Poster NIB09

Silvia R. Delgado | sdelgado1@med.miami.edu

Baseline Serum Neurofilament Light Chain Levels Predict Future Disease Activity Irrespective of Race/Ethnicity: Results From the Phase 3 ASCLEPIOS I/II Trials

Silvia R. Delgado,[1] Enrique Alvarez,[2] Eric Thouvenot,[3,4] Annette F. Okai,[5] Alit Bhatt,[6] Wenjia Wei,[7] Jason Freeman,[8] Tjalf Ziemssen[9]

[1]Department of Neurology, University of Miami, Miller School of Medicine, Miami, FL, USA; [2]Department of Neurology, Rocky Mountain MS Center at the University of Colorado Anschutz Medical Center, University of Colorado, Aurora, CO, USA; [3]Department of Neurology, CHU Nîmes, University Montpellier, Nîmes, France; [4]Institut de Génomique Fonctionnelle, University Montpellier, CNRS, INSERM, Montpellier, France; [5]North Texas Institute of Neurology and Headache, Plano, TX, USA; [6]Novartis Healthcare Pvt. Ltd., Hyderabad, India; [7]Novartis Pharma AG, Basel, Switzerland; [8]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [9]Department of Neurology, University Clinic Carl-Gustav Carus, Dresden, Germany

KEY FINDINGS & CONCLUSIONS

- In racial/ethnic subpopulations (described as Asian, Black, and Other) in the ASCLEPIOS I/II trials, baseline serum neurofilament light chain levels were prognostic for new or enlarging T2 lesions
- These results were consistent with those found in the White subgroup and overall cohort of patients in ASCLEPIOS I/II

Scan to obtain a copy of the poster

https://bit.ly/CMSCNIB09

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

This study is sponsored by Novartis Pharma AG, Basel, Switzerland

Poster presented at the Consortium of Multiple Sclerosis Centers (CMSC) 38th Annual Meeting • May 29-June 1, 2024 • Nashville, TN, USA

Previously presented at the American Academy of Neurology Annual Meeting • April 13-18, 2024 • Denver, CO, USA and at Americas Committee for Treatment and Research in Multiple Sclerosis (ACTRIMS) Forum • February 29–March 2, 2024 • West Palm Beach, FL, USA

INTRODUCTION

- A challenge encountered in clinical practice with relapsing multiple sclerosis (RMS) is the difficulty in prognosticating the risk of future disease activity because of the variable disease course across patients[1]
- There are disparities in multiple sclerosis (MS) prognosis and disease severity between different racial/ethnic groups, with certain ethnic groups often experiencing increased disease burden as compared with White groups[2]
 - A biomarker that can prognosticate disease activity may help optimize individualized patient care and limit irreversible neurological damage, even in the absence of overt clinical symptoms or radiological signs[3]
- In the phase 3 ASCLEPIOS I/II trials (ofatumumab vs teriflunomide in people with RMS [pwRMS; NCT02792218/ NCT02792231]), a preplanned analysis of baseline serum neurofilament light chain (sNfL) levels, based on being above or below the baseline median, showed that sNfL levels were prognostic for on-study lesion formation and brain volume loss in the overall population and in recently diagnosed treatment-naïve participants[4]
- The prognostic use of sNfL for future disease activity among diverse racial/ethnic subgroups from the trials has not yet been explored
 - Such data can help support the generalizability of sNfL as a prognostic biomarker for pwRMS with different racial/ ethnic backgrounds

OBJECTIVE

- To evaluate the prognostic value of baseline sNfL for future magnetic resonance imaging (MRI) disease activity in diverse racial/ethnic subpopulations (Asian, Black, and Other) of pwRMS in ASCLEPIOS I/II

METHODS

Study Design

- ASCLEPIOS I/II were 2 phase 3, double-blind, active-controlled trials in which participants with RMS were randomized to receive either ofatumumab or teriflunomide for ≤30 months
- Participants aged 18-55 years with a diagnosis of RMS, Expanded Disability Status Scale (EDSS) score 0-5.5, ≥1 relapse in the year before screening or ≥2 relapses in the last 2 years before screening, or ≥1 gadolinium-enhancing (Gd+) lesion on MRI in the year before randomization were included
- Due to the event-driven design, participants were switched to open-label ofatumumab following a variable duration in the core study (**Figure 1**)
 - The first switches to open-label treatment occurred during Year 1, and all participants were switched by the end of Year 3
 - The median time in the core study was 1.6 years (1.5 years in ASCLEPIOS I and 1.6 years in ASCLEPIOS II) and >30% of the participants had a time in trial longer than 2 years
- The baseline sNfL cutoff was predefined in the clinical study protocol (ie, before measuring sNfL or any clinical or radiological outcomes) as the median sNfL value for the overall population across ASCLEPIOS I/II (9.3 pg/mL)
- Participants were stratified as having high (≥9.3 pg/mL) vs low (<9.3 pg/mL) sNfL based on the median baseline sNfL level

Figure 1. Time Spent in the ASCLEPIOS I/II Trials

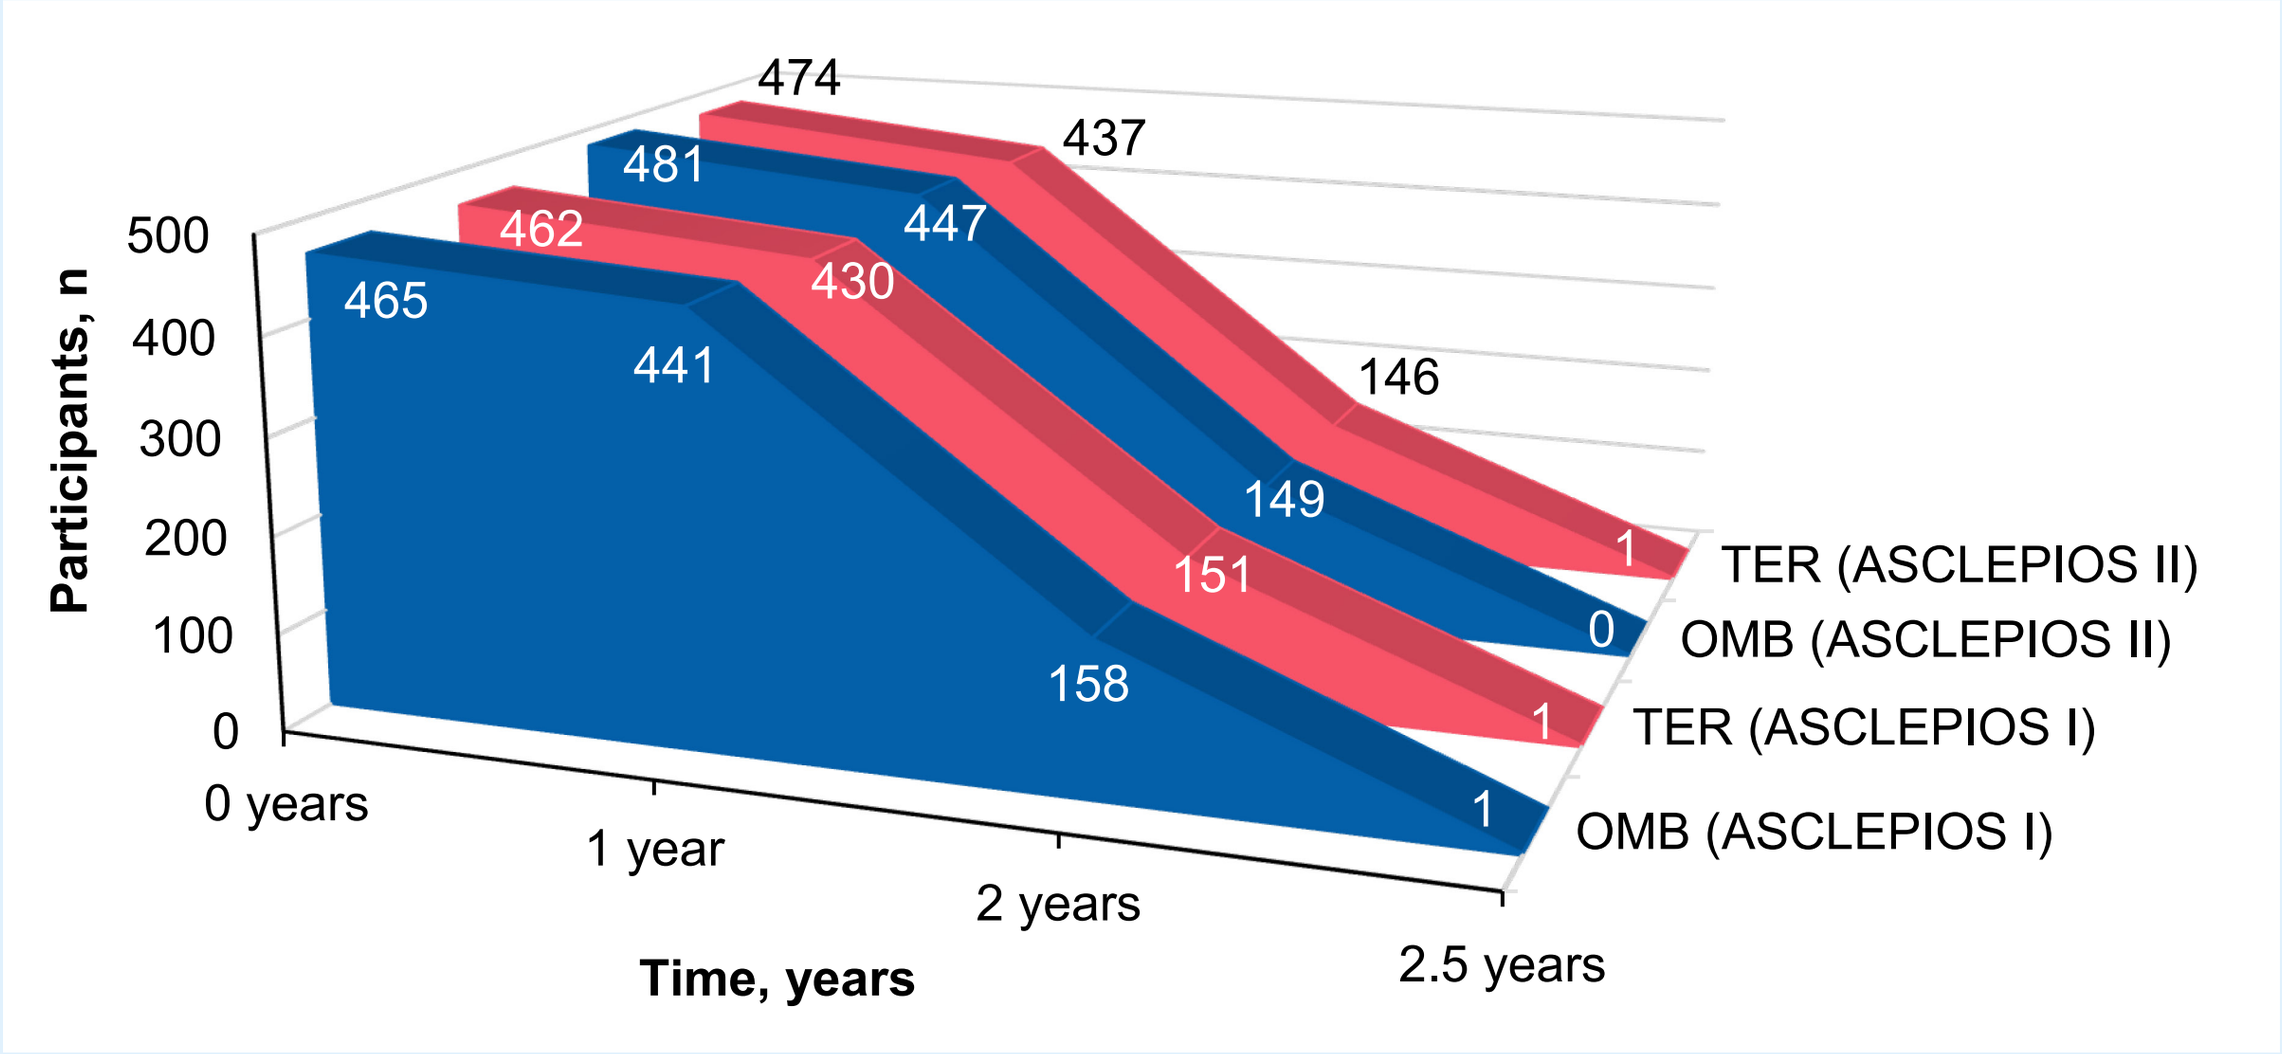
Participants, n
500
400
300
200
100
0
0 years
1 year
2 years
2.5 years
Time, years
465
441
158
1
462
430
151
1
481
447
149
0
474
437
146
1
TER (ASCLEPIOS II)
OMB (ASCLEPIOS II)
TER (ASCLEPIOS I)
OMB (ASCLEPIOS I)

OMB, ofatumumab; TER, teriflunomide
Time in the trial is defined as the time each participant spent in the trial (Day 1 to the end of the study)

Assessments

- Quantification of sNfL levels was performed centrally (Navigate BioPharma Services, Carlsbad, CA, USA) as a single batch at the end of the trials, using a validated Quanterix Simoa® NF-light advantage kit
- MRI scans were performed at baseline, Months 12 and 24, and end of treatment/study
- The prognostic value of high vs low baseline sNfL for the annualized rate of new or enlarging T2 (neT2) lesions was assessed, irrespective of treatment, in the overall population and racial/ethnic subgroups (including White, Asian, Black, and Other [ie, racial subgroups described as “other” or “unknown” upon data collection]) from ASCLEPIOS I/II

Statistical Analyses

- The neT2 lesion number on the last available scan (relative to baseline scan) was analyzed using a negative binomial model with time (in years) between the 2 scans as offset, adjusting for baseline sNfL groups
- The prognostic value was assessed via the lesion rate ratio attained using this single cutoff threshold for high vs low sNfL

RESULTS

Participant Characteristics

- Of the 1882 participants randomized in the ASCLEPIOS I/II trials, 1746 had baseline sNfL data, including 1678 participants with neT2 lesion and sNfL data available
- Baseline characteristics were similar between the sNfL groups, except the mean number of Gd+ lesions and T2 lesion volume, which were considerably higher in participants with high vs low baseline sNfL (**Table 1**)

Table 1. Demographic and Disease Characteristics of Participants by Baseline sNfL Category

Characteristic	Low sNfL category (<9.3 pg/mL) (N=876*)	High sNfL category (≥9.3 pg/mL) N=870*)
Age, years	38.6 (8.5)	37.8 (9.7)
Female, n (%)	588 (67.1)	602 (69.2)
Race, n (%)		
White	780 (89.0)	763 (87.7)
Asian	33 (3.8)	32 (3.7)
Black	27 (3.1)	34 (3.9)
Other†	36 (4.1)	41 (4.7)
MS duration since first symptom, years	8.3 (7.2)	7.9 (6.9)
Previously treated with DMT, n (%)	537 (61.3)	507 (58.3
Number of relapses in the year before the study	1.2 (0.7)	1.3 (0.7)
Time since onset of most recent relapse, months	7.8 (13.5)	7.0 (9.3)
EDSS score	2.8 (1.3)	2.9 (1.4)
Normalized brain volume, cm³	1447.6 (74.8)	1437.2 (81.0)
Number of Gd+ T1 lesions	0.4 (1.2)	2.6 (5.4)
Participants free of Gd+ T1 lesions, n (%)	679 (77.5)	383 (44.0)
T2 lesion volume, cm³	9.4 (10.6)	16.7 (15.0)
Median sNfL, pg/mL	6.76	14.23

DMT, disease-modifying therapy; EDSS, Expanded Disability Status Scale; Gd+, gadolinium-enhancing; MS, multiple sclerosis; sNfL, serum neurofilament light chain
Data are expressed as mean (SD) unless specified otherwise
*Only participants with nonmissing baseline sNfL values are included; †Racial subgroups were described as “other” or “unknown” upon data collection

Prognostic Value of Baseline sNfL for neT2 Lesions

- The annualized mean rate of neT2 lesions was consistently higher in participants with high vs low sNfL levels across all racial/ethnic subgroups; these results were also significantly higher in participants with high vs low sNfL levels for the overall ASCLEPIOS I/II population (**Figure 2**)

Figure 2. Mean Annualized Rate of neT2 Lesions by Racial/Ethnic Subgroups and in the Overall Population per Baseline sNfL*

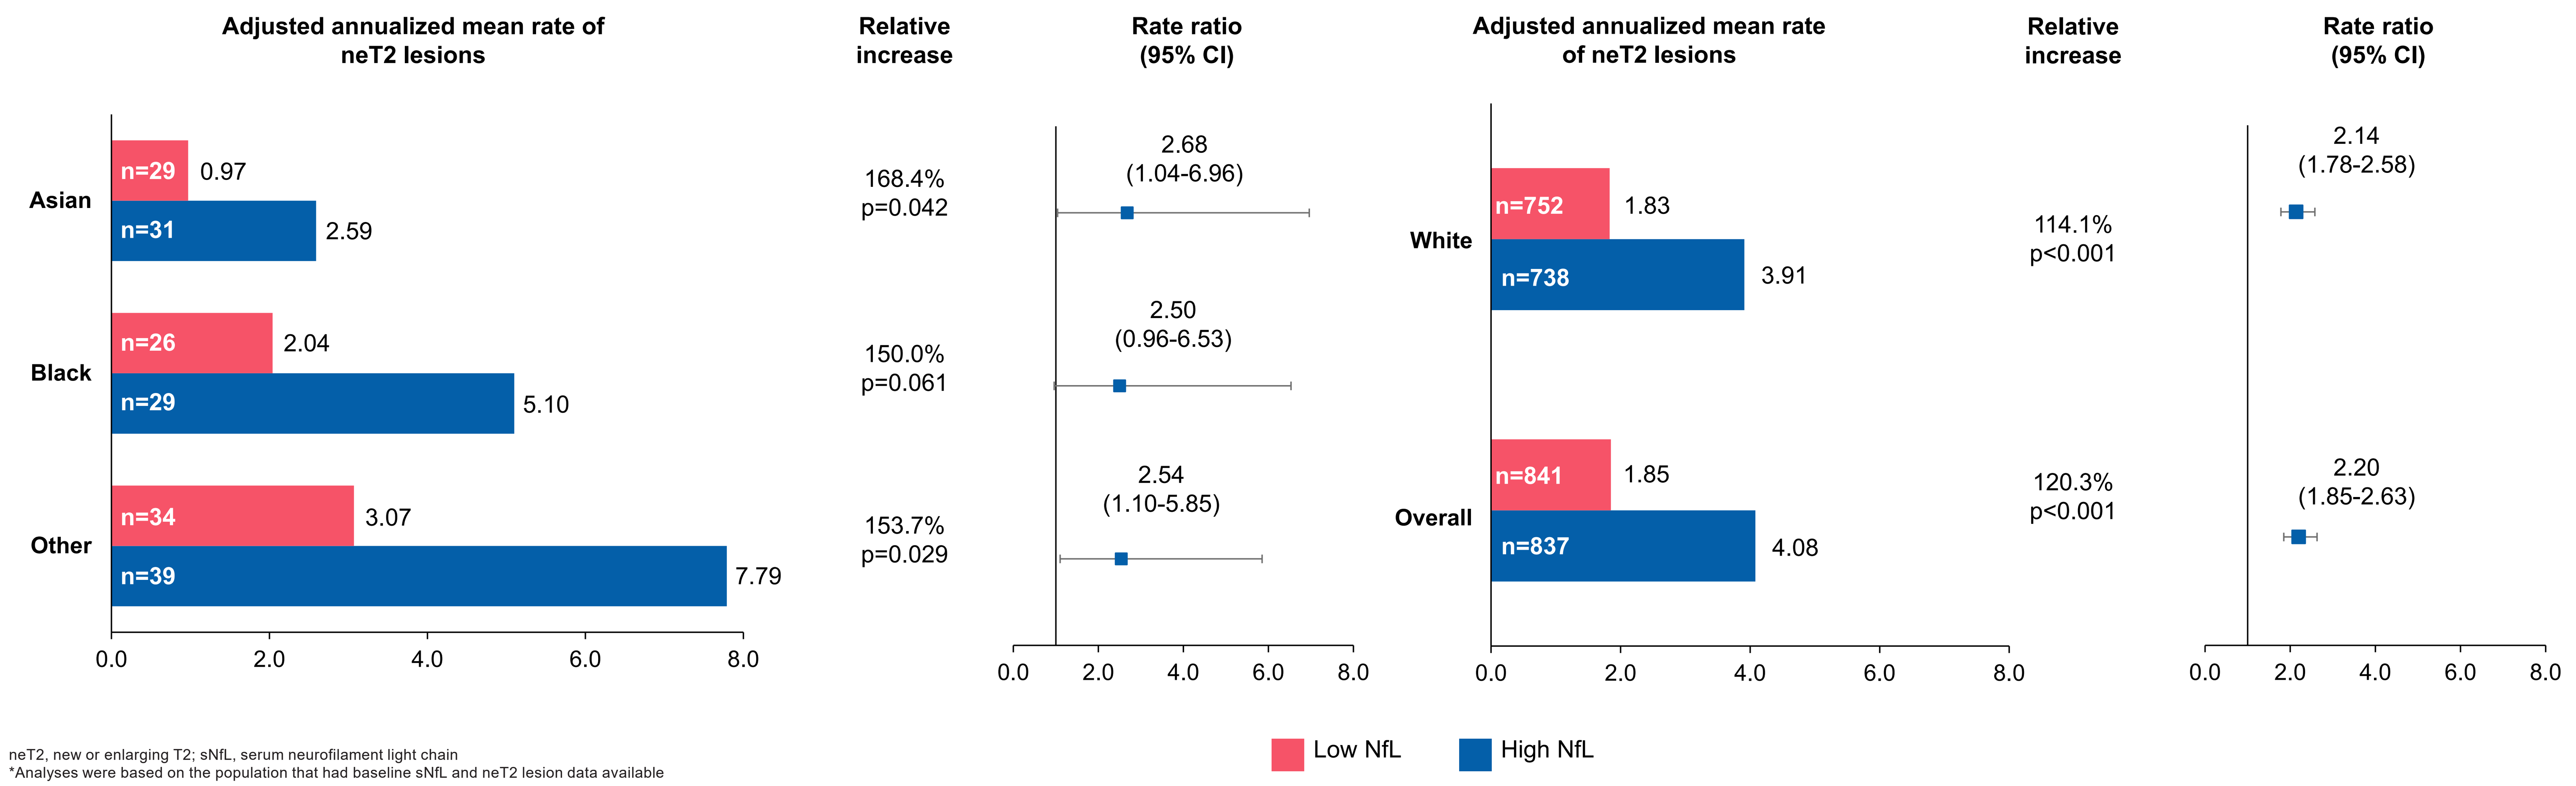
Adjusted annualized mean rate of neT2 lesions
Relative increase
Rate ratio (95% CI)
Asian
n=29 0.97
n=31 2.59
168.4% p=0.042
2.68 (1.04-6.96)
Black
n=26 2.04
n=29 5.10
150.0% p=0.061
2.50 (0.96-6.53)
Other
n=34 3.07
n=39 7.79
153.7% p=0.029
2.54 (1.10-5.85)
White
n=752 1.83
n=738 3.91
114.1% p<0.001
2.14 (1.78-2.58)
Overall
n=841 1.85
n=837 4.08
120.3% p<0.001
2.20 (1.85-2.63)
0.0
2.0
4.0
6.0
8.0
Low NfL
High NfL

neT2, new or enlarging T2; sNfL, serum neurofilament light chain
*Analyses were based on the population that had baseline sNfL and neT2 lesion data available

Limitations

- This analysis evaluated the prognostic value of sNfL irrespective of treatment; treatment effect on sNfL was not assessed
- The data presented for the racial/ethnic subgroups from ASCLEPIOS I/II are based on small sample sizes
- Based on the preplanned nature of the analysis, participants were stratified by baseline median sNfL value into “high” or “low” with the intention to divide a typical RMS population for a phase 3 trial into groups of equal size with higher vs lower than median sNfL
- Results reported here are based on the protocol-defined single sNfL threshold; future work should evaluate how this single sNfL threshold could be optimized with a specific target and population in mind
- The use of a single NfL threshold may be applicable mainly to relatively young RMS populations (18-55 years), such as the population included in these trials; since prognostication of MS disease activity is primarily a concern in patients who are younger and/or are early in their disease course, this is not considered a strong limitation
- The data presented in this study are based on a population that was selected according to the ASCLEPIOS inclusion/exclusion criteria, and although this population represents a typical population suitable for phase 3 trials/regulatory purposes, it may not reflect the broader population of individuals with RMS seen in everyday clinical practice

References

1. Oh J, et al. *Curr Opin Neurol*. 2018;31(6):752-759.
2. Rivas-Rodríguez E, Amezcua L. *Neurol Clin*. 2018;36(1):151-162.
3. Thebault S, et al. *Mult Scler*. 2022;28(10):1491-1497.
4. Ziemssen T, et al. *Front Immunol*. 2022;13:852563.

Acknowledgments

Medical writing support was provided by Brianna Fitzmaurice (employee of Novartis Ireland Ltd., Dublin, Ireland), and creative design support by Edward Kattekola (employee of Novartis Healthcare Pvt. Ltd., Hyderabad, India). Editorial assistance for this poster was provided by Envision Pharma Group and was funded by Novartis Pharmaceuticals Corporation. The final responsibility for the content lies with the authors

Disclosures

Silvia R. Delgado has received research support from EMD Serono and Novartis. **Enrique Alvarez** has received compensation for consulting from Alexion, Biogen, Celgene/Bristol Myers Squibb, EMD Serono/Merck, Genentech/Roche, Horizon/Amgen, Motric Bio, Novartis, Sanofi, Scionic, and TG Therapeutics and for research from Atara, Biogen, Genentech/Roche, the National Institutes of Health, the National Multiple Sclerosis Society, Novartis, Patient-Centered Outcomes Research Initiative, Rocky Mountain MS Center, Sanofi, and TG Therapeutics. **Eric Thouvenot** has received personal compensation for serving as a consultant for Biogen, Merck, Novartis, and Teva and for serving on a Scientific Advisory or Data Safety Monitoring board for Novartis and Roche, and unconditional research support from Biogen and Novartis. **Annette F. Okai** has received speaking and consulting fees from Biogen, Bristol Myers Squibb, EMD Serono, Genentech, and Sanofi Genzyme and has received research support from Alexion, Biogen, EMD Serono, Genentech/Roche, Novartis, Sanofi Genzyme, and TG Therapeutics. **Jason Freeman**, **Alit Bhatt**, and **Wenjia Wei** are employees of Novartis. **Tjalf Ziemssen** has received personal compensation for participating in advisory boards, trial steering committees, and data and safety monitoring committees, as well as for scientific talks and project support from Almirall, BAT, Bayer, Biogen, Celgene, Merck, Novartis, Roche, Sanofi Genzyme, Teva, and Vitaccess