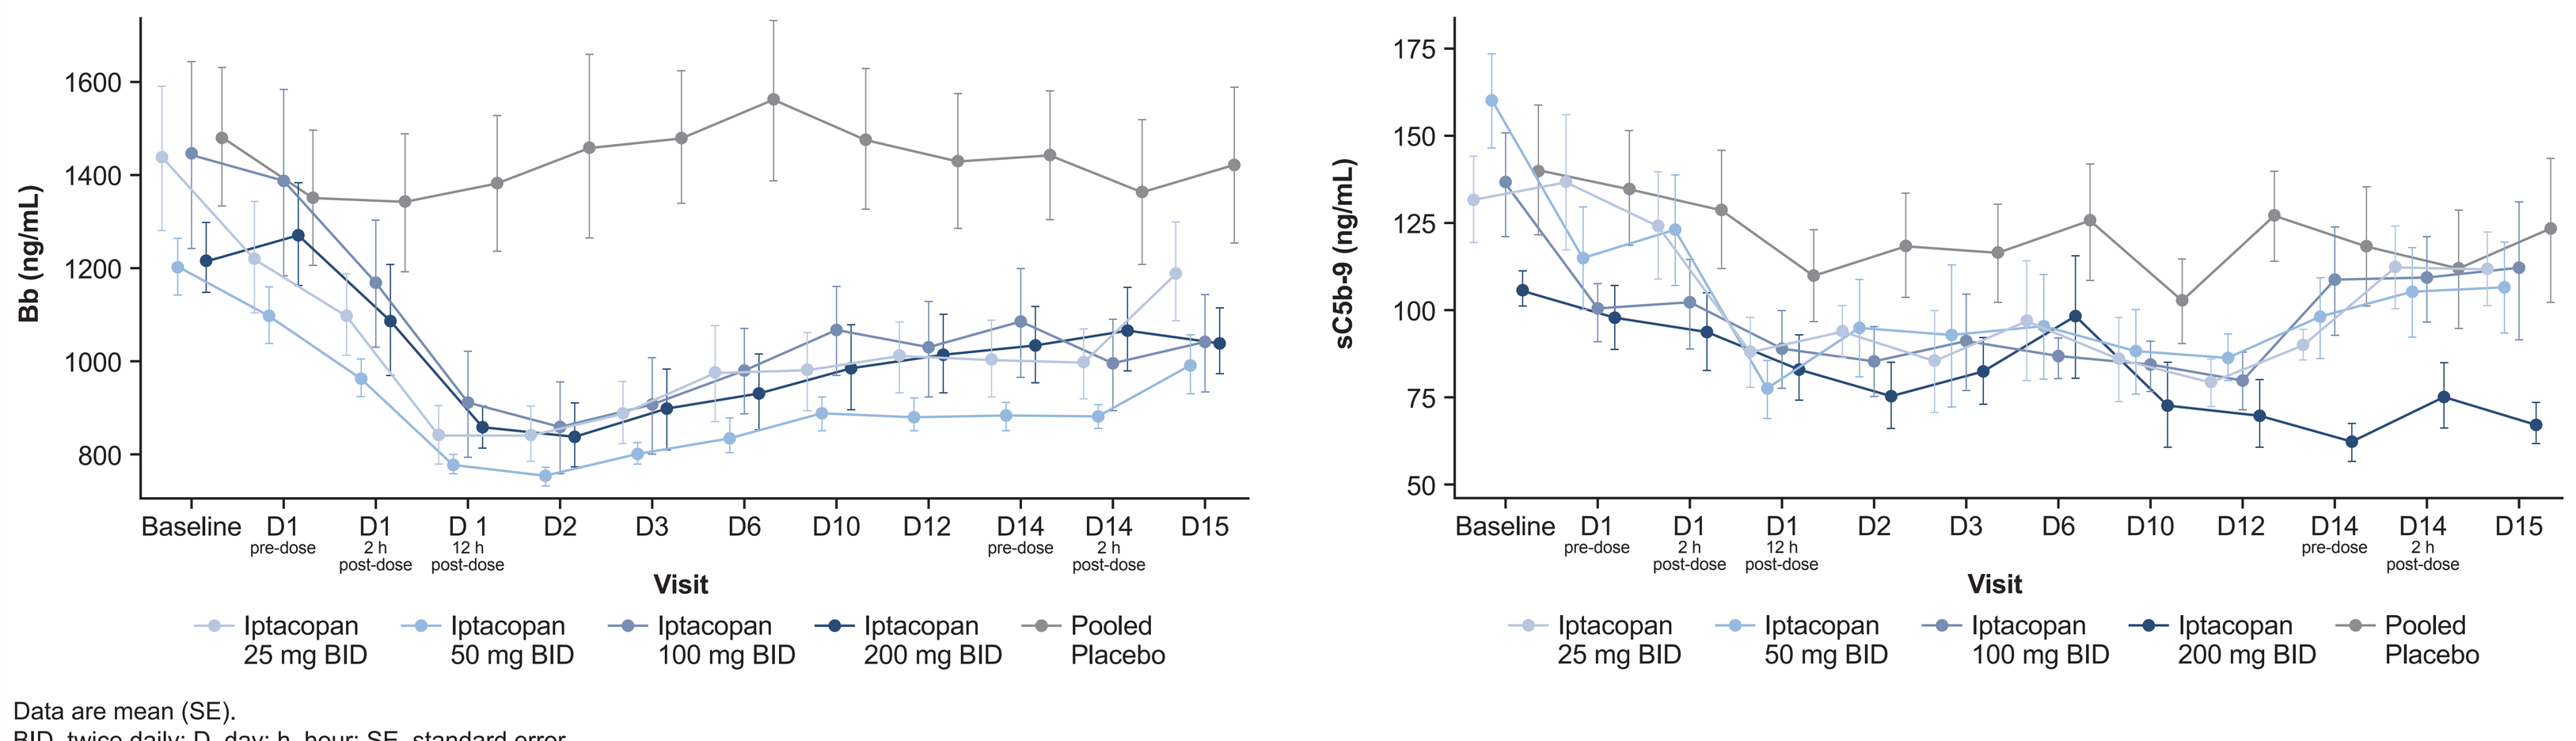

Bb (ng/mL)
sC5b-9 (ng/mL)
Baseline
D1 pre-dose
D1 2 h post-dose
D1 12 h post-dose
D2
D3
D6
D10
D12
D14 pre-dose
D14 2 h post-dose
D15
Visit
Iptacopan 25 mg BID
Iptacopan 50 mg BID
Iptacopan 100 mg BID
Iptacopan 200 mg BID
Pooled Placebo

Data are mean (SE).
BID, twice daily; D, day; h, hour; SE, standard error.

Safety assessment

- 20/32 participants (62.5%) reported 38 AEs; no serious AEs were reported, and no AEs led to discontinuation of study drug
- The iptacopan 50 mg BID and 100 mg BID dose groups showed the highest occurrence of AEs (83.3%), followed by the 200 mg BID (66.7%) and 25 mg BID (16.7%) dose groups
 - All AEs reported in the iptacopan dosing groups were of mild intensity
- Overall, headache (15.6%), medical device-site reaction (9.4%), back pain (6.3%), and sunburn (6.3%) were the most commonly reported AEs; all other AEs occurred in only one participant per AE
 - In the iptacopan dosing groups, headache and medical device-site reaction were the most commonly reported AEs, occurring in 4/24 (16.7%) and 2/24 (8.3%) of participants who received iptacopan, respectively

References

1. Schubart A et al. *Proc Natl Acad Sci U S A*. 2019;116:7926–31.
2. ClinicalTrials.gov. NCT04820530. Accessed: January 19, 2023.
3. ClinicalTrials.gov. NCT04817618. Accessed: January 19, 2023.
4. ClinicalTrials.gov. NCT04578834. Accessed: January 19, 2023.
5. ClinicalTrials.gov. NCT04889430. Accessed: January 19, 2023.
6. ClinicalTrials.gov. NCT05755386. Accessed: March 7, 2023.
7. Wieslab® Complement System Alternative Pathway. Instructions For Use. Svar Life Science AB; December 2018. Accessed February 24, 2023.

Abbreviations

AE, adverse event; AP, alternative complement pathway; BID, twice daily; C3, complement 3; CP, classical complement pathway; D, day; ECG, electrocardiogram; EoS, end-of-study; FB, factor B; h, hour; IgA, immunoglobulin A; LLOQ, lower limit of quantification; MAD, multiple-ascending doses; PD, pharmacodynamics; SAD, single-ascending dose; sC5b-9, soluble terminal complement complex C5b-9; SD, standard deviation; SE, standard error; ULOQ, upper limit of quantification.

Disclosures

RS is an employee of Novartis Pharmaceuticals Corporation, East Hanover, New Jersey, USA. **GJ** is an employee of Novartis Pharma AG, Basel, Switzerland. **JM*** is an employee of Novartis Institutes of BioMedical Research, Basel, Switzerland. **PKN** is an employee of Novartis Healthcare Pvt. Ltd., Hyderabad, India. **KK** is an employee of Novartis Institutes of BioMedical Research, Cambridge, Massachusetts, USA.

**Affiliation at the time of analysis.*

Funding source

This study was funded by Novartis Pharma AG, Basel, Switzerland.

Acknowledgments

Medical writing support and editorial support were provided by Ella Brooks (BOLDSCIENCE Ltd. UK) and were funded by Novartis Pharmaceuticals Corporation. This poster was developed in accordance with Good Publication Practice (GPP) guidelines. The authors had full control of the content and made the final decision on all aspects of this publication.
A copy of this poster will be available for download at the following URL: http://novartis.medicalcongressposters.com/Default.aspx?doc=3659f

Note: Downloading data may incur costs that can vary depending on your service provider and may be high if you are using your smartphone abroad. Please check your phone tariff or contact your service provider for more details.

Poster presented at: ISN World Congress of Nephrology 2023. Bangkok, Thailand. March 30–April 2, 2023.

Scan QR code to download this poster