WCN23-0405

An adaptive, dose-exploration, Phase 2 trial evaluating efficacy and safety of iptacopan in combination with standard-of-care with and without oral corticosteroids in active lupus nephritis

Udaykiran Veldandi[1], Hans-Joachim Anders[2], Tak Mao Chan[3], David Jayne[4], Piyush Mahajan[1], Janina Linnik[5], Nicholas J A Webb[5], Matthias Meier[5], Brad H. Rovin[6]

[1]Novartis Healthcare Pvt Ltd, Hyderabad, India; [2]University Hospital, LMU Munich, Munich, Germany; [3]School of Clinical Medicine, University of Hong Kong, Hong Kong; [4]University of Cambridge, Cambridge, United Kingdom; [5]Novartis Pharma AG, Basel, Switzerland; [6]The Ohio State University, Ohio, USA

Summary

- This adaptive proof-of-concept and dose-exploration Phase 2 study is designed to ascertain whether iptacopan can improve outcomes in patients with lupus nephritis while also allowing reduction (or elimination) of oral corticosteroids

Figure 1. Study design

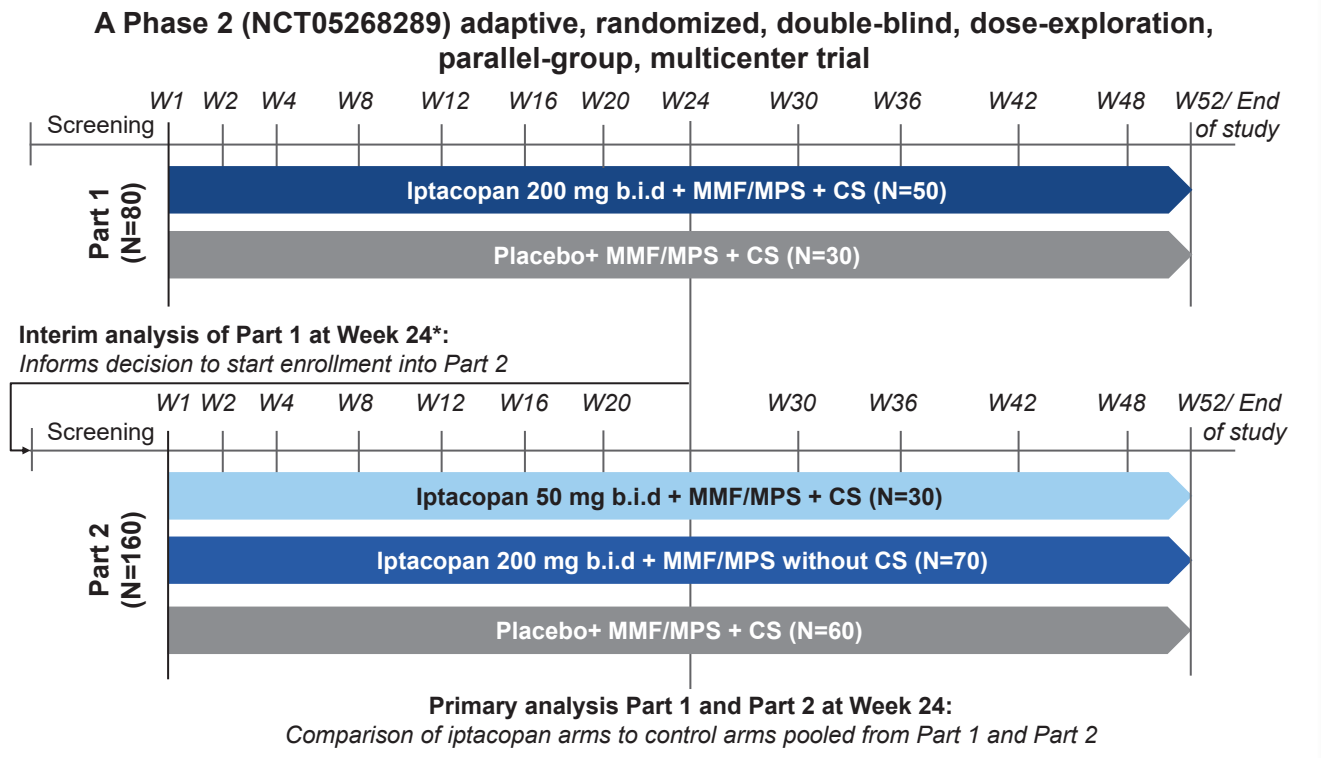

A Phase 2 (NCT05268289) adaptive, randomized, double-blind, dose-exploration, parallel-group, multicenter trial
Screening
W1 W2 W4 W8 W12 W16 W20 W24 W30 W36 W42 W48 W52/ End of study
Part 1 (N=80)
Iptacopan 200 mg b.i.d + MMF/MPS + CS (N=50)
Placebo+ MMF/MPS + CS (N=30)
Interim analysis of Part 1 at Week 24*:
Informs decision to start enrollment into Part 2
W1 W2 W4 W8 W12 W16 W20 W30 W36 W42 W48 W52/ End of study
Screening
Part 2 (N=160)
Iptacopan 50 mg b.i.d + MMF/MPS + CS (N=30)
Iptacopan 200 mg b.i.d + MMF/MPS without CS (N=70)
Placebo+ MMF/MPS + CS (N=60)
Primary analysis Part 1 and Part 2 at Week 24:
Comparison of iptacopan arms to control arms pooled from Part 1 and Part 2

* A pre-specified interim analysis (IA) will be performed when ~80 participants have completed Week 24 visit. The totality of efficacy and safety data up to the IA cut-off point and outcome of the prespecified IA will determine the start of Part 2

Figure 2. Study participants

Key Inclusion Criteria*

- Patients aged ≥18 years
- Positive ANA (titer ≥1:80) and/or presence of anti-double stranded DNA antibodies at screening
- Active biopsy-proven Class III–IV LN, with/without co-existing features of Class V, within 3 months of screening
- Documented active renal disease at the time of screening necessitating therapy with CS in combination with MMF/MPS
- eGFR ≥30 mL/min/1.73 m^2
- Vaccination against *Neisseria meningitidis*, *Streptococcus pneumoniae* and *Haemophilus influenzae*
- Supportive care including stable dose regimen of anti-malarials (e.g. hydroxychloroquine) unless contraindicated. Maximal or locally approved daily dose of ACEi or ARB at screening and should remain stable throughout the study
- First presentation or flare of LN

Key Exclusion Criteria*

- Induction treatment with cyclophosphamide or calcineurin inhibitors 3 months prior to screening
- Rapidly progressive glomerulonephritis as defined by a 50% decline in eGFR within 3 months prior to screening
- Renal biopsy presenting with interstitial fibrosis/tubular atrophy or glomerulosclerosis >50%, or likely to be unresponsive to immunosuppressive therapy per investigators' judgment
- Treatment with systemic CS (>5 mg/day prednisone or equivalent) for indications other than SLE or LN, e.g., acute asthma and inflammatory bowel disease
- Treatment with systemic CS for SLE or LN with >10 mg/day average of prednisone (or equivalent) in the previous 4 weeks and >20 mg/day average in the previous 1 week

* Other protocol required inclusion/ exclusion criteria may apply

Introduction

- Lupus nephritis (LN) is a form of glomerulonephritis that constitutes a severe organ manifestation of systemic lupus erythematosus, seen in up to 60% patients[1]
- The current SoC for LN includes intense immunosuppressive therapy- typically high-dose followed by a tapering regimen of oral corticosteroids [CS] plus mycophenolate mofetil/sodium (MMF/MPS) or cyclophosphamide[1]
- However, only 30–50% of patients achieve a complete renal response (CRR) after treatment, and up to 35% of responders may relapse. Further, chronic CS therapy is associated with short and long-term adverse events[1]
- Dysregulation of the alternative complement pathway (AP) is implicated in the pathogenesis of LN; nearly 30% of patients with LN also have anti-C3 autoantibodies that contribute to overactivation of the AP. Thus, a targeted therapy to prevent activation of the AP might be beneficial for treatment in LN[2-5]
- Iptacopan is an oral, proximal complement inhibitor that specifically binds factor B and inhibits the activation of AP

Purpose

- To evaluate the efficacy, safety, and tolerability of iptacopan in combination with SoC with and without oral corticosteroids in adult patients with active LN class III–IV, ±V

Study design

- This Phase 2 (NCT05268289) adaptive, randomized, double-blind, dose-exploration, parallel-group, placebo-controlled, multicenter trial consists of two-parts of 52 weeks each (**Figure 1**)[6]
- Part 1 will evaluate whether the use of iptacopan 200 mg twice-a-day (b.i.d) in combination with MMF/MPS and CS, is efficacious and safe compared with SoC (MMF/MPS + CS). An interim analysis (IA) will be performed when ~80 patients in Part 1 have completed the Week 24 visit
- If iptacopan is found to be effective (in terms of reduction in proteinuria and other renal endpoints) and safe in the IA, Part 2 (N~160) will be initiated to evaluate the efficacy and safety of (i) iptacopan 50 mg b.i.d + MMF/MPS + CS, and (ii) iptacopan 200 mg b.i.d + MMF/MPS without CS (**Figure 1**)
- The key inclusion and exclusion criteria are shown in **Figure 2**

Primary objective

- To assess the proportion of patients treated with iptacopan achieving CRR at Week 24 in the absence of renal flares, compared with SoC alone
 - CRR is defined as: eGFR ≥90 mL/min/1.73m^2 or no less than 85% of baseline value and 24-h UPCR ≤ 0.5 g/g
 - The proportion of patients achieving CRR at Week 24 will be assessed in the following regimen vs SoC alone:
 - **Part 1:** iptacopan 200 mg b.i.d + MMF/MPS + CS
 - **Part 2:** iptacopan 50 mg b.i.d + MMF/MPS + CS and iptacopan 200 mg b.i.d + MMF/MPS without CS
 - The SoC treatment in the control arms of Part 1 and Part 2 is identical (MMF/MPS + CS)

Secondary objectives

To assess the following endpoints compared to SoC:

- Proportion of patients achieving CRR in the absence of renal flares at Week 52
- Proportion of patients achieving CRR or partial renal response* (PRR) in the absence of renal flares at Weeks 24 and 52
- Proportion of patients achieving ≥25% UPCR reduction in the absence of renal flares compared to baseline at Week 24 and the frequency of renal flares between Weeks 24 and 52
- The dose-exposure response for reduction in proteinuria at Week 24
- Change from baseline in FACIT-Fatigue, SLEDAI-2K, and BILAG-2004 scores at Weeks 24 and 52
- Time-to-CRR based on first morning void urine samples
- Safety and tolerability

Statistical analysis

- CRR rates at Weeks 24 and 52 will be estimated using a logistic regression model
 - Pre-specified IA: The decision to start recruitment for Part 2 will be taken if difference in CRR rates at Week 24 between iptacopan 200 mg b.i.d + MMF/MPS+ CS arm versus MMF/MPS + CS is statistically significant at α=0.1 one-sided. However, the totality of efficacy and safety data up to the IA cut-off point and the outcome will determine the start of Part 2
- Time-to-CRR will be described using Kaplan-Meier estimates by treatment arms and comparison will be conducted using a Cox proportional hazard model
- A negative binomial model will be used to compare rates of renal flares or rate of use of CS at a dose exceeding an average of 20 mg/day
- A mixed model for repeated measures will be used to analyze the change in patient reported outcomes
- The control arms of Part 1 and Part 2 will be pooled at the primary and final analysis to achieve greater power

Footnotes

*Partial renal response defined as:
eGFR ≥90 mL/min/1.73m^2 or no less than 80% of baseline value and ≥50% reduction in 24h UPCR < 1.0 g/g.

Abbreviations

ANA, anti-nuclear antibody; ACEi, angiotensin-converting enzyme inhibitor; ARB, angiotensin receptor blocker; b.i.d, twice-a-day; BILAG, British Isles lupus activity; CRR, complete renal response; CS, corticosteroids; eGFR, estimated glomerular filtration rate; FACIT, functional assessment of chronic illness therapy; IA, interim analysis; LN, lupus nephritis; MMF/MPS, mycophenolate mofetil/sodium; SLEDAI-2K, systemic lupus erythematosus disease activity index 2000; SoC, standard of care; UPCR, urine protein creatinine ratio; W, week.

References

1. Anders H-J, et al. *Nat Rev Dis Primers*. 2020; 6(1):7.
2. Elliott MK, et al. *Kidney Int*. 2004;65(1):129–138.
3. Vasilev VV, et al. *J Biol Chem*. 2015; 290(42):25343-25355.
4. Birmingham DJ, et al. *Clin J Am Soc Nephrol*. 2016;11(1):47–53.
5. Schubart A, et al. *Proc Natl Acad Sci* USA. 2019;116(16):7926–7931.
6. https://clinicaltrials.gov/ct2/show/NCT05268289 (Accessed February 2023).

Conflict of Interest

UKV, PM, JL, NW, MM are employees of Novartis. TMC has received unrestricted research grant from Astellas Pharma and consultancy fee from AstraZeneca and Otsuka. H-JA has received consultancy fees from GSK, Novartis, Janssen, Sanofi, Bayer, Boehringer-Ingelheim, PreviPharma, Kezar, Otsuka, Vifor DJ has received unrestricted research grants from GSK, Roche and Vfor and consulting fees from AstraZeneca, Boehringer-Ingelheim, Chemocentryx, Novartis, Otsuka, Roche, Takeda and Vifor. BR received consultation fees from Novartis.

Acknowledgments

Professional medical writing assistance was provided by Shivani Vadapalli and design support was provided by Rupa De [both Novartis Healthcare Pvt Ltd, Hyderabad, India].

Funding

This study is sponsored by Novartis Pharma AG.

Scan QR Code for a copy of this poster

Poster presented at: the ISN World Congress of Nephrology 2023; 30 March–2 April 2023; Bangkok, Thailand.