Poster DMT20

Derrick S. Robertson | dsrobert@usf.edu

Longer-Term (Up to 6 Years) Efficacy of Ofatumumab in People With Recently Diagnosed and Treatment-Naïve Relapsing Multiple Sclerosis

Gabriel Pardo,[1] Stephen L. Hauser,[2] Amit Bar-Or,[3] Ralf Gold,[4] Xavier Montalban,[5] Jeffrey A. Cohen,[6] Derrick S. Robertson,[7] Carrie M. Hersh,[8] Robert T. Naismith,[9] Kumaran Deiva,[10] Alit Bhatt,[11] Haoyi Fu,[12] Ibolya Boer,[13] Sven G. Meuth,[14] Anne H. Cross,[15] Jutta Gärtner,[16] Ludwig Kappos[17]

[1]Oklahoma Medical Research Foundation, Oklahoma City; OK, USA; [2]UCSF Weill Institute for Neurosciences, University of California, San Francisco, San Francisco, CA, USA; [3]Center for Neuroinflammation and Experimental Therapeutics and Department of Neurology, Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA, USA; [4]Department of Neurology, Katholisches Klinikum Bochum, Ruhr-Universität Bochum, Bochum, Germany; [5]Department of Neurology-Neuroimmunology, Centre d'Esclerosi Múltiple de Catalunya (Cemcat), Hospital Universitari Vall d'Hebron, Barcelona, Spain; [6]Department of Neurology, Mellen MS Center, Neurological Institute, Cleveland Clinic, Cleveland, OH, USA; [7]Multiple Sclerosis Division, Department of Neurology, University of South Florida, Tampa, FL, USA; [8]Cleveland Clinic Lou Ruvo Center for Brain Health, Las Vegas, NV, USA; [9]Department of Neurology, Washington University School of Medicine, Saint Louis, MO, USA; [10]Department of Pediatric Neurology, University Hospitals Paris Saclay, Hôpital Bicêtre, National Reference Center for Rare Inflammatory Brain and Spinal Diseases, Le Kremlin Bicêtre, France; [11]Novartis Healthcare Pvt. Ltd., Hyderabad, India; [12]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [13]Novartis Pharma AG, Basel, Switzerland; [14]Department of Neurology, Medical Faculty, Heinrich-Heine University Düsseldorf, Düsseldorf, Germany; [15]Department of Neurology, Division of Neuroimmunology, Washington University School of Medicine, Saint Louis, MO, USA; [16]Department of Paediatrics and Adolescent Medicine, Division of Paediatric Neurology, University Medical Centre Göttingen, Georg August University Göttingen, Göttingen, Germany; [17]Neurologic Clinic and Policlinic and MS Center, Department of Head, Spine and Neuromedicine, University Hospital Basel, Basel, Switzerland; Research Center for Clinical Neuroimmunology and Neuroscience Basel (RC2NB), Departments of Biomedicine and Clinical Research, University Hospital and University of Basel, Basel, Switzerland

KEY FINDINGS & CONCLUSIONS

- In recently diagnosed (≤3 years) and treatment-naïve (RDTN) people with relapsing multiple sclerosis (pwRMS), first-line ofatumumab (OMB) treatment for up to 6 years showed sustained efficacy with an adjusted rate of 1 relapse for every 20 years during the extension phase and profound suppression of magnetic resonance imaging (MRI) lesion activity; these results are consistent with those of the overall study population[1]
- Participants who switched from teriflunomide (TER) to OMB in the extension phase showed pronounced reductions in relapses and MRI lesion activity after the switch
- By Year 6 of treatment, 9 of 10 participants were free from disease activity (no evidence of disease activity [NEDA-3]) in the continuous and switch groups
 - High rates of NEDA-3 were achieved within 2 years with first-line OMB, whereas rates of NEDA-3 increased rapidly after switching from TER to OMB
- First-line OMB was associated with significantly fewer confirmed disability worsening events and lower rates of progression independent of relapse activity up to 6 years compared with participants who switched from TER to OMB
 - The efficacy benefit of first-line OMB in delaying disability worsening cannot be recovered in those switching from TER to OMB
- These long-term efficacy results up to 6 years, combined with the favorable benefit–risk profile demonstrated in the overall study population,[1] support the use of OMB as first-line therapy for RDTN pwRMS

Scan to obtain a copy of the poster

https://bit.ly/CMSCDMT20

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

This study is sponsored by Novartis Pharma AG, Basel, Switzerland

Poster presented at the Consortium of Multiple Sclerosis Centers (CMSC) 38th Annual Meeting • May 29-June 1, 2024 • Nashville, TN, USA

Previously presented at the American Academy of Neurology Annual Meeting • April 13-18, 2024 • Denver, CO, USA

INTRODUCTION

- Ofatumumab (OMB), a fully human anti-CD20 monoclonal antibody with a 20-mg subcutaneous monthly dosing regimen, is approved for treating relapsing multiple sclerosis (RMS) in adults[2]
- In the phase 3 ASCLEPIOS I/II trials in people with RMS (pwRMS), OMB demonstrated superior efficacy in reducing the annualized relapse rate (ARR), suppressing magnetic resonance imaging (MRI) lesion activity, and delaying disability worsening, while maintaining a favorable safety profile vs teriflunomide (TER)[3]
- In the subgroup of recently diagnosed (≤3 years) and treatment-naïve (RDTN) participants, OMB showed a superior benefit–risk profile compared with TER, with an almost complete abrogation of inflammatory disease activity and no unexpected safety signals, supporting its use as a first-line treatment in early RMS[4]
- Results previously reported from the ASCLEPIOS I/II trials and ALITHIOS open-label extension study demonstrated sustained efficacy for up to 4 years in RDTN participants[5]

OBJECTIVE

- To assess the long-term efficacy of OMB for up to 6 years in RDTN pwRMS

METHODS

- These analyses include cumulative data from the RDTN subgroups randomized to OMB in the core ASCLEPIOS studies (continuous group) and those originally randomized to TER in the ASCLEPIOS studies and switching to OMB in the ALITHIOS extension study (switch group) (**Figure 1**)
- Data cutoff: Sep 25, 2022 (up to 5 years)/Sep 25, 2023 (up to 6 years)

Figure 1. Participant Disposition (RDTN Subgroup) and Key Assessments

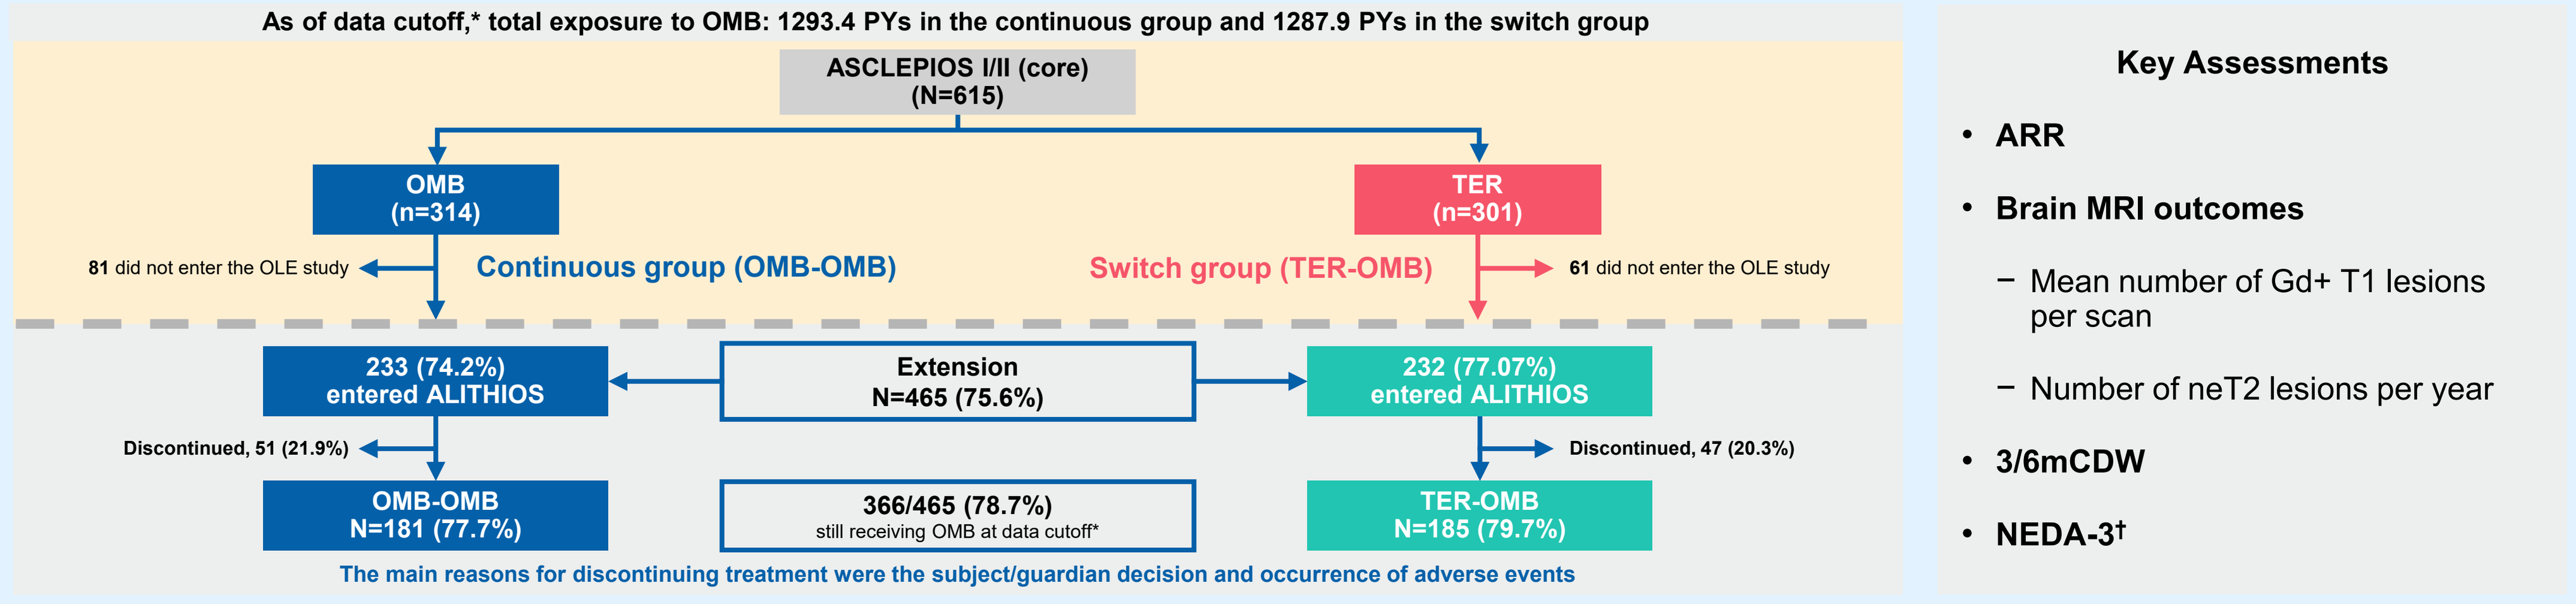

As of data cutoff,* total exposure to OMB: 1293.4 PYs in the continuous group and 1287.9 PYs in the switch group
ASCLEPIOS I/II (core) (N=615)
OMB (n=314)
TER (n=301)
81 did not enter the OLE study
Continuous group (OMB-OMB)
Switch group (TER-OMB)
61 did not enter the OLE study
233 (74.2%) entered ALITHIOS
Extension N=465 (75.6%)
232 (77.07%) entered ALITHIOS
Discontinued, 51 (21.9%)
Discontinued, 47 (20.3%)
OMB-OMB N=181 (77.7%)
366/465 (78.7%) still receiving OMB at data cutoff*
TER-OMB N=185 (79.7%)
The main reasons for discontinuing treatment were the subject/guardian decision and occurrence of adverse events
Key Assessments
ARR
Brain MRI outcomes
Mean number of Gd+ T1 lesions per scan
Number of neT2 lesions per year
3/6mCDW
NEDA-3†

3/6mCDW, 3- and 6-month confirmed disability worsening; ARR, annualized relapse rate; Gd+, gadolinium-enhancing; MRI, magnetic resonance imaging; NEDA-3, no evidence of disease activity; neT2 lesions, new or enlarging T2 lesions; OLE, open-label extension; OMB, ofatumumab; OMB-OMB, continuous ofatumumab; PY, patient year; RDTN, recently diagnosed (≤3 years) and treatment-naive; TER, teriflunomide; TER-OMB, switch from teriflunomide to ofatumumab
*Data cutoff: September 25, 2023 (≤6 years); †Defined as no 6mCDW, no confirmed multiple sclerosis relapse, no neT2 lesions compared with baseline, and no Gd+ T1 lesions

RESULTS

- Characteristics of RDTN participants were typical of patients with early RMS and were generally balanced between treatment groups (**Table 1**)
- Over a period of up to 6 years, first-line continuous vs later initiation of OMB was associated with a 44% reduction in the cumulative number of relapses (**Figure 2A**)
- ARR remained low with first-line continuous OMB, reaching an adjusted rate in the extension period that corresponded to 1 relapse for every 20 years (**Figure 2B**)
- Switching from TER to OMB resulted in a pronounced 71.3% reduction in ARR (**Figure 2B**)

Table 1. Baseline Demographics and Disease Characteristics

Characteristics*	Continuous OMB-OMB group: Baseline from core (N=314)	Switch TER-OMB group (N=301): Baseline from core (N=301)	Switch TER-OMB group (N=301): Baseline from OLE (N=232)
Age, years	36.8 (9.40)	35.7 (9.03)	37.7 (8.99)
BMI, kg/m²	25.93 (6.15)	26.19 (6.06)	25.71 (5.71)
Female, n (%)	217 (69.1)	195 (64.8)	155 (66.8)
Time since MS diagnosis, years	0.58 (0.63)	0.53 (0.51)	2.44 (0.60)
Time since first MS symptom, years	3.41 (3.96)	3.25 (4.28)	5.16 (4.23)
EDSS at baseline	2.30 (1.2)	2.28 (1.2)	2.20 (1.2)
Number of relapses in the past 12 months prior to screening	1.30 (0.70)	1.4 (0.72)	0.10 (0.41)
Number of Gd+ T1 lesions	1.8 (4.35)	1.4 (2.79)	0.7 (2.01)
Proportion of participants free of Gd+ T1 lesions, n (%)	173 (55.1)	171 (56.8)	169 (72.8)
Total volume of T2 lesions, cm³	10.1 (12.23)	8.3 (8.83)	NA†

BMI, body mass index; EDSS, Expanded Disability Status Scale; Gd+, gadolinium-enhancing; MS, multiple sclerosis; OLE, open-label extension, OMB, ofatumumab; OMB-OMB, continuous ofatumumab; TER-OMB, switch from teriflunomide to ofatumumab
*Data are represented as mean (SD) unless specified otherwise; for participants newly switched to OMB, their baseline values from the extension study contribute to the overall summary; †Data are not collected for baseline from extension

Figure 2. (A) Cumulative Number of Relapses Up to 6 Years; (B) Within-Group Comparison* Between the Core and Extension Phase

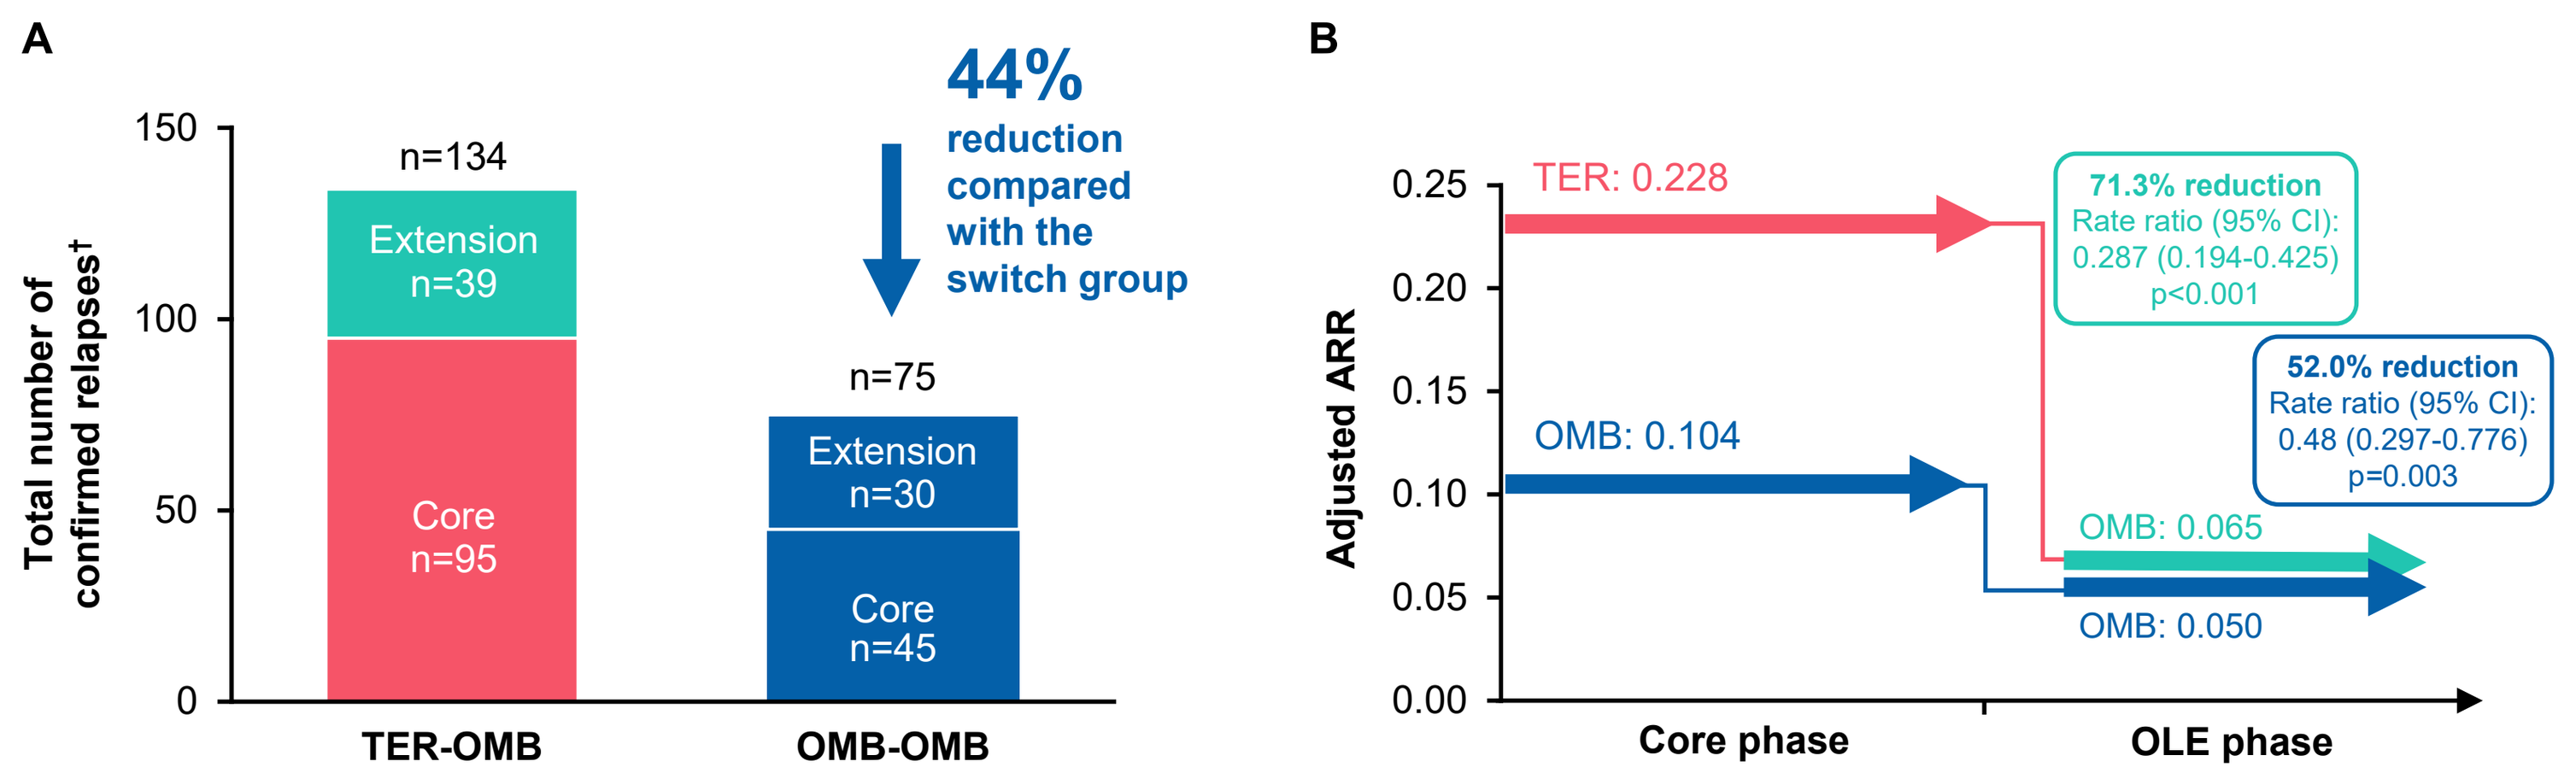

A
Total number of confirmed relapses†
150
100
50
0
n=134
Extension n=39
Core n=95
n=75
Extension n=30
Core n=45
TER-OMB
OMB-OMB
44% reduction compared with the switch group
B
Adjusted ARR
0.25
0.20
0.15
0.10
0.05
0.00
TER: 0.228
OMB: 0.104
71.3% reduction Rate ratio (95% CI): 0.287 (0.194-0.425) p<0.001
52.0% reduction Rate ratio (95% CI): 0.48 (0.297-0.776) p=0.003
OMB: 0.065
OMB: 0.050
Core phase
OLE phase

ARR, annualized relapse rate; EDSS, Expanded Disability Status Scale; Gd+, gadolinium-enhancing; OLE, open-label extension; OMB, ofatumumab; OMB-OMB, continuous ofatumumab; TER, teriflunomide; TER-OMB, switch from teriflunomide to ofatumumab
*Obtained from fitting a piecewise negative binomial model for the core phase and extension phase time period with log-link, adjusted for treatment and region as factors, number of relapses in previous year, baseline EDSS, baseline number of Gd+ lesions and the patient's age at baseline as covariates. The natural log of the time-in-study (in years) by period is used as offset to annualize the relapse rate in each period. Baseline variables are from the core study baseline; †Confirmed relapses are those accompanied by a clinically relevant change in the EDSS

- Over a period of ≤6 years, first-line continuous vs later initiation of OMB was associated with a 96.4% reduction in the cumulative number of Gd+ T1 lesions (**Figure 3A**)

Figure 3. (A) Cumulative Number of Gd+ T1 Lesions Up to 6 Years; (B) Within-Group Comparison* Between the Core and Extension Phase

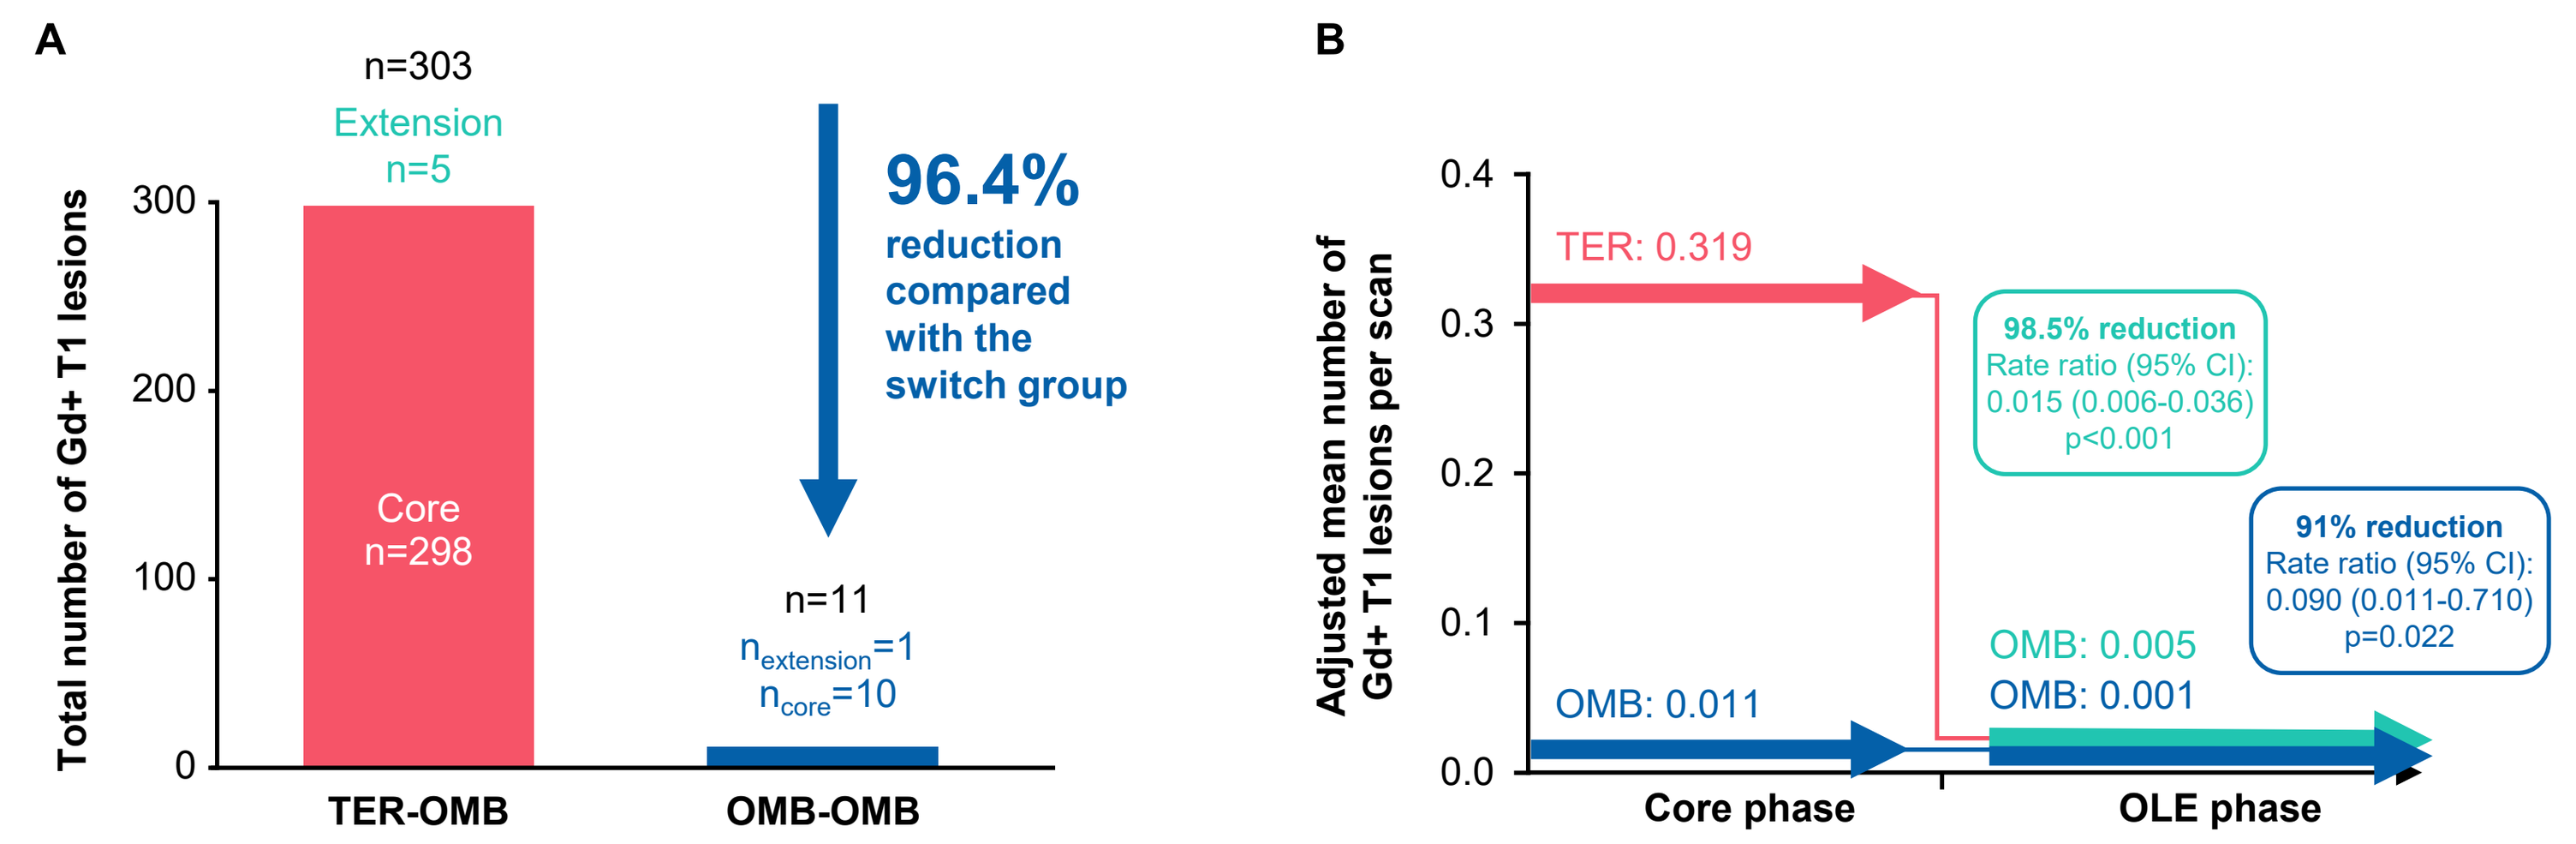

A
Total number of Gd+ T1 lesions
300
200
100
0
n=303
Extension n=5
Core n=298
n=11
n extension=1
n core=10
TER-OMB
OMB-OMB
96.4% reduction compared with the switch group
B
Adjusted mean number of Gd+ T1 lesions per scan
0.4
0.3
0.2
0.1
0.0
TER: 0.319
98.5% reduction Rate ratio (95% CI): 0.015 (0.006-0.036) p<0.001
91% reduction Rate ratio (95% CI): 0.090 (0.011-0.710) p=0.022
OMB: 0.011
OMB: 0.005
OMB: 0.001
Core phase
OLE phase

Gd+, gadolinium-enhancing; OLE, open-label extension; OMB, ofatumumab; OMB-OMB, continuous ofatumumab; TER, teriflunomide; TER-OMB, switch from teriflunomide to ofatumumab
*Estimated from fitting a piecewise negative binomial model for the core phase and extension phase time period with log-link, adjusted for treatment and region as factors and baseline number of Gd+ T1 lesions and patient's age at baseline as covariates. The natural log of the number of scans with evaluable Gd+ lesion counts by period is used as offset to obtain the lesion rate per scan in each period. Baseline variables are from the core study baseline. All p-values are nominal

- First-line continuous OMB treatment maintained an almost complete suppression of Gd+ T1 lesion activity up to Year 6 (**Figure 3A**)
- Switching from TER to OMB led to a rapid suppression of Gd+ T1 lesion activity to closely match the continuous OMB group (**Figure 3B**)
- A significant and sustained reduction in the number of new or enlarging T2 (neT2) lesions was observed in RDTN participants receiving OMB for up to 6 years (**Figure 4**)
- Over a period of up to 6 years, first-line initiation of OMB was associated with an 82.7% reduction in the cumulative number of neT2 lesions (**Figure 4A**)
- First-line continuous OMB profoundly suppressed the number of neT2 lesions up to Year 6 (**Figure 4A**)
- Switching from TER to OMB resulted in a profound reduction in the number of neT2 lesions (**Figure 4B**)

Figure 4. (A) Cumulative Number of neT2 Lesions Up to 6 Years; (B) Within-Group Comparison Between the Core and Extension Phase

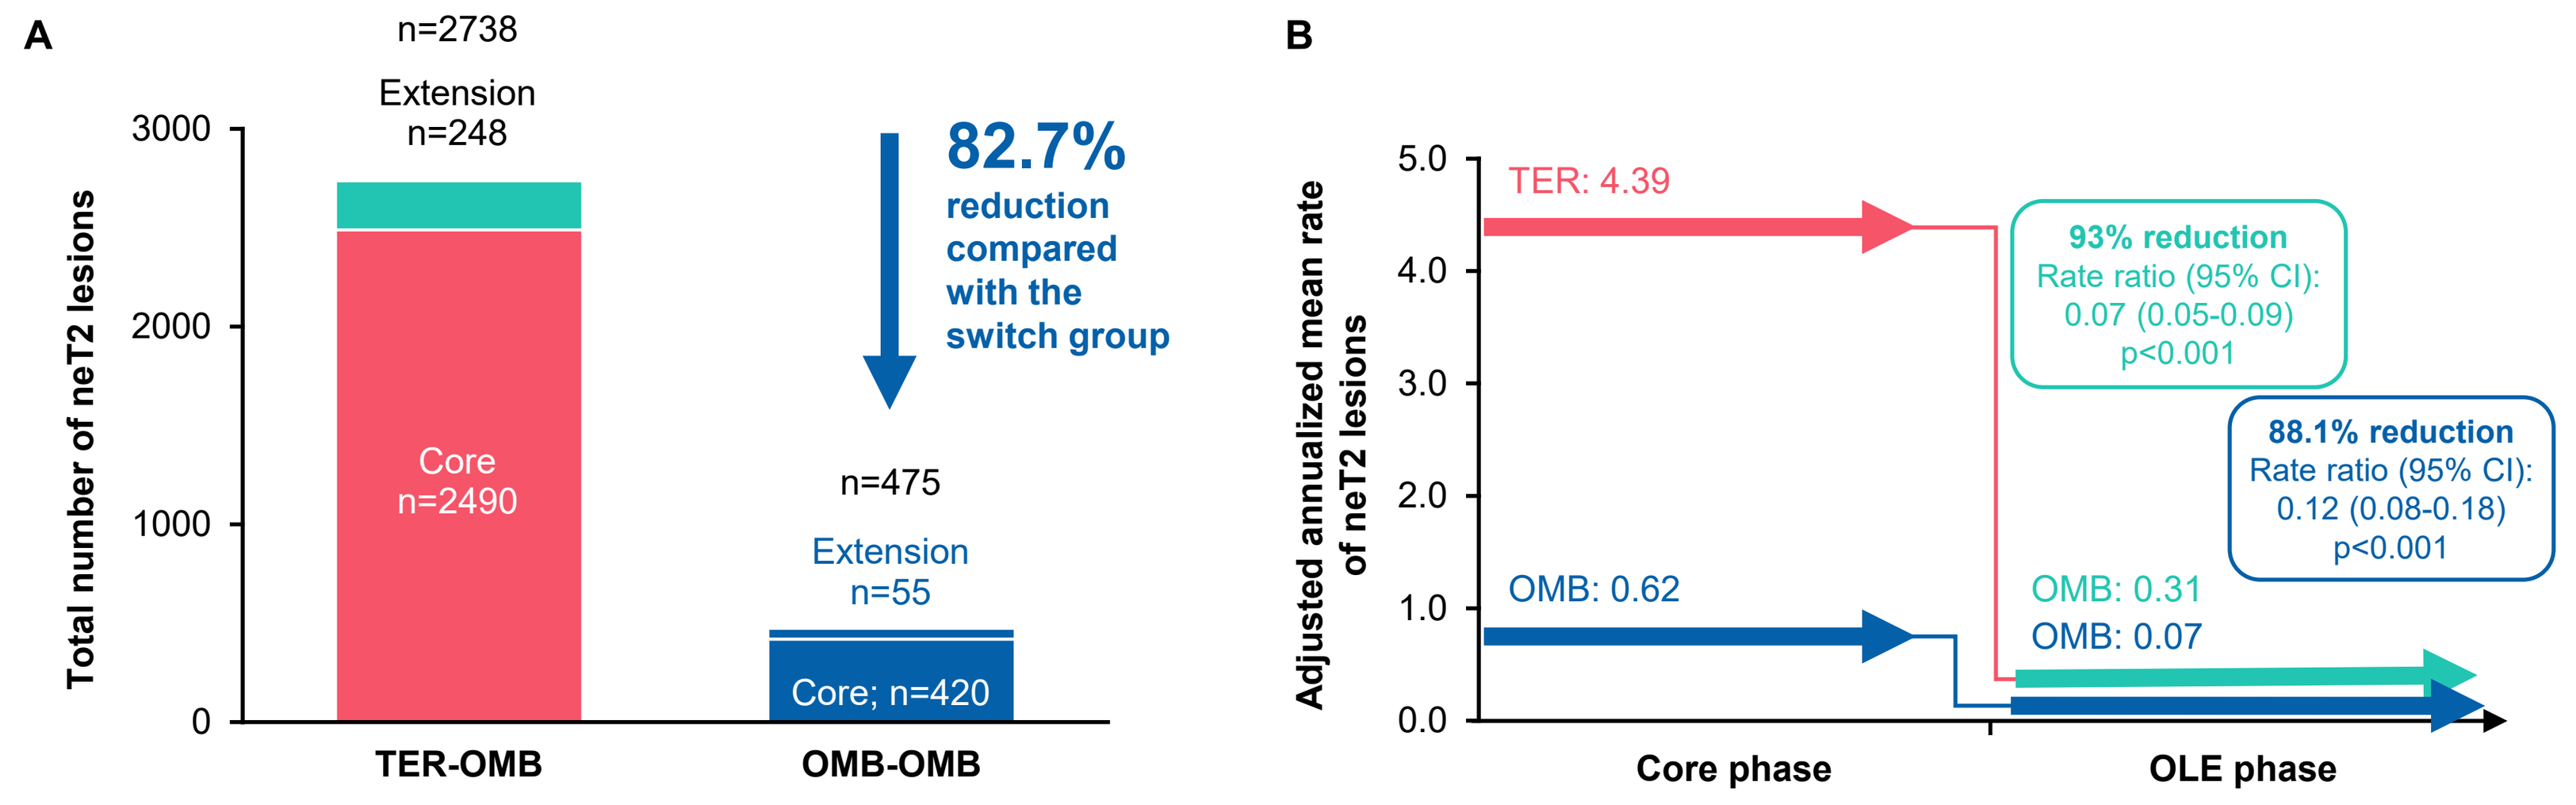

A
Total number of neT2 lesions
3000
2000
1000
0
n=2738
Extension n=248
Core n=2490
n=475
Extension n=55
Core; n=420
TER-OMB
OMB-OMB
82.7% reduction compared with the switch group
B
Adjusted annualized mean rate of neT2 lesions
5.0
4.0
3.0
2.0
1.0
0.0
TER: 4.39
OMB: 0.62
93% reduction Rate ratio (95% CI): 0.07 (0.05-0.09) p<0.001
88.1% reduction Rate ratio (95% CI): 0.12 (0.08-0.18) p<0.001
OMB: 0.31
OMB: 0.07
Core phase
OLE phase

neT2 lesion, new or enlarging T2 lesion; OLE, open-label extension; OMB, ofatumumab; OMB-OMB, continuous ofatumumab; TER, teriflunomide; TER-OMB, switch from teriflunomide to ofatumumab
All p-values are nominal

- First-line OMB treatment and first-line continuous OMB treatment in RDTN participants was associated with a significantly lower number of 3-month confirmed disability worsening (3mCDW) events up to 6 years (**Figure 5**)
- 3-month progression independent of relapse activity (3mPIRA) events occurred in 14.3% vs 20.4% of RDTN participants in the continuous and switch groups, respectively
 - 3mPIRA is defined as a 3mCDW event with either no prior relapse or an onset >90 days after the start date of the last investigator-reported relapse (irrespective of the Expanded Disability Status Scale [EDSS] confirmation). In addition, to qualify as a PIRA event, no relapse must occur within 30 days after confirmation of EDSS worsening. Cutoff for the core and extension periods refers to the first dose of OMB in the extension period
- The significant efficacy benefit of first-line OMB on 3mCDW in the core phase cannot be recovered in those initially randomized to TER and later switched to OMB (**Figure 5B**)

Figure 5. (A) Cumulative Number of 3mCDW Events Up to 6 Years; (B) Cumulative Rate of 3mCDW Events

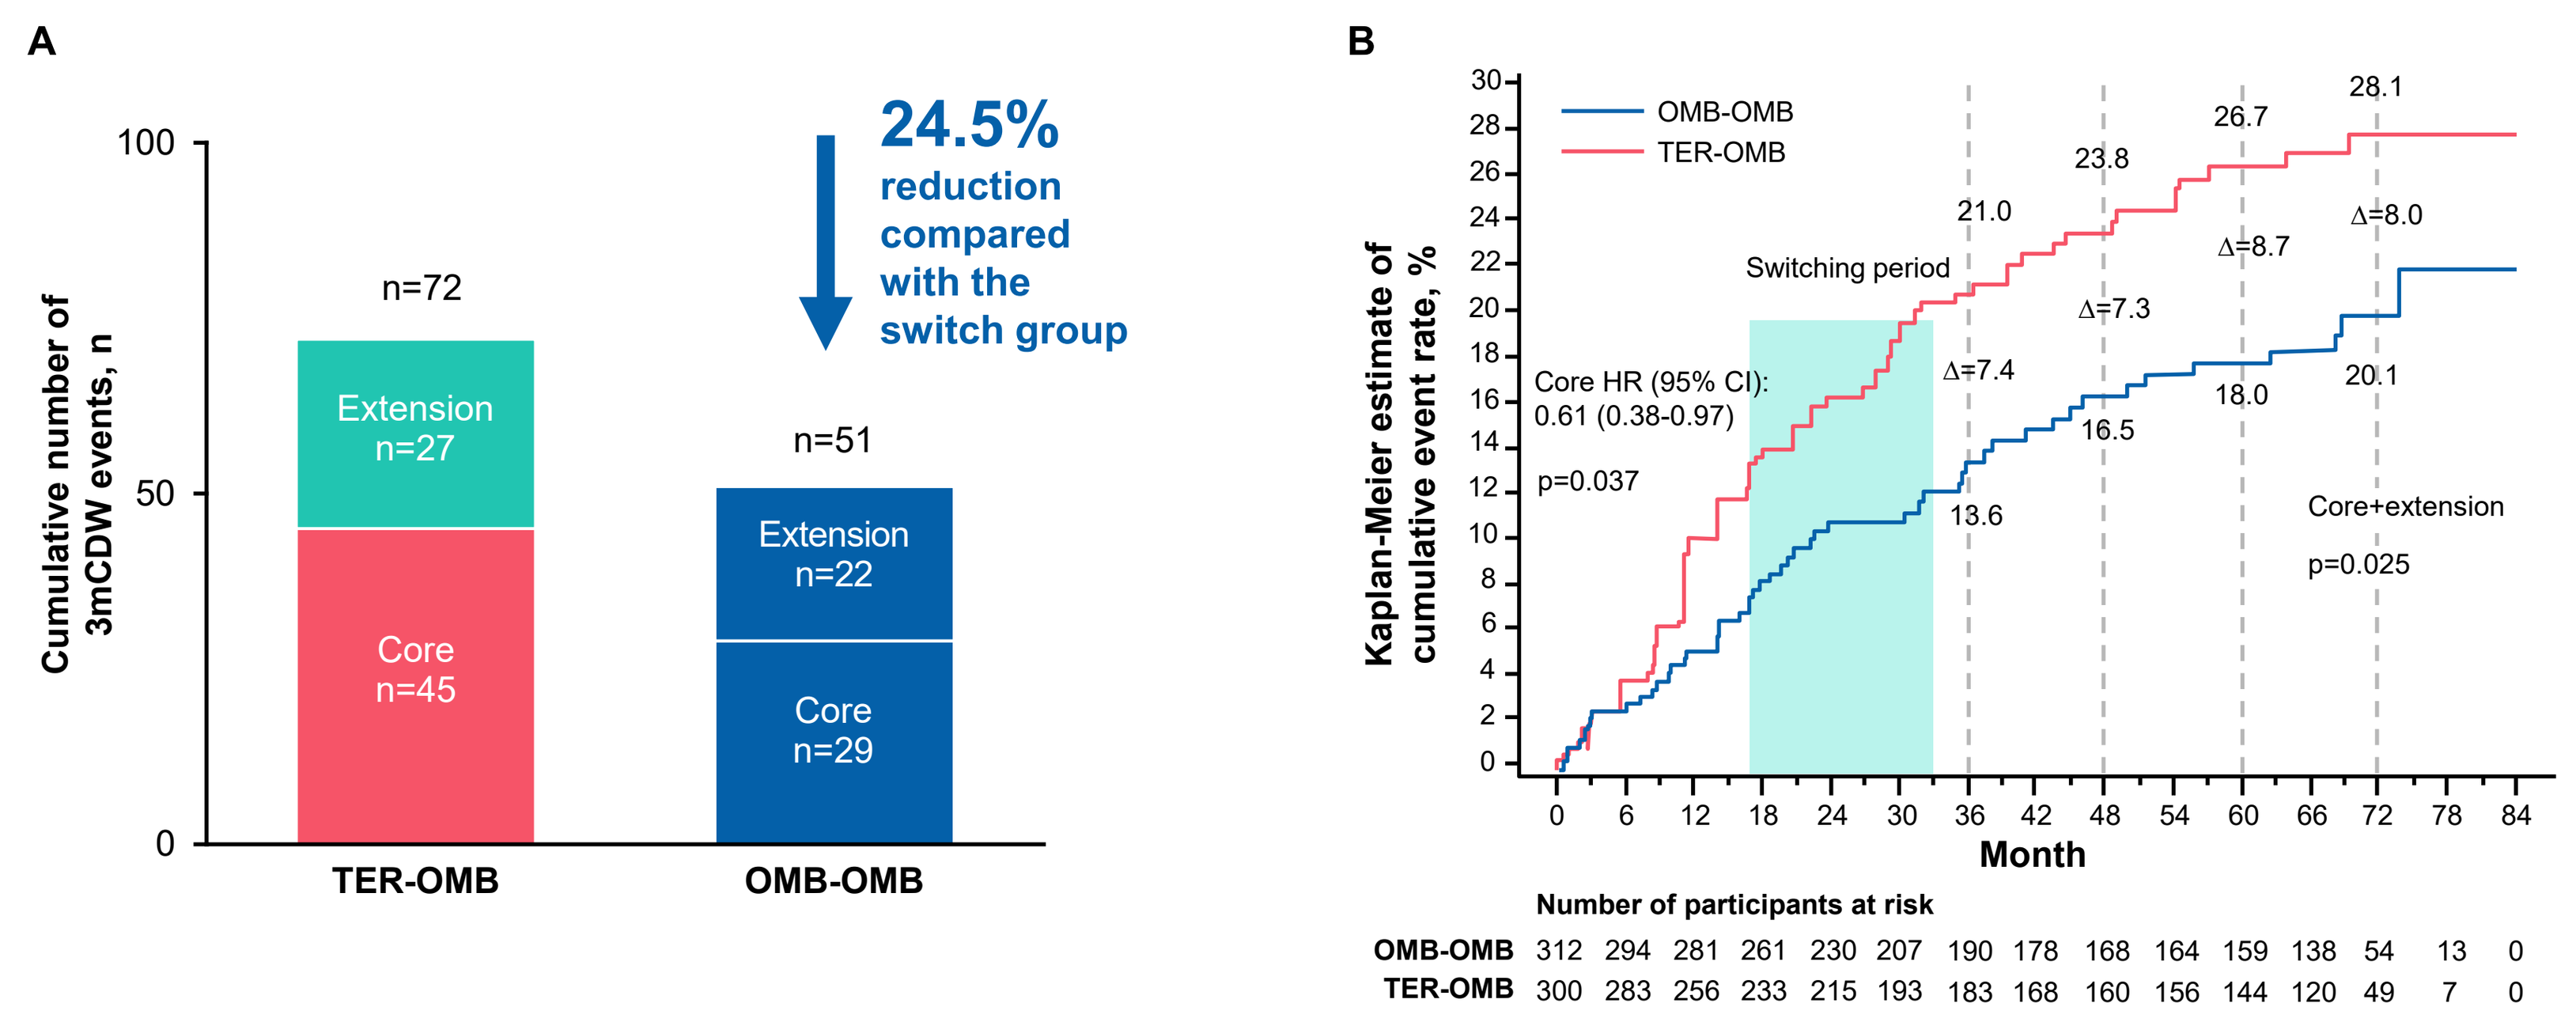

A
Cumulative number of 3mCDW events, n
100
50
0
n=72
Extension n=27
Core n=45
n=51
Extension n=22
Core n=29
TER-OMB
OMB-OMB
24.5% reduction compared with the switch group
B
Kaplan–Meier estimate of cumulative event rate, %
OMB-OMB
TER-OMB
Switching period
Core HR (95% CI): 0.61 (0.38-0.97)
p=0.037
21.0
23.8
26.7
28.1
Δ=7.4
Δ=7.3
Δ=8.7
Δ=8.0
13.6
16.5
18.0
20.1
Core+extension
p=0.025
Month
0 6 12 18 24 30 36 42 48 54 60 66 72 78 84
Number of participants at risk
OMB-OMB 312 294 281 261 230 207 190 178 168 164 159 138 54 13 0
TER-OMB 300 283 256 233 215 193 183 168 160 156 144 120 49 7 0

Δ, difference in Kaplan–Meier estimates (TER-OMB minus OMB-OMB); 3mCDW, 3-month confirmed disability worsening; HR, hazard ratio; OMB-OMB, continuous ofatumumab; TER-OMB, switch from teriflunomide to ofatumumab
HR was determined by Cox regression model; p-value represents log-rank test

- First-line OMB treatment and first-line continuous OMB treatment in RDTN participants was associated with a significantly lower number of 6-month confirmed disability worsening (6mCDW) events up to 6 years (**Figure 6**)
- 6-month progression independent of relapse activity (6mPIRA) events occurred in 11.1% vs 16.8% of RDTN participants in the continuous and switch groups, respectively
 - 6mPIRA is defined as a 6mCDW event with either no prior relapse or an onset >90 days after the start date of the last investigator-reported relapse (irrespective of the EDSS confirmation). In addition, to qualify as a PIRA event, no relapse must occur within 30 days after confirmation of EDSS worsening. Cutoff for the core and extension periods refers to the first dose of OMB in the extension period
- The significant efficacy benefit of first-line OMB on 6mCDW in the core phase cannot be recovered in those initially randomized to TER and later switched to OMB (**Figure 6B**)

Figure 6. (A) Cumulative Number of 6mCDW Events Up to 6 Years; (B) Cumulative Rate of 6mCDW Events

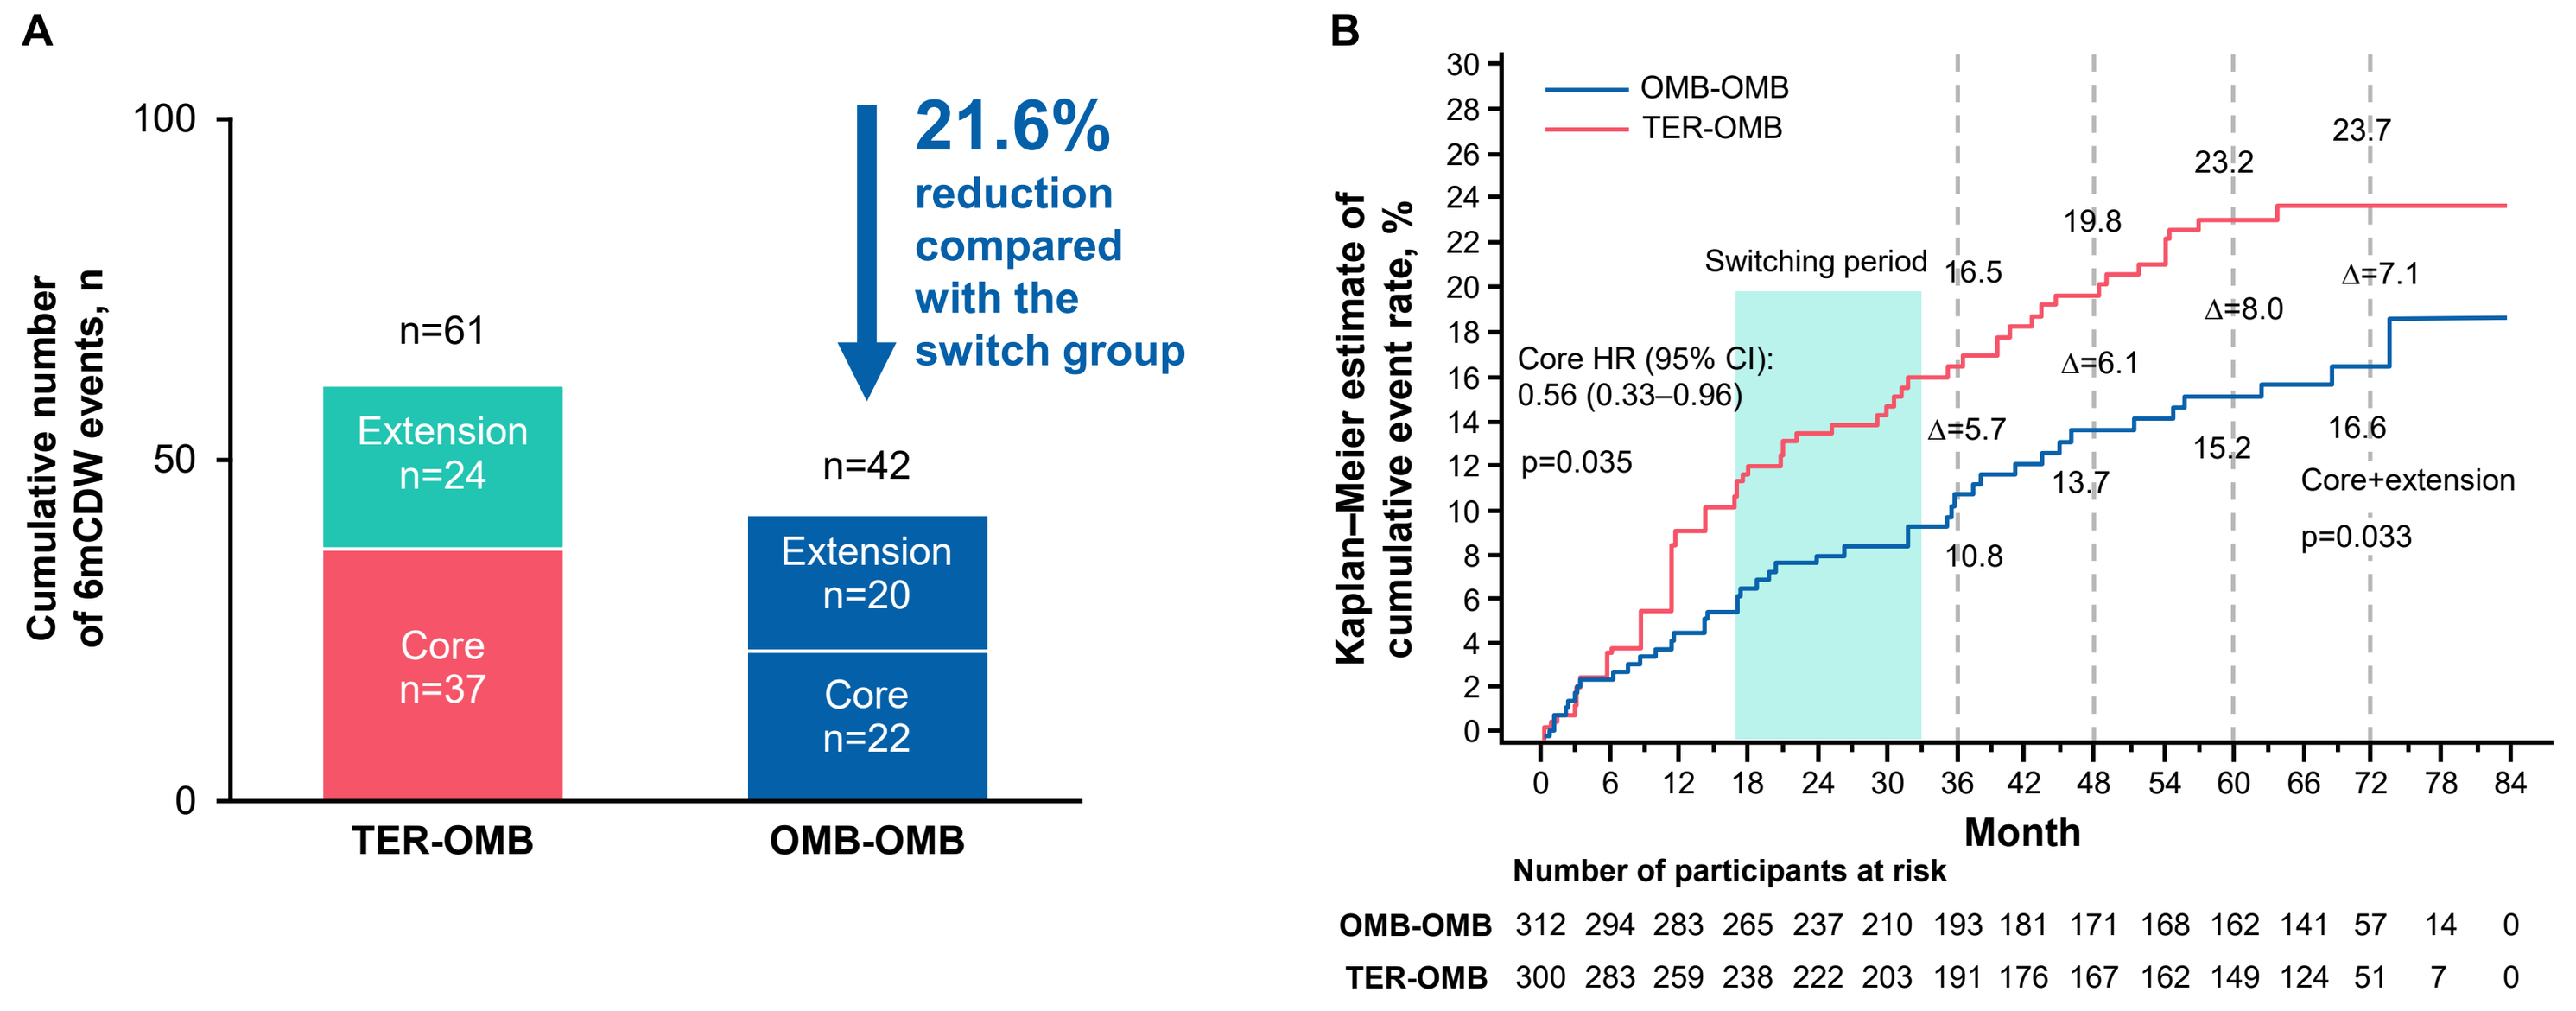

A
Cumulative number of 6mCDW events, n
100
50
0
n=61
Extension n=24
Core n=37
n=42
Extension n=20
Core n=22
TER-OMB
OMB-OMB
21.6% reduction compared with the switch group
B
Kaplan–Meier estimate of cumulative event rate, %
OMB-OMB
TER-OMB
Switching period
Core HR (95% CI): 0.56 (0.33–0.96)
p=0.035
16.5
19.8
23.2
23.7
Δ=5.7
Δ=6.1
Δ=8.0
Δ=7.1
10.8
13.7
15.2
16.6
Core+extension
p=0.033
Month
0 6 12 18 24 30 36 42 48 54 60 66 72 78 84
Number of participants at risk
OMB-OMB 312 294 283 265 237 210 193 181 171 168 162 141 57 14 0
TER-OMB 300 283 259 238 222 203 191 176 167 162 149 124 51 7 0

Δ, difference in Kaplan–Meier estimates (TER-OMB minus OMB-OMB); 6mCDW, 6-month confirmed disability worsening; HR, hazard ratio; OMB-OMB, continuous ofatumumab; TER-OMB, switch from teriflunomide to ofatumumab
HR was determined by Cox regression model; p-value represents log-rank test

- By Year 6 of treatment, 9 of 10 participants were free from disease activity (no evidence of disease activity [NEDA-3]) in the continuous and switch groups (**Figure 7**)
 - The observed rapid increase in the proportion of participants with NEDA-3 with first-line continuous OMB was maintained over 6 years
 - Participants who were initially on TER had significantly lower NEDA-3 rates, but a rapid increase in NEDA-3 was observed after switching to OMB

Figure 7. NEDA-3* Status Up to 6 Years of OMB Treatment (mFAS†)

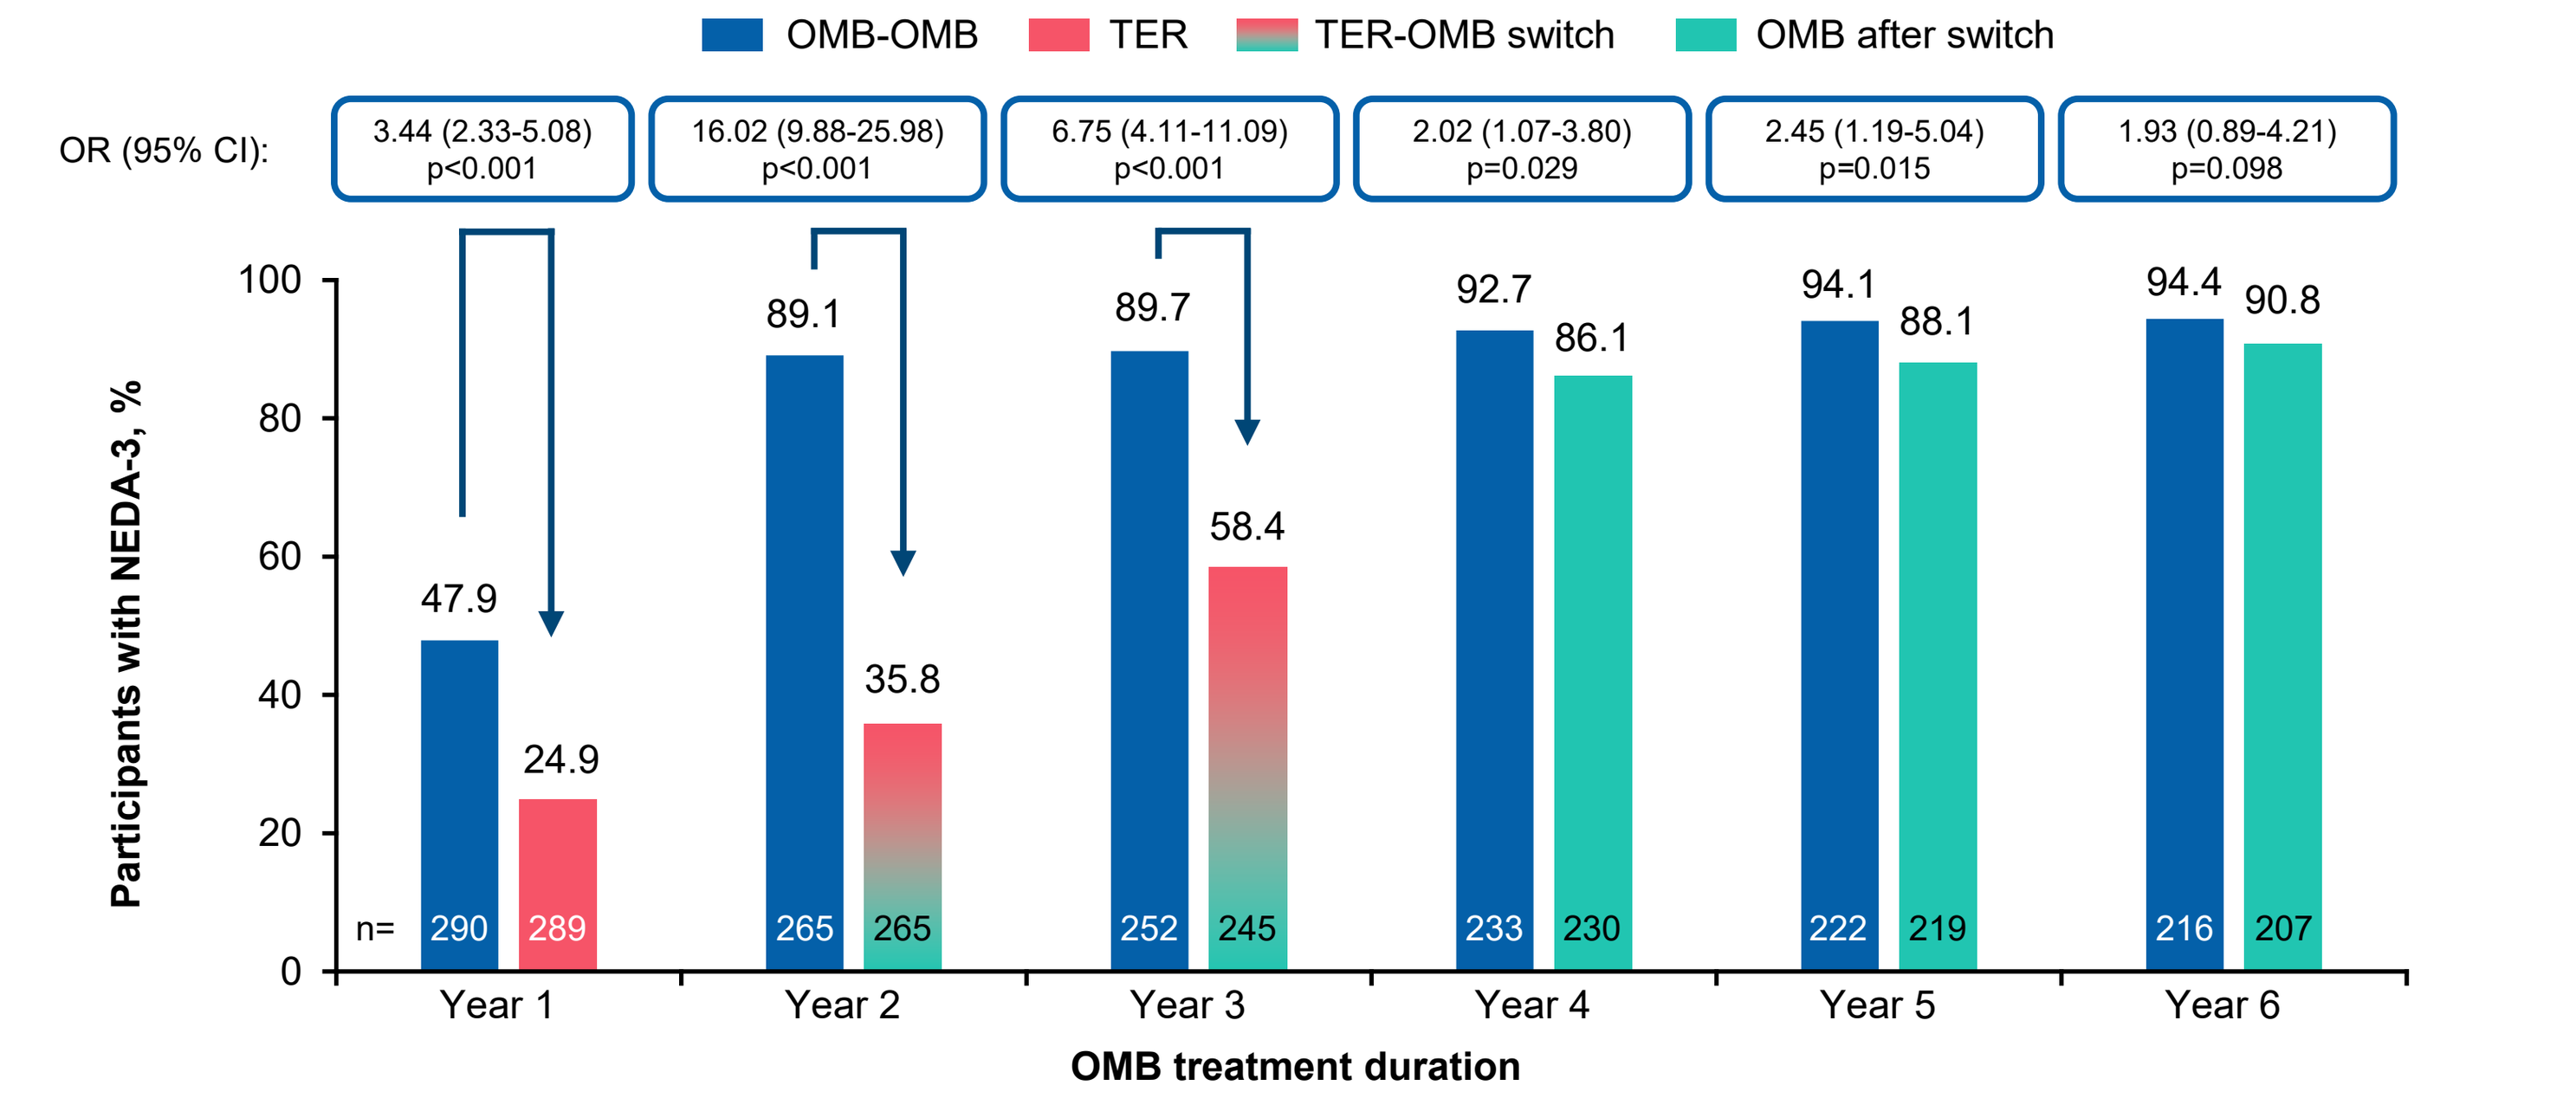

OMB-OMB
TER
TER-OMB switch
OMB after switch
OR (95% CI):
3.44 (2.33-5.08) p<0.001
16.02 (9.88-25.98) p<0.001
6.75 (4.11-11.09) p<0.001
2.02 (1.07-3.80) p=0.029
2.45 (1.19-5.04) p=0.015
1.93 (0.89-4.21) p=0.098
Participants with NEDA-3, %
47.9
24.9
89.1
35.8
89.7
58.4
92.7
86.1
94.1
88.1
94.4
90.8
n= 290 289
265 265
252 245
233 230
222 219
216 207
Year 1
Year 2
Year 3
Year 4
Year 5
Year 6
OMB treatment duration

6mCDW, 6-month confirmed disability worsening; Gd+, gadolinium-enhancing; mFAS, modified full analysis set; MS, multiple sclerosis; n, the total number of participants in the treatment group with response variable defined; NEDA-3, no evidence of disease activity; neT2 lesions, new or enlarging T2 lesions; OMB, ofatumumab; OMB-OMB, continuous ofatumumab; OR, odds ratio; TER, teriflunomide; TER-OMB, switch from teriflunomide to ofatumumab
*NEDA-3 is defined as no 6mCDW, no confirmed MS relapse, no neT2 lesions compared with baseline, and no Gd+ T1 lesions. Statistical model used logistic regression adjusting for treatment and region as factors and age, baseline EDSS, and number of Gd+ T1 lesions at baseline as covariates; †The mFAS for NEDA-3 contained all participants in the full analysis set according to the intent-to-treat principle, but participants who discontinued from the study drug prematurely for reasons other than "lack of efficacy" or "death" and had NEDA-3 before early discontinuations were excluded

References

1. Wiendl H et al. Poster presented at: *AAN* 2024; P9.010. **2.** Novartis Pharmaceuticals Corporation. Prescribing information. Kesimpta® 2024. Accessed February 15, 2024. https://www.novartis.us/sites/www.novartis.us/files/kesimpta.pdf; **3.** Hauser SL et al. *N Engl J Med.* 2020;383:546-557. **4.** Gärtner J et al. *Mult Scler.* 2022;28:1562-1575. **5.** Gärtner J et al. Poster presented at: *ECTRIMS* 2022; P052.

Acknowledgments

We thank the participants, their families, the investigators, and the staff at the participating study sites. Medical writing support was provided by Sivaram Vedantam (Novartis Healthcare Pvt. Ltd. Hyderabad) and Paul Coyle (Novartis Ireland, Ltd, Dublin), and graphic designing support was provided by Mantosh Roy (Novartis Healthcare Pvt. Ltd. Hyderabad), which was funded by Novartis Pharma AG, Basel, Switzerland. Editorial assistance for this poster was provided by Envision Pharma Group and was funded by Novartis Pharmaceuticals Corporation. The final responsibility for the content lies with the authors.

Disclosures

Gabriel Pardo has received personal compensation for serving as a consultant for Biogen, Celgene, EMD Serono, Genentech, Genzyme, Greenwich Neuroscience, Horizon Therapeutics, Novartis, and TG Therapeutics. He has also received personal compensation for serving on a speakers' bureau for Biogen, BMS, Celgene, EMD Serono, Novartis, and Viela Bio. **Stephen L. Hauser** serves on the board of trustees for Neurona and serves on scientific advisory boards for Accure, Alector, and Annexon and has received travel reimbursement and writing assistance for CD-20–related meeting and presentations from Roche and Novartis. **Amit Bar-Or** has participated as a speaker in meetings sponsored by and received consulting fees and/or grant support from Accure, Atara Biotherapeutics, Biogen, BMS/Celgene/Receptos, GlaxoSmithKline, Gossamer, Janssen/Actelion, Medimmune, Merck/EMD Serono, Novartis, Roche/Genentech, and Sanofi-Genzyme. **Ralf Gold** has received compensation for consulting or speaking from Bayer Healthcare, Biogen Idec, Merck Serono, Novartis, and Teva Neuroscience. He, or the institution he works for, has received research support from Bayer Healthcare, Biogen Idec, Merck Serono, Novartis, and Teva Neuroscience. He has also received honoraria as a journal editor from SAGE and Thieme Verlag. **Xavier Montalban** has received speaking honoraria and travel expenses for participation in scientific meetings, has been a steering committee member of clinical trials, or participated in advisory boards of clinical trials in the past years with Actelion, Alexion, Bayer, Biogen, Celgene, EMD Serono, Excemed, Genzyme, Immunic, Medday, Merck, MSIF, Mylan, Nervgen, NMSS, Novartis, Roche, Sanofi-enzyme, Teva Pharmaceutical, and TG Therapeutics. **Jeffrey A. Cohen** has received personal compensation for consulting from Biogen, Bristol-Myers Squibb, Convelo, Genentech, Janssen, NervGen, Novartis, and PSI; speaking from H3 Communications; and serving as an editor for *Multiple Sclerosis* journal. **Derrick Robertson** has received fees for consulting, contracted research, and speaker's bureau from Biogen, Celgene, EMD Serono, Genentech, Janssen, Sanofi Genzyme, and TG therapeutics; consulting fees and speakers bureau from Alexion, Bristol Myers Squibb, and Horizon; consulting fees and contracted research from Novartis; consulting fees from Greenwich Biosciences; and contracted research from Atara Biotherapeutics, CorEvitas, GW Pharmaceuticals, and PCORI. **Carrie M. Hersh** has received speaking, consulting, and advisory board fees from Alexion, Bristol Myers Squibb, Biogen, EMD-Serono, Genentech, Genzyme, Horizon Therapeutics, Novartis, and TG Therapeutics. She has received research support paid to her institution by Biogen, Bristol Myers Squibb, NIH - NINDS 1U01NS111678-01A1 sub-award, Novartis, and Patient-Centered Outcomes Research Institute (PCORI). **Robert T. Naismith** has consulted for Alexion Pharmaceuticals, Biogen, Bristol Myers Squibb, Celltrion, Genentech, Genzyme, EMD Serono, Horizon Therapeutics, Novartis, Sandoz, TG Therapeutics. **Kumaran Deiva** has received personal compensation for speaker activities from Novartis and Sanofi. **Sven G. Meuth** has received honoraria for consulting from Alexion, Almirall, Amicus Therapeutics Germany, Bayer Healthcare, Biogen, Celgene, Chugai Pharma, Diamed, Genzyme, MedDay Pharmaceuticals, Merck Serono, Novartis, Novo Nordisk, ONO Pharma, QuintilesIMS, Roche, Sanofi-Aventis, and Teva. He received a research grant from Bundesinstitut für Risikobewertung (BfR), Deutsche Forschungsgemeinschaft (DFG), Else Kröner Fresenius Foundation, Gemeinsamer Bundesausschuss (G-BA), German Academic Exchange Service, German Foundation Neurology, German Ministry for Education and Research (BMBF), Hertie Foundation, Interdisciplinary Center for Clinical Studies (IZKF) Muenster, and Alexion, Almirall, Amicus Therapeutics Germany, Biogen, Diamed, Fresenius Medical Care, Genzyme, HERZ Burgdorf, Merck Serono, Novartis, ONO Pharma, Roche, and Teva. **Anne H. Cross** has received consulting fees, support, and honoraria from Academic CME, ACTRIMS, Biogen, Bristol Myers Squibb, Celgene, CME Outfitters, Conrad N. Hilton Foundation, EMD Serono, Genentech, Greenwich Biosciences (Jazz Pharmaceuticals), Horizon Therapeutics, Janssen (subsidiary of Johnson & Johnson), Merck, Novartis, Potomac Center for Medical Education, Projects In Knowledge, Roche, TG Therapeutics, The Consortium of Multiple Sclerosis Centers, and WebMD; has received a grant from the Department of Defense, USA; has been the secretary (elected) of The Consortium of Multiple Sclerosis Centers, member of the scientific advisory board of Race to Erase MS, program committee (chair) of ACTRIMS, member of the COVID-19 advisory committee of the National Multiple Sclerosis Society and National Multiple Sclerosis Society, and representative on the Progressive MS Alliance; has participated on the data safety monitoring board or advisory board for Biogen, Celgene/Bristol Myers Squibb, EMD Serono, National Multiple Sclerosis Society, Novartis, Race to Erase MS (charity), and TG Therapeutics; and has received a patent for "Yablonskiy DA, Sukstansky AL, Wen J, Cross AH. Methods for simultaneous multi-angular relaxometry of tissue using magnetic resonance imaging. Patent 15060-630 (015875)." **Jutta Gärtner**, in the past 3 years, has received fees for lectures and consultancy fees from Bayer, Biogen, Merck, Novartis, and Sanofi, as well as funding for a research project from Novartis. **Ludwig Kappos** has received consultancy fees from Actelion, Bayer Healthcare, Biogen, BMS, Genzyme, Janssen, Japan Tobacco, Merck, Novartis, Roche, Sanofi, Santhera, and TG therapeutics; contracted research from Bayer Healthcare, Biogen, European Union, InnoSwiss, Merck, Novartis, Roche, Swiss MS Society, and Swiss National Research Foundation; speaker fees from Bayer Healthcare, Biogen, Merck, Novartis, Roche, and Sanofi; serves on the steering committee for Actelion, Bayer Healthcare, Biogen, BMS, Genzyme, Janssen, Japan Tobacco, Merck, Novartis, Roche, Sanofi, Santhera, and TG Therapeutics; support of educational activities from Allergan, Bayer Healthcare, Biogen, CSL Behring, Desitin, Genzyme, Merck, Novartis, Roche, Pfizer, Sanofi, Shire, and Teva; and license fees for Neurostatus products. **Alit Bhatt, Haoyi Fu,** and **Ibolya Boer** are employees of Novartis