Poster DMT38

Abhijit Gadkari | abhijit.gadkari@novartis.com

Real-World Persistence and Adherence to Ofatumumab vs Ocrelizumab in Patients With Multiple Sclerosis

Ming-Hui Tai,[1] Qiujun Shao,[1] Brandon Brown,[1] Riley Taiji,[2] Ariane Faucher,[2] Abhijit Gadkari[1]

[1]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [2]StatLog Inc., Montreal, QC, Canada

KEY FINDINGS & CONCLUSIONS

- In a real-world sample of patients with multiple sclerosis, those on ofatumumab (OMB) demonstrated persistence and adherence that was comparable to those on ocrelizumab
- The proportion of patients adherent to OMB at 18 and 24 months was 77% and 71%, respectively
- These findings underscore the convenience of OMB as an at-home self-injectable disease-modifying therapy, translating into high adherence and persistence

Scan to obtain a copy of the poster

https://bit.ly/CMSCDMT38

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

The study was supported by Novartis Pharmaceuticals Corporation

Poster presented at the Consortium of Multiple Sclerosis Centers (CMSC) 38th Annual Meeting • May 29-June 1, 2024 • Nashville, TN, USA

INTRODUCTION

- Multiple sclerosis (MS) is a chronic, irreversible autoimmune disease of the central nervous system[1] that affects close to 1 million US adults[2]
- Currently, no curative treatment exists for MS; however, several disease-modifying therapies (DMTs) were shown to reduce relapse rates and slow disability worsening and disease progression[3]
- Ofatumumab (OMB) and ocrelizumab (OCR) are 2 B-cell monoclonal antibodies with different routes of administration and dosing schedules (OMB: self-administered subcutaneously once monthly; OCR: biannual infusion)
- Real-world (RW) data comparing persistence and adherence to OMB vs OCR are needed, as these factors impact therapeutic success and patient quality of life

OBJECTIVES

- To compare 18- and 24-month persistence and adherence to OMB vs OCR in a RW sample of patients with MS using US administrative claims data

METHODS

Study Design

- A retrospective cohort study was conducted using Optum® Clinformatics® Data Mart Database (August 2019-May 2023; ***study period***), a longitudinal database of medical and pharmacy administrative claims for patients enrolled in commercial insurance and Medicare Advantage plans in the United States
- ***Index date*** = the date of the first OMB or OCR claim, which was by design after OMB US Food and Drug Administration approval on August 20, 2020
- The sample included adults (age ≥18 years) with:
 - ≥1 inpatient primary MS diagnosis (International Classification of Diseases, 10th Revision, Clinical Modification: G35) or ≥2 outpatient MS diagnoses in any position ≥30 days apart in the 12 months prior to (***preindex period***) or 6 months following the index date;
 - First OMB or OCR claim recorded on or after August 20, 2020 (index date); and
 - Continuous enrollment in a health care plan ≥12 months before and ≥24 months after the index date
- Patients treated with OMB were matched 1:1 to patients treated with OCR (***matched OMB*** and ***OCR cohorts***) using greedy nearest neighbor propensity score matching without replacement and a caliper of 0.1. The propensity score was constructed from age at the index date (continuous), sex, race, region, payer type, number of MS relapses in the preindex period (0, 1, 2+), prior DMTs in the preindex period, MS disability in the preindex period, Deyo-Charlson Comorbidity Index (DCCI) in the preindex period (continuous), and Psychiatric Diagnosis Group index in the preindex period (continuous)
- Persistence and adherence were compared between the matched OMB and OCR cohorts in the ***postindex period***, the varying ≥24-month period from the index date until the end of enrollment/study period

Measurements

- ***Persistence*** was defined as the time in months from the index date until treatment discontinuation, where discontinuation was defined as a gap of ≥60 days in index therapy or switch to another DMT. The end date of supply prior to the gap is taken as the discontinuation date. Switch to another DMT is defined as ≥1 claim for another MS-related DMT. The claim date of the other DMT is taken as the switch date
- ***Adherence*** was defined as the proportion of days covered (PDC) ≥0.8. PDC was calculated as: (number of covered days in the period/number of days in the period)
- For OMB, days of supply were determined directly from claims. Overlapping days of supply were accounted for by adding the extra days to the supply of the next dose
- For OCR, days of supply for each infusion was imputed as: min (182 days [date of next infusion – date of current infusion + 1]). No adjustments were made for early administrations

Statistical Analyses

- The percentages of persistent and adherent patients at 18 and 24 months are reported
 - For persistence, a Kaplan–Meier approach is used, and unadjusted p-values at each time point are calculated using Log-rank tests on survival curves truncated at the given time point. Comparisons of matched cohorts are stratified by the matched pair identifier to control for within-pair correlation
 - For percentage adherent, unadjusted p-values are calculated using chi-square tests

RESULTS

Preindex Patient Characteristics

- A total of 498 patients met the inclusion criteria (OMB, n=102; OCR, n=396)
- Prior to matching, the OMB cohort had a higher percentage of female patients, patients residing in the South, those with commercial insurance, those indexed in 2021, individuals identified as Black or African American, and patients with prior DMT use during the preindex period
- The matched cohorts included 98 patients each and were balanced with respect to the majority of demographic and disease characteristics as well as treatment history (**Table 1**). Year of index date, race, presence of fatigue or malaise, and MS disability remained imbalanced after matching
 - In the matched OMB and OCR cohorts (n=196), mean (SD) age at the index date was 51 (11.4) years (**Table 1**). Age at the index date varied widely, ranging from 21 to 79 years. Most (80%) patients were female, 77% were White, 42% were enrolled in a Medicare Advantage plan, and 72% had no relapse in the preindex period
 - Included patients had relatively mild MS disability, with 80% of patients having no or mild Expanded Disability Status Scale–related symptoms. The most common MS-related symptoms and secondary conditions were fatigue or malaise (34%), anxiety (33%), and sensory problems (19%). The mean DCCI value of 0.9 indicates a relatively low level of comorbidity burden in the matched cohorts

MS Treatment History

- One-third (30%) of patients in the matched OMB and OCR cohorts received no DMT in the preindex period (**Table 1**)
- In the matched OMB cohort, the most common DMT in the preindex period was natalizumab (14%), followed by OCR (13%) and dimethyl fumarate (12%) (**Figure 1**)
- In the matched OCR cohort, the most common DMTs received were dimethyl fumarate (28%), followed by natalizumab (18%) and glatiramer acetate (11%) (**Figure 1**)

Key Outcomes

- Persistence and adherence to OMB and OCR were comparable in the matched OMB and OCR cohorts (**Table 2, Figure 2**)
 - The proportions (95% CI) of patients persistent at 18 and 24 months post index in the matched OMB vs OCR cohorts were 69% (61-79%) vs 70% (62-80%) (p=1.00) and 63% (54-74%) vs 59% (50-70%) (p=0.62), respectively
 - Median persistence (95% CI) was 28 months (25, not reached) in the matched OMB cohort and 26 months (24-31) in the matched OCR cohort
 - The proportions of patients adherent at 18 and 24 months post index in the matched OMB vs OCR cohorts were 77% vs 74% (p=0.74) and 71% vs 67% (p=0.54), respectively
 - Mean (SD) PDC was 0.8 (0.3) in the matched OMB and OCR cohorts
 - The nonsignificant differences in persistence and adherence between the matched OMB and OCR cohorts were also observed after adjusting for demographic and clinical characteristics that remained imbalanced after matching (**Figures 3** and **4**)
- Similar patterns of comparable persistence and adherence between OMB and OCR were observed in sensitivity analyses that had less restrictive continuous enrollment requirements post index (12 and 18 months) and that required ≥2 OMB claims or ≥2 OCR infusions 13 to 21 days apart for cohort eligibility (instead of ≥1 claim for both cohorts)

Table 1. Preindex Patient Characteristics

	All (N=196)	Matched OMB cohort (N=98)	Matched OCR cohort (N=98)	p-Value*	SMD†
Age, mean (SD), years	50.62 (11.40)	50.02 (10.95)	51.21 (11.87)	0.468	0.104
Female, n (%)	156 (79.59)	76 (77.55)	80 (81.63)	0.595	0.101
Payer type, n (%)				0.885	0.041
Commercial	114 (58.16)	58 (59.18)	56 (57.14)		
Medicare	82 (41.84)	40 (40.82)	42 (42.86)		
Race, n (%)				**0.581**	**0.206**
White	150 (76.53)	72 (73.47)	78 (79.59)		
Black or African American	22 (11.22)	11 (11.22)	11 (11.22)		
Hispanic	17 (8.67)	10 (10.20)	7 (7.14)		
Unknown	7 (3.57)	5 (5.10)	2 (2.04)		
DCCI, mean (SD)	0.88 (1.49)	0.87 (1.54)	0.90 (1.43)	0.537	0.021
PDG index, mean (SD)	1.01 (1.05)	1.05 (1.10)	0.97 (1.01)	0.714	0.077
MS relapses in preindex period,‡ n (%)				0.933	0.053
0	142 (72.45)	70 (71.43)	72 (73.47)		
1	36 (18.37)	19 (19.39)	17 (17.35)		
2+	18 (9.18)	9 (9.18)	9 (9.18)		
Top 5 MS-related symptom or secondary conditions, n (%)					
Fatigue or malaise	66 (33.67)	39 (39.80)	27 (27.55)	**0.096**	**0.261**
Anxiety	65 (33.16)	35 (35.71)	30 (30.61)	0.544	0.109
Sensory problems	37 (18.88)	18 (18.37)	19 (19.39)	1.000	0.026
Urinary tract infection	34 (17.35)	20 (20.41)	14 (14.29)	0.346	0.162
Muscle weakness	32 (16.33)	15 (15.31)	17 (17.35)	0.847	0.055
MS disability,§ n (%)				**0.233**	**0.303**
No EDSS-related symptoms	154 (78.57)	78 (79.59)	76 (77.55)		
Mild	3 (1.53)	3 (3.06)	0 (0.00)		
Moderate	11 (5.61)	6 (6.12)	5 (5.10)		
Severe	28 (14.29)	11 (11.22)	17 (17.35)		
DMT use in the preindex period, n (%)	138 (70.41)	68 (69.39)	70 (71.43)	0.876	0.045

DCCI, Deyo-Charlson Comorbidity Index; DMT, disease-modifying therapy; ED, emergency department; EDSS, Expanded Disability Status Scale; IV, intravenous; MS, multiple sclerosis; OCR, ocrelizumab; OMB, ofatumumab; OP, outpatient; PDG, Psychiatric Diagnosis Group; SMD, standardized mean difference

*p-Values are estimated using independent samples t-tests for normally distributed continuous variables and using the Wilcoxon Rank Sum test for non-normally distributed continuous variables. Normality was assessed using the Shapiro–Wilk test. For categorical variables where all cells have an expected count ≥5, p-values are estimated using a chi-square test. For categorical variables where ≥1 cell has an expected count <5, p-values are estimated using the Fisher exact test; †SMD values of 0.2-0.5, 0.5-0.8, and >0.8 represent small, medium, and large effect sizes, respectively; ‡Relapse events were defined using a validated claims-based algorithm as a hospitalization with primary MS diagnosis or OP or ED visit with MS diagnosis in any position with a claim for (≥500 mg/day), IV methylprednisolone, corticotropin, or plasma exchange within 7 days.[4] Corticosteroid use occurring within ±5 days of a DMT infusion (for which steroid premedication is indicated) was excluded from the definition of relapse.[5] The date of relapse event was set to the hospital admission date or ED or OP visit date. Multiple qualifying relapse events within 30 days were collapsed into relapse episodes;§MS disability is based on observance of EDSS-related symptoms and durable medical equipment use observed in claims data weighted by severity score. Using a published algorithm, disability levels and definitions are as follows: severe = defined as having ≥1 EDSS-related symptom with a severity score = 3 in any functional system; moderate: defined as having ≥1 EDSS-related symptom with a severity score = 2 in any functional system or having ≥2 functional systems with severity score = 1; mild: defined as having only 1 EDSS-related symptom with a severity score = 1 or having no EDSS-related symptoms observed during the measurement period[6]

Figure 1. Summary of DMTs in the Preindex Period

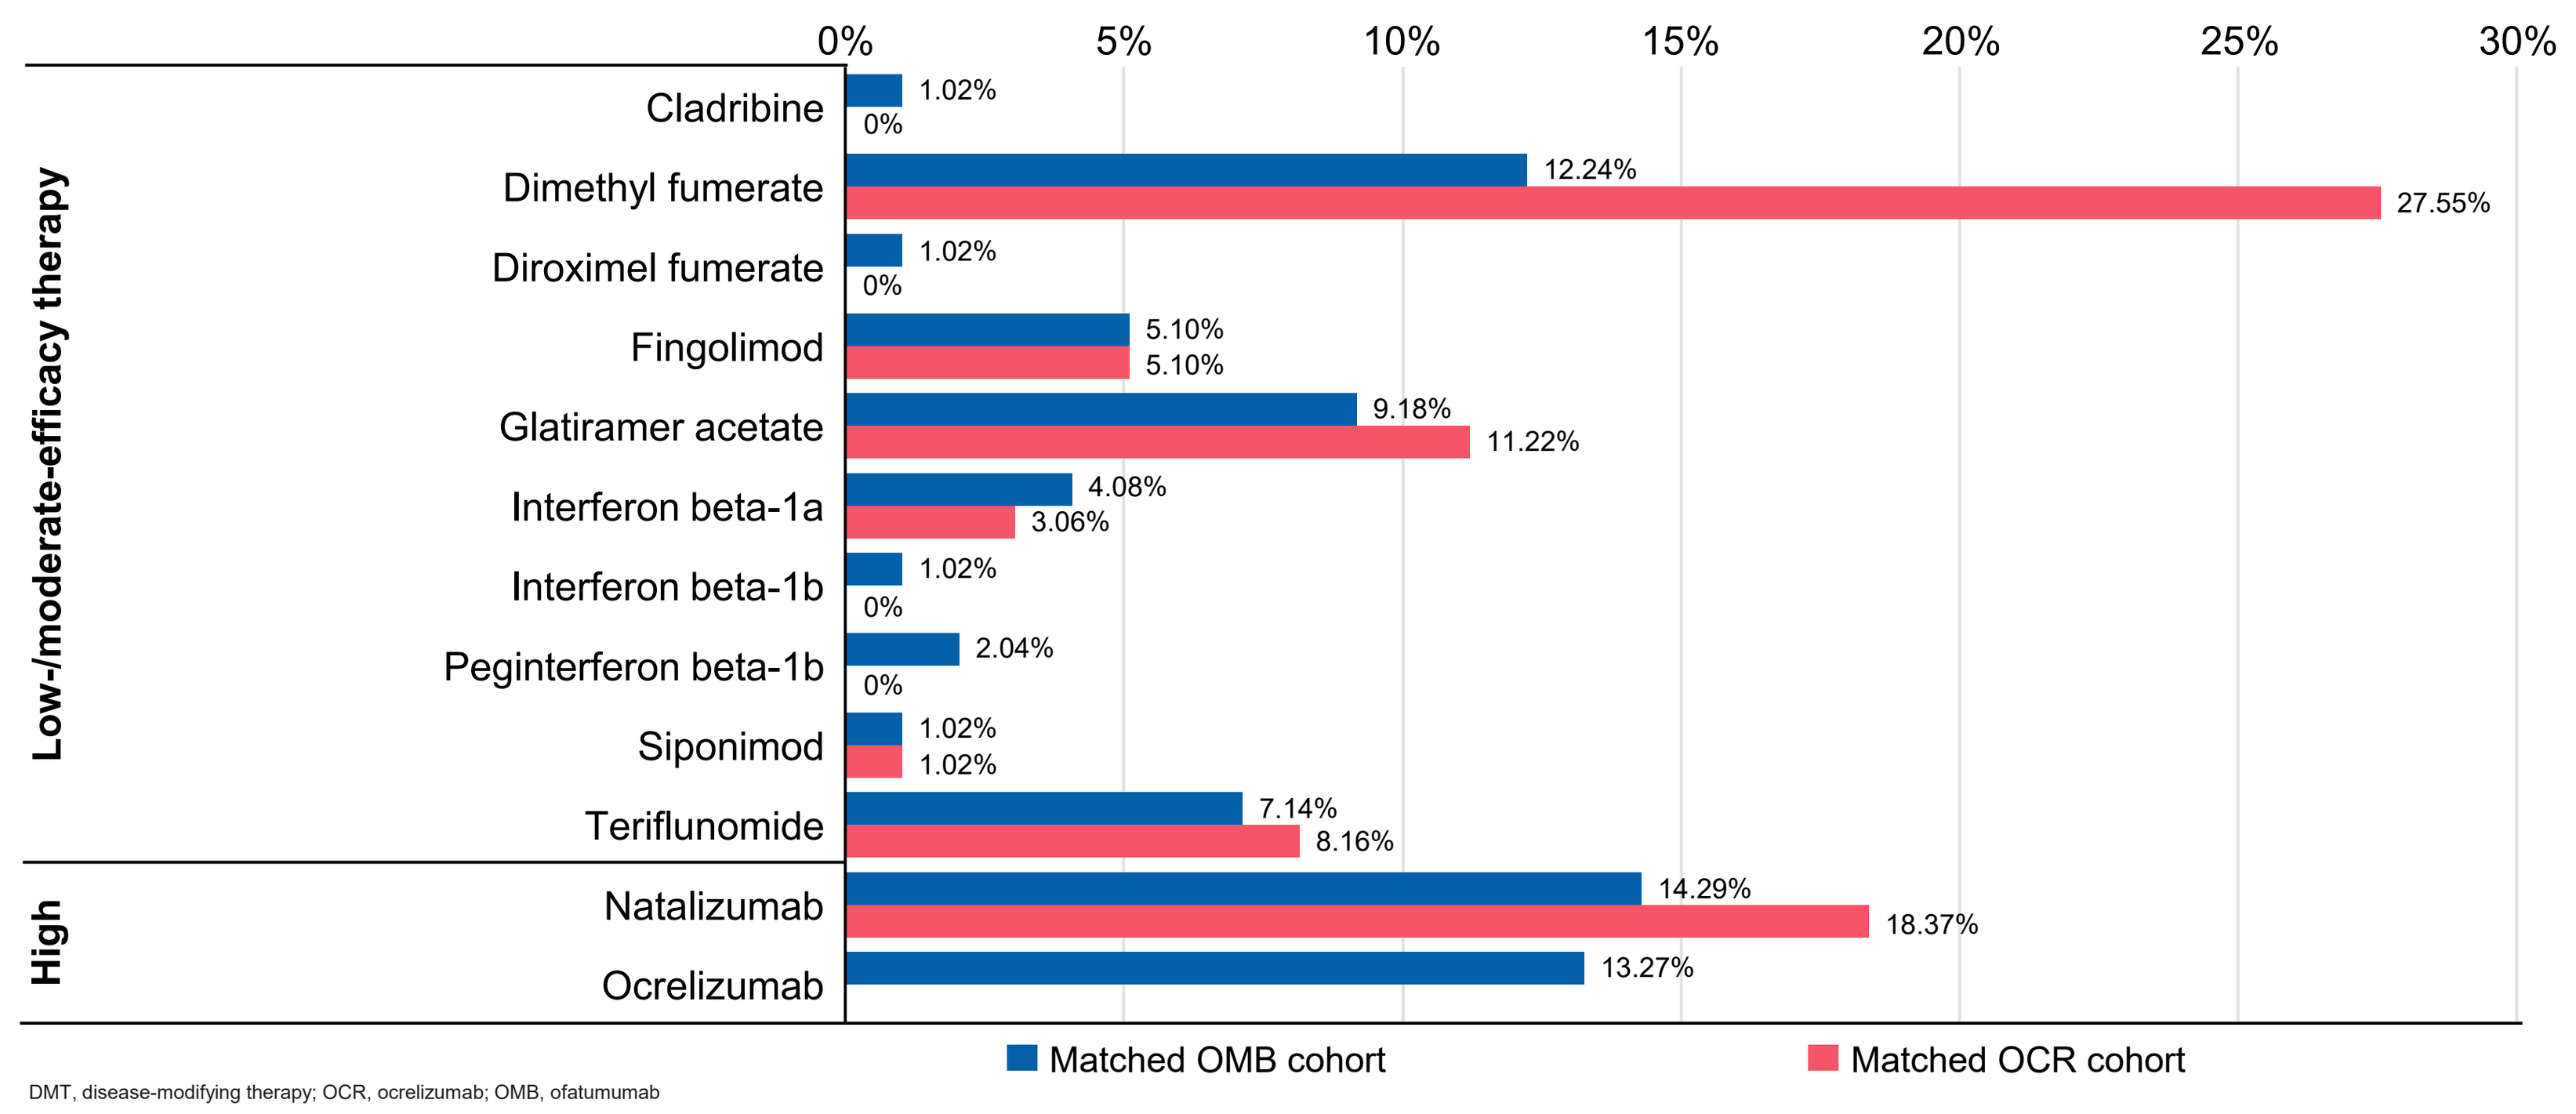
0%
5%
10%
15%
20%
25%
30%
Low-/moderate-efficacy therapy
High
Cladribine 1.02% 0%
Dimethyl fumerate 12.24% 27.55%
Diroximel fumerate 1.02% 0%
Fingolimod 5.10% 5.10%
Glatiramer acetate 9.18% 11.22%
Interferon beta-1a 4.08% 3.06%
Interferon beta-1b 1.02% 0%
Peginterferon beta-1b 2.04% 0%
Siponimod 1.02% 1.02%
Teriflunomide 7.14% 8.16%
Natalizumab 14.29% 18.37%
Ocrelizumab 13.27%
Matched OMB cohort
Matched OCR cohort

DMT, disease-modifying therapy; OCR, ocrelizumab; OMB, ofatumumab

Table 2. Summary of Persistence and Adherence in the Matched OMB and OCR Cohorts

	Matched OMB cohort (N=98)	Matched OCR cohort (N=98)	p-Value*
Persistence			
Persistence at 18 months (95% CI), %	69.39 (60.84-79.14)	70.41 (61.93-80.05)	1.000
Persistence at 24 months (95% CI), %	63.27 (54.41-73.57)	59.18 (50.21-69.76)	0.617
Median persistence in months (95% CI)	27.86 (24.90-NR)	26.22 (24.15-31.34)	–
Adherence			
Adherence at 18 months, %	76.53	74.49	0.739
Adherence at 24 months, %	71.43	67.35	0.538
PDC during 24-month follow-up, mean (SD)	0.80 (0.25)	0.80 (0.26)	–

NR, not reached; OCR, ocrelizumab; OMB, ofatumumab; PDC, proportion of days covered

*For proportion adherent, unadjusted p-values are calculated using binary logistic regression. For proportion persistent at time point, a Kaplan–Meier approach is used; unadjusted p-values at each time point are calculated using Log-rank test on survival curves truncated at the given time point (ie, if survival time > time point, then censor). Comparisons of matched cohorts are stratified by the matched pair identifier to control for within-pair correlation

Figure 2. Kaplan–Meier Curves of Time to Discontinuation (Persistence) in the Matched OMB and OCR Cohorts

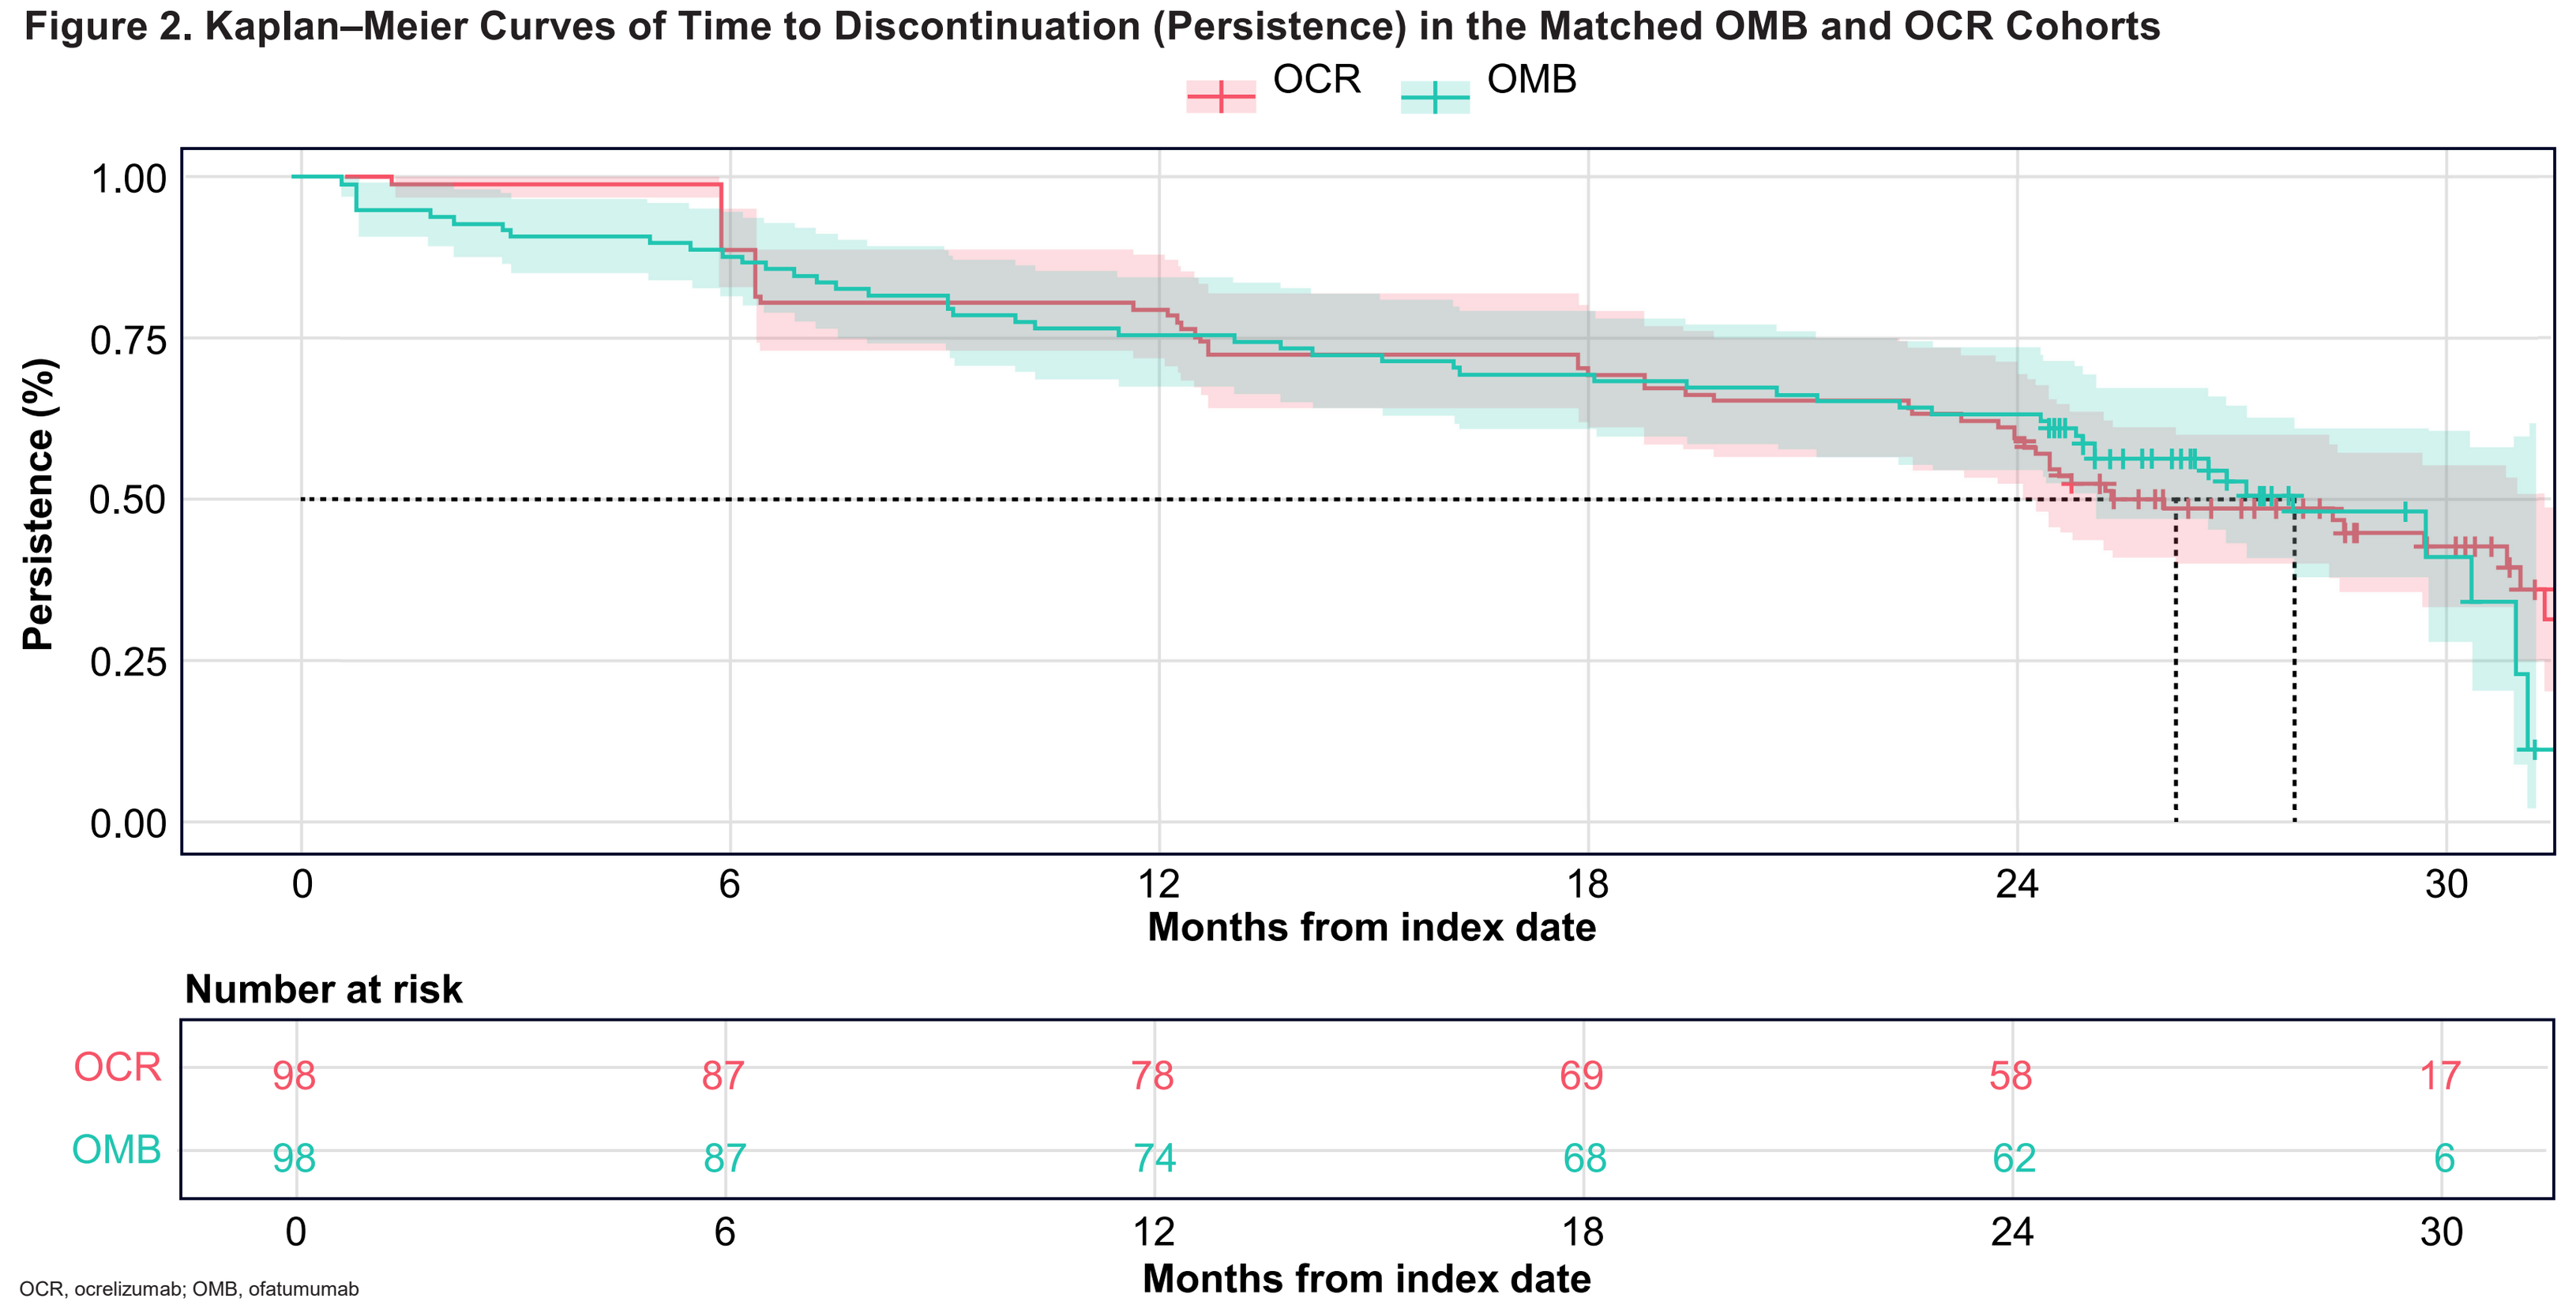
OCR
OMB
Persistence (%)
1.00
0.75
0.50
0.25
0.00
0
6
12
18
24
30
Months from index date
Number at risk
OCR 98 87 78 69 58 17
OMB 98 87 74 68 62 6
0
6
12
18
24
30
Months from index date

OCR, ocrelizumab; OMB, ofatumumab

Figure 3. Cox Proportional Hazards Model of Time to Discontinuation (OMB vs OCR Matched Cohorts)*

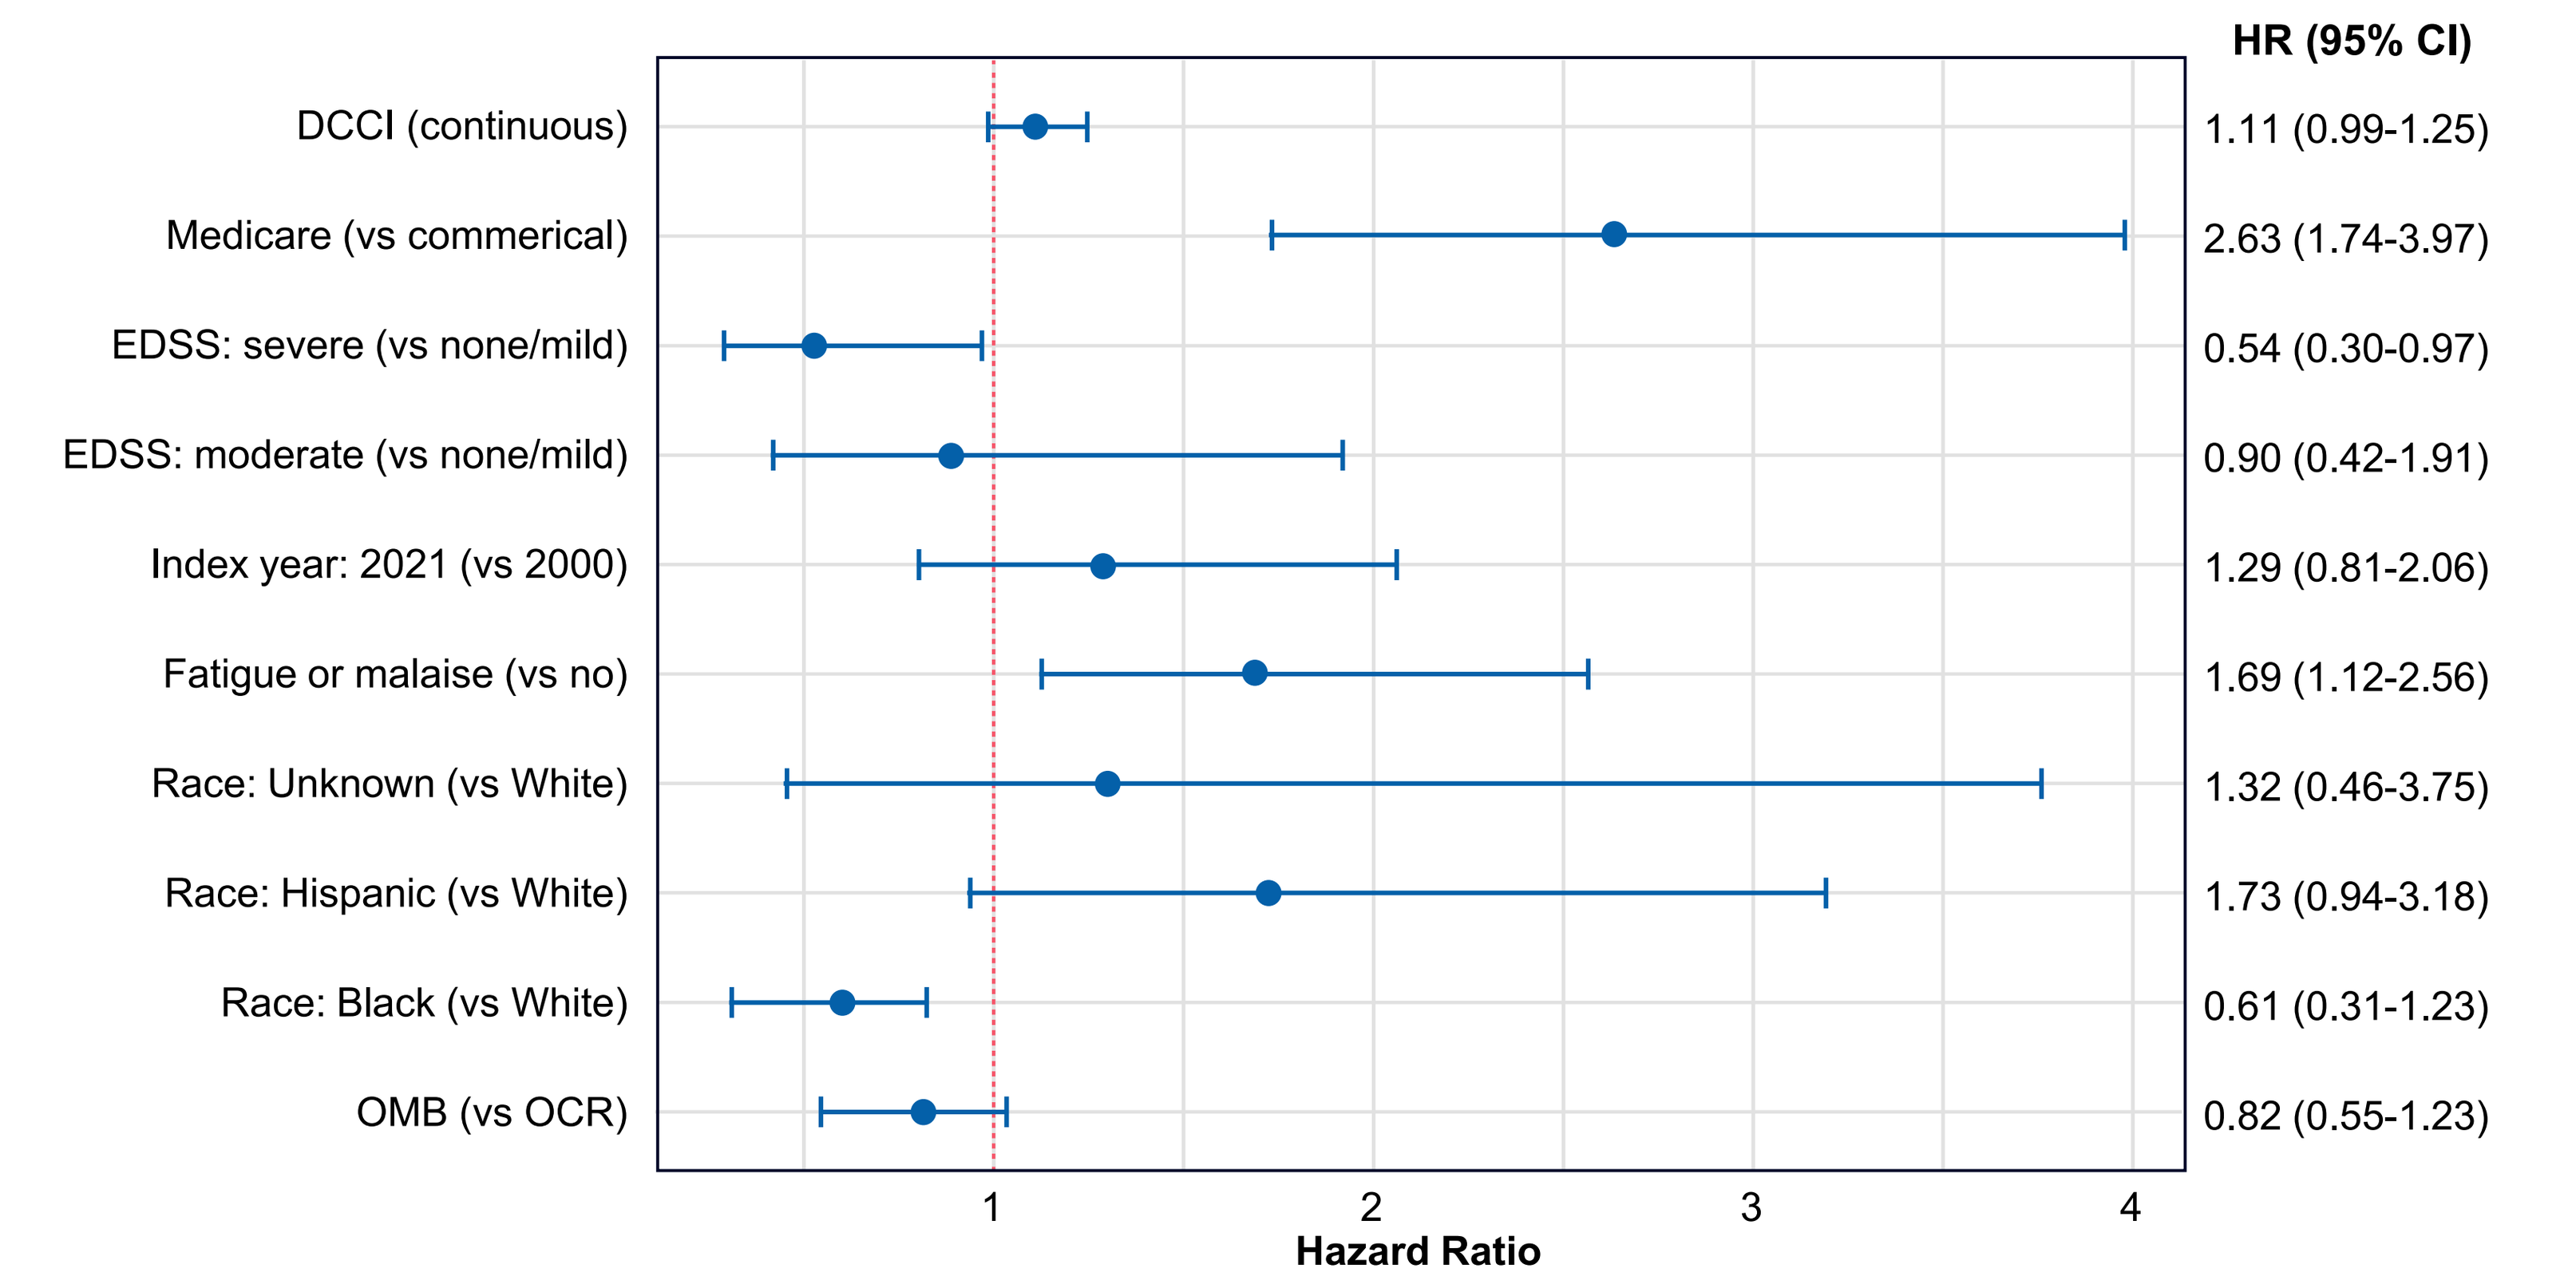
HR (95% CI)
DCCI (continuous) 1.11 (0.99-1.25)
Medicare (vs commerical) 2.63 (1.74-3.97)
EDSS: severe (vs none/mild) 0.54 (0.30-0.97)
EDSS: moderate (vs none/mild) 0.90 (0.42-1.91)
Index year: 2021 (vs 2000) 1.29 (0.81-2.06)
Fatigue or malaise (vs no) 1.69 (1.12-2.56)
Race: Unknown (vs White) 1.32 (0.46-3.75)
Race: Hispanic (vs White) 1.73 (0.94-3.18)
Race: Black (vs White) 0.61 (0.31-1.23)
OMB (vs OCR) 0.82 (0.55-1.23)
1
2
3
4
Hazard Ratio

DCCI, Deyo-Charlson Comorbidity Index; EDSS, Expanded Disability Status Scale; HR, hazard ratio; OCR, ocrelizumab; OMB, ofatumumab
*An HR <1.0 indicates that the covariate is associated with a lower risk of discontinuation

Figure 4. Conditional Logistic Regression Model of Adherence at 24 Months (OMB vs OCR Matched Cohorts)*

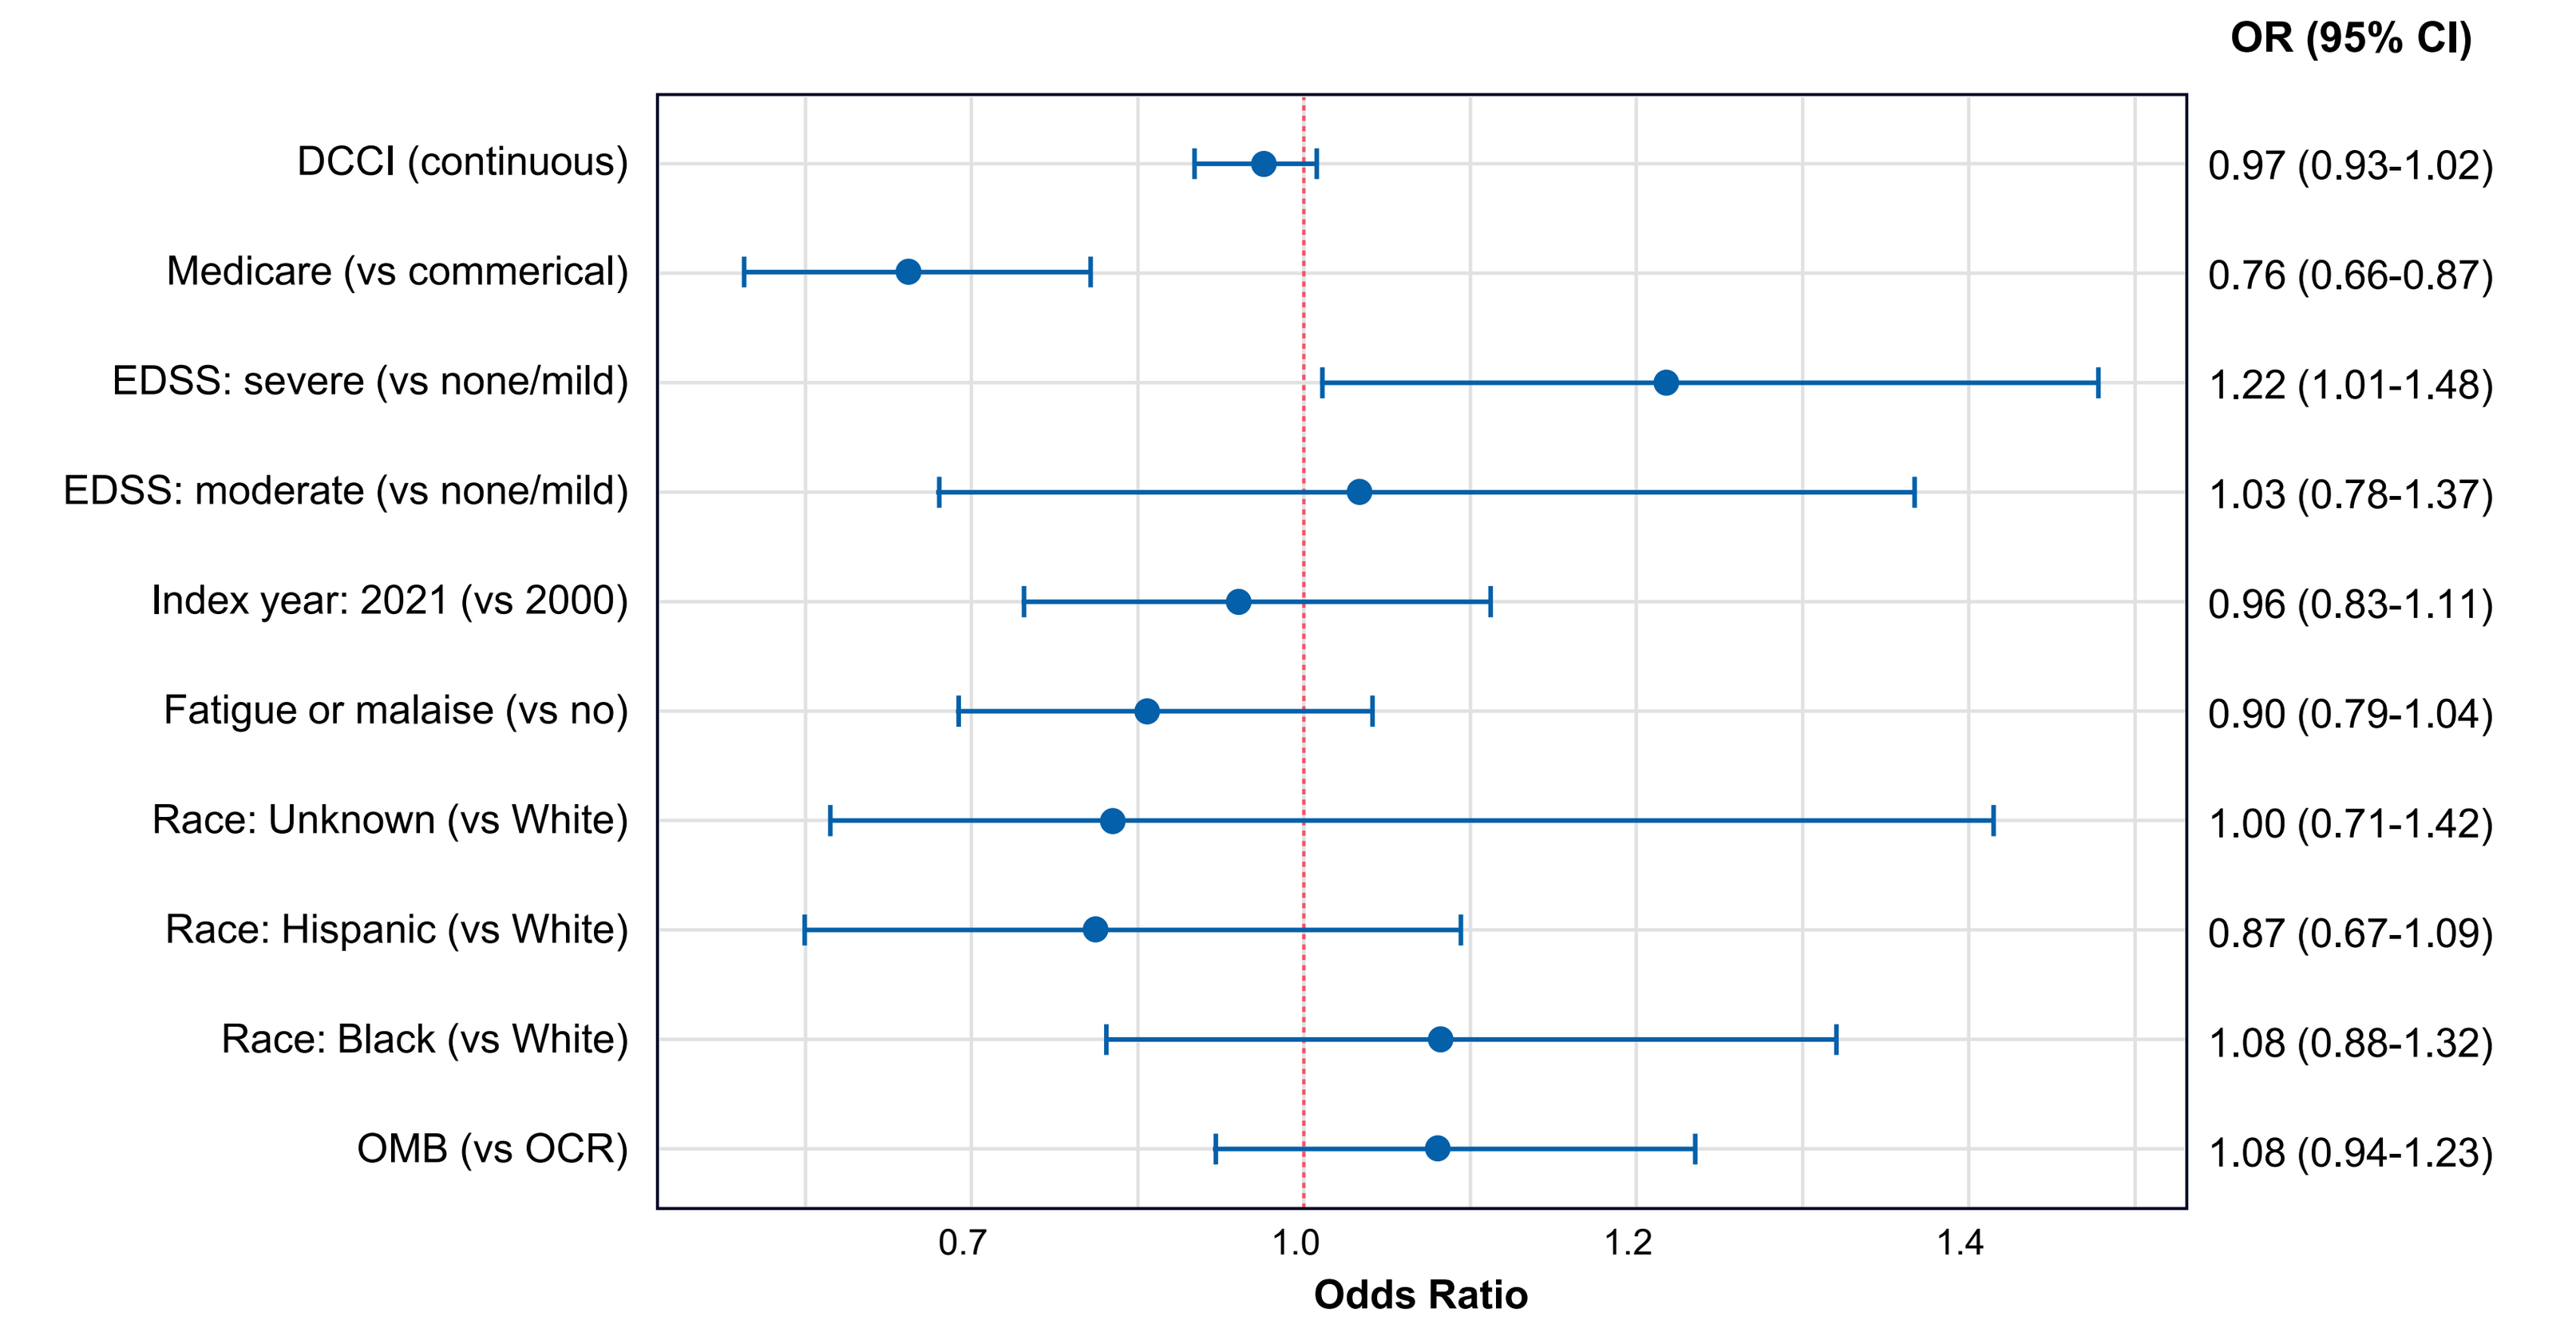
OR (95% CI)
DCCI (continuous) 0.97 (0.93-1.02)
Medicare (vs commerical) 0.76 (0.66-0.87)
EDSS: severe (vs none/mild) 1.22 (1.01-1.48)
EDSS: moderate (vs none/mild) 1.03 (0.78-1.37)
Index year: 2021 (vs 2000) 0.96 (0.83-1.11)
Fatigue or malaise (vs no) 0.90 (0.79-1.04)
Race: Unknown (vs White) 1.00 (0.71-1.42)
Race: Hispanic (vs White) 0.87 (0.67-1.09)
Race: Black (vs White) 1.08 (0.88-1.32)
OMB (vs OCR) 1.08 (0.94-1.23)
0.7
1.0
1.2
1.4
Odds Ratio

DCCI, Deyo-Charlson Comorbidity Index; EDSS, Expanded Disability Status Scale; HR, hazard ratio; MS, multiple sclerosis; OCR, ocrelizumab; OR, odds ratio; OMB, ofatumumab
*An HR <1.0 indicates that the covariate is associated with a lower risk of discontinuation

LIMITATIONS

- Analyses using claims data are dependent on the accuracy and specificity of entered diagnostic codes
- Early discontinuation may be overestimated if treatment occurred outside the purview of the claims data source (eg, if patients continued treatment under a different payer structure, including cash payments or within a clinical trial)
- Data are primarily from employer-sponsored plans and therefore underrepresent patients older than 65 years and those on Medicaid, potentially limiting the generalizability of these findings
- OMB received approval in August 2020, resulting in a relatively limited number of RW patients with sufficient follow-up to assess persistence and adherence

References

1. Martin R et al. *Eur J Immunol*. 2016;46(9):2078-2090. **2.** Wallin MT et al. *Neurology*. 2019;92(10):e1029-e1040. Erratum in: *Neurology*. 2019;93(15):688. **3.** Finkelsztejn A. *Perspect Medicin Chem*. 2014;6:65-72. **4.** Ollendorf DA et al. *J Manag Care Pharm*. 2002;8(6):469-476. **5.** Hersh CM et al. Presentation at: *ECTRIMS 2022*. **6.** Berkovich R et al. *J Med Econ*. 2021;24(1):46-53.

Acknowledgements

Medical writing support was provided by Frankie Sorrell, PhD, of Envision Pharma, Inc. and was funded by Novartis Pharmaceuticals Corporation. This poster was developed in accordance with Good Publication Practice (GPP4) guidelines. Authors had full control of the content and made the final decision on all aspects of this poster

Disclosures

Ming-Hui Tai, **Brandon Brown**, and **Abhijit Gadkari** are employees of Novartis Pharmaceuticals Corporation. **Qiujun Shao** is a former employee of Novartis Pharmaceuticals Corporation. **Riley Taiji** and **Ariane Faucher** are employees of StatLog Inc., which has received funding from Novartis Pharmaceuticals Corporation